A High Throughput LC-MS/MS Method for the Quantitation of Voriconazole in 0.05 mL of Human Plasma

Elizabeth M. Peterson, Erica Nachi, Chris J. Kafonek, Corey Ohnmacht, Curtis Sheldon
Celerion, Lincoln, NE

PURPOSE:

- The purpose of this project was to develop a sensitive and high-throughput LC-MS/MS method suitable for the determination of voriconazole in human plasma samples from bioequivalence or drug interaction studies using a small sample volume.

BACKGROUND:

- Voriconazole is an antifungal medication that is generally used to treat serious, invasive fungal infections.
- Peak plasma concentrations of approximately 2.3 µg/mL are achieved 1 to 2 hours after oral administration of 400 mg in the fasted state.
- The pharmacokinetics are non-linear due to metabolic saturation of the human hepatic cytochrome P450 enzymes, CYP2C19, CYP2C9 and CYP3A4. CYP2C19 accounts for a large part of voriconazole metabolism and exhibits genetic polymorphism in humans.
- The range of the assay including dilution integrity is appropriate for analysis of samples from pharmacokinetic and drug interaction studies.
- Using a low sample volume allowed for less blood draw volume and the ability to measure additional analytes from one sample.

METHODS:

- **Extraction:** Voriconazole human plasma (K2 EDTA) samples (0.0500 mL) were diluted with an ammonium acetate buffer, extracted with n-butyl chloride using an automated sample handling system, evaporated to dryness and reconstituted in an acetonitrile:water solution.
- **Chromatography:** Voriconazole was chromatographically separated from other matrix components on an Advanced Chromatography Technologies, ACE® C18 analytical column with an isocratic mobile phase consisting of acetonitrile, water, and formic acid.
- **Detection:** An AB SCIEX 4000, using an ESI interface, detected positive ions in the multiple reaction monitoring mode with a runtime of 1.5 minutes.

RESULTS:

- The calibration standards were linear over the range of 5.00 to 2000 ng/mL. Inter-batch precision (% CV) of voriconazole quality control samples was less than 5.2% (Table 1). Inter-batch accuracy (% Bias) of voriconazole quality controls was -10.6 to 2.0% (Table 1).
- Samples having a concentration above the upper limit of the calibration standard range were diluted with blank human plasma, and results show that samples with a concentration up to 15,000 ng/mL could be quantified after the application of an appropriate dilution factor.
- Assay selectivity was demonstrated by the quantitation of ten separate lots of human plasma fortified with known concentrations of voriconazole. No significant matrix effect was observed in any of the ten lots fortified at the LLOQ concentration or at the high quality control sample concentration for voriconazole (Table 2).
- Stability of the analyte in freshly collected human whole blood was established for up to 120 minutes in polypropylene tubes at ambient temperature under white light.
- A post-column infusion showed no areas of significant enhancement or suppression at the retention time of the analyte or internal standard (Figure 2).
- Post-preparative stability in injection solvent (quantitation against freshly extracted standards) was established for 158 hours at 5°C.
- Processed sample stability in injection solvent (reinjection) was established for 163 hours at 5°C.
- Short-term stability in plasma was established for 56 hours at ambient temperature under white light.
- Freeze and thaw stability in plasma was established for six freeze (-20°C) and thaw cycles (ambient temperature).

Figure 1. Voriconazole Structure

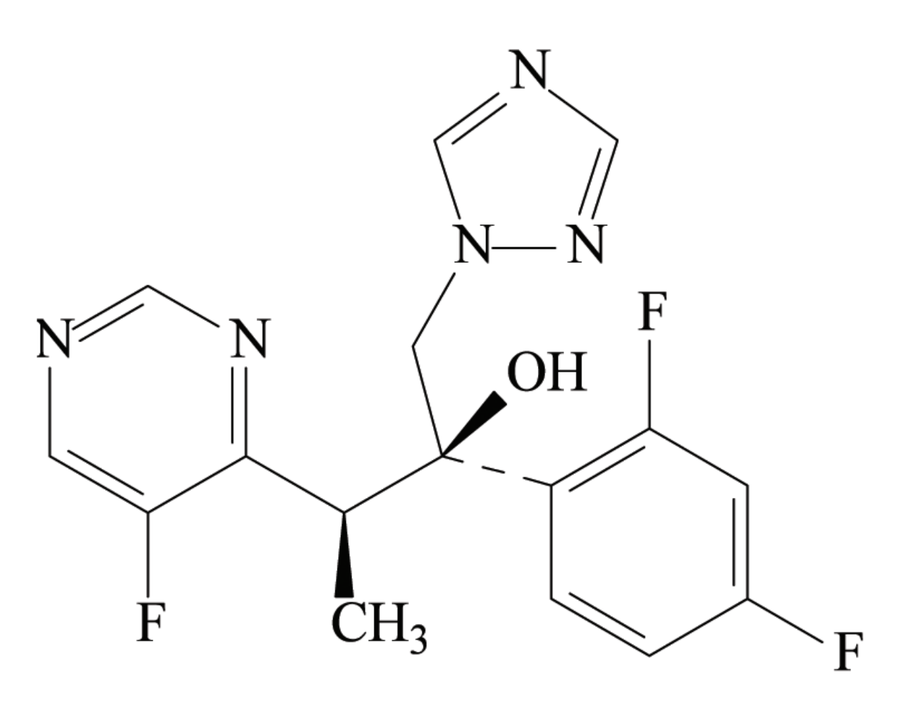

CONCLUSIONS:

- The bioanalytical assay for the quantitation of voriconazole in human plasma met acceptance criteria for precision, accuracy, sensitivity, selectivity, and stability. The low sample volume required for analysis enabled the use of an automated liquid-liquid extraction procedure that greatly increased sample throughput. The method has successfully been used to support clinical studies.

Table 1. Inter-batch Precision and Accuracy

	LLOQ QC 5.00 ng/mL	QC A 15.0 ng/mL	QC B 300 ng/mL	QC C 1500 ng/mL
Inter-Batch Mean	4.47	14.6	306	1500
Inter-Batch SD	0.234	0.513	6.29	34.3
Inter-Batch % CV	5.2	3.5	2.1	2.3
Inter-Batch % Bias	-10.6	-2.7	2.0	0.0

Table 2. Matrix Effect for Voriconazole in Human Plasma (EDTA)

		LLOQ		High	
	Lot#	5.00 ng/mL	% Bias	1500 ng/mL	% Bias
	1	5.10	+2.0	1460	-2.7
	2	5.70	+14.0	1460	-2.7
	3	5.40	+8.0	1420	-5.3
	4	5.21	+4.2	1440	-4.0
	5	5.14	+2.8	1480	-1.3
	6	4.96	-0.8	1500	+0.0
	7	5.02	+0.4	1500	+0.0
	8	4.62	-7.6	1460	-2.7
	9	5.06	+1.2	1500	+0.0
	10	5.22	+4.4	1410	-6.0
Mean		5.14		1460	
% CV		5.5		2.2	
% Bias		2.8		-2.7	
n		10		10	

Figure 2. Post-Column Matrix Infusion

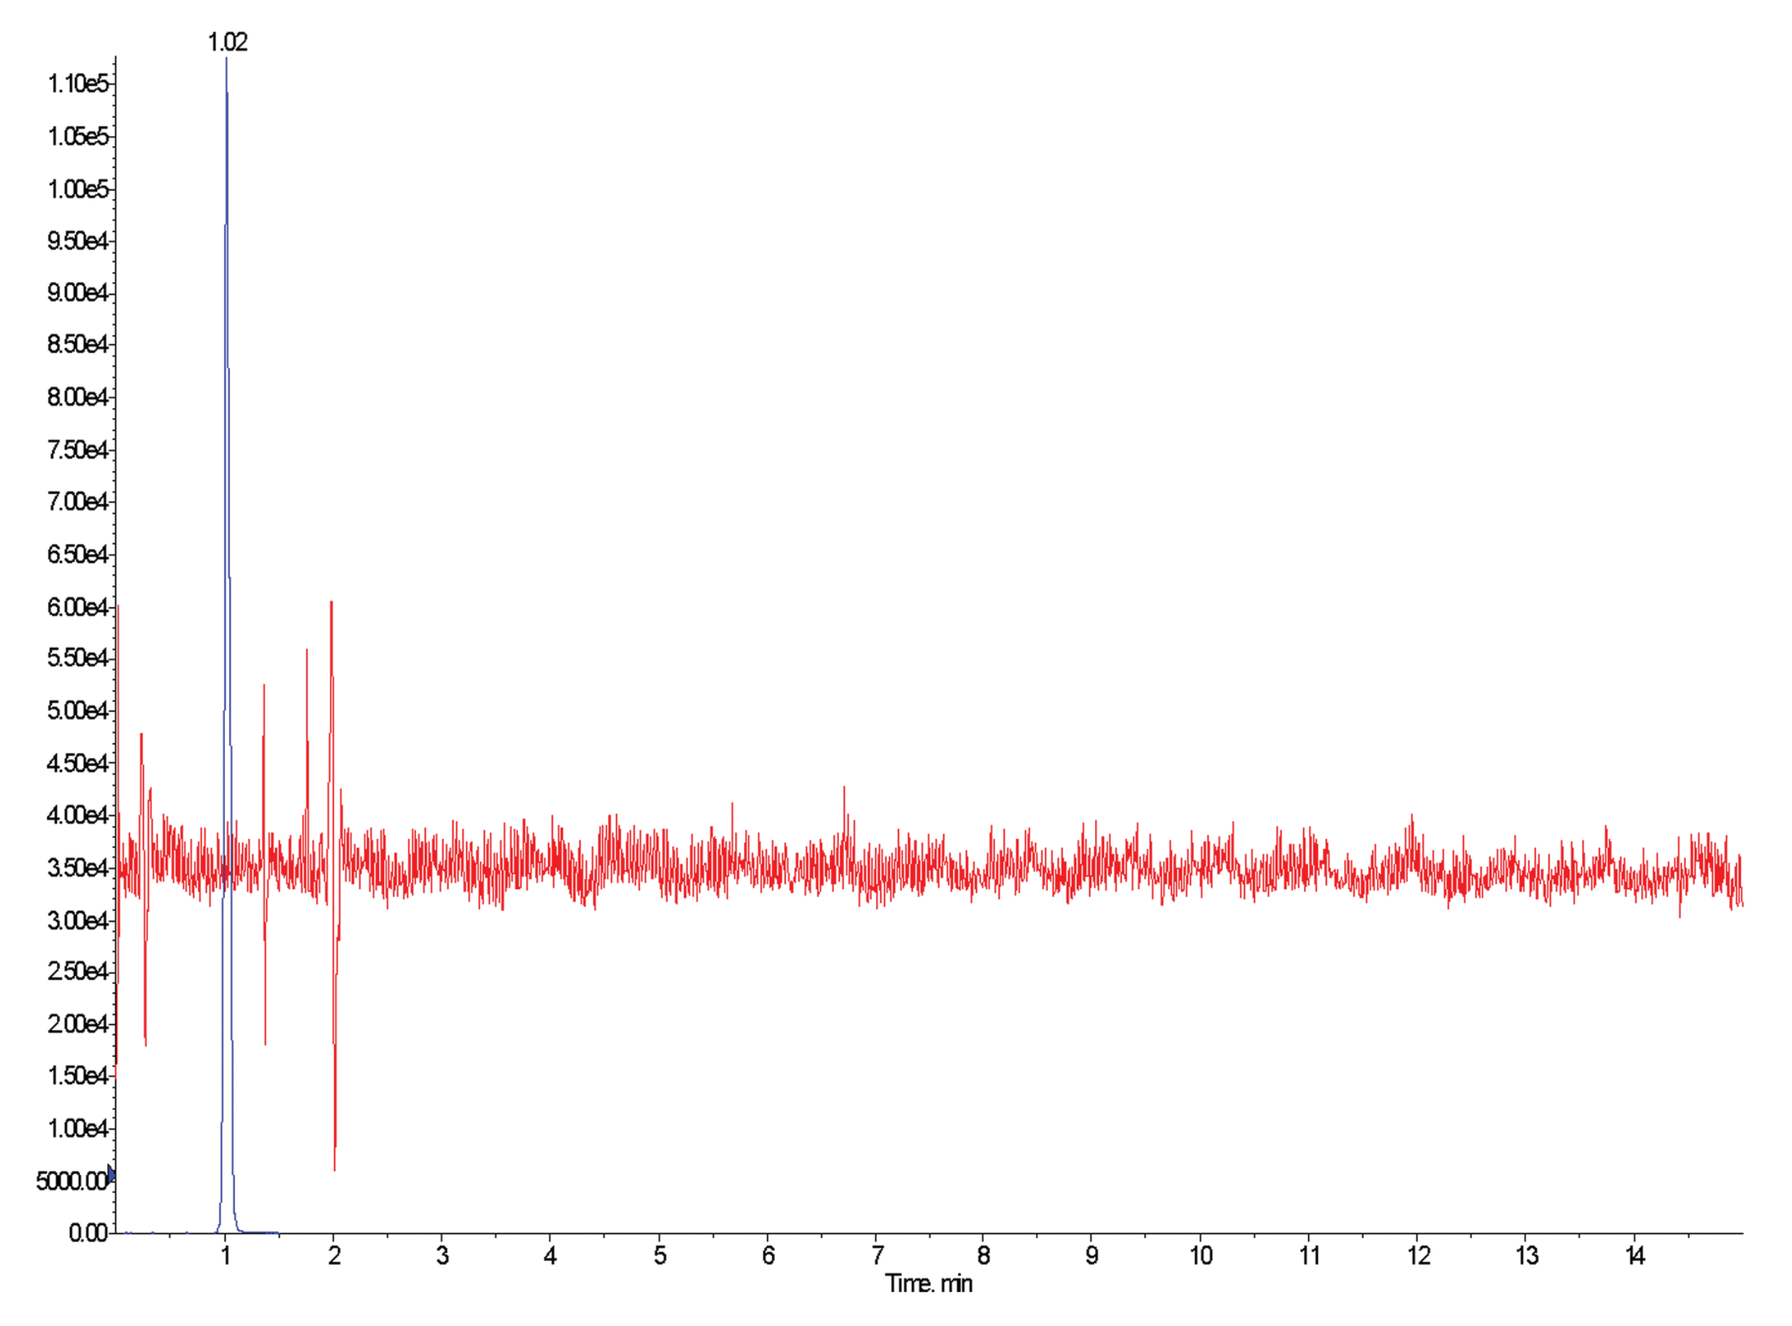
1.02
Time, min

Figure 3. Representative Chromatograms of Voriconazole and d_3-Voriconazole (IS) from an Extracted Blank Human Plasma (EDTA) Sample

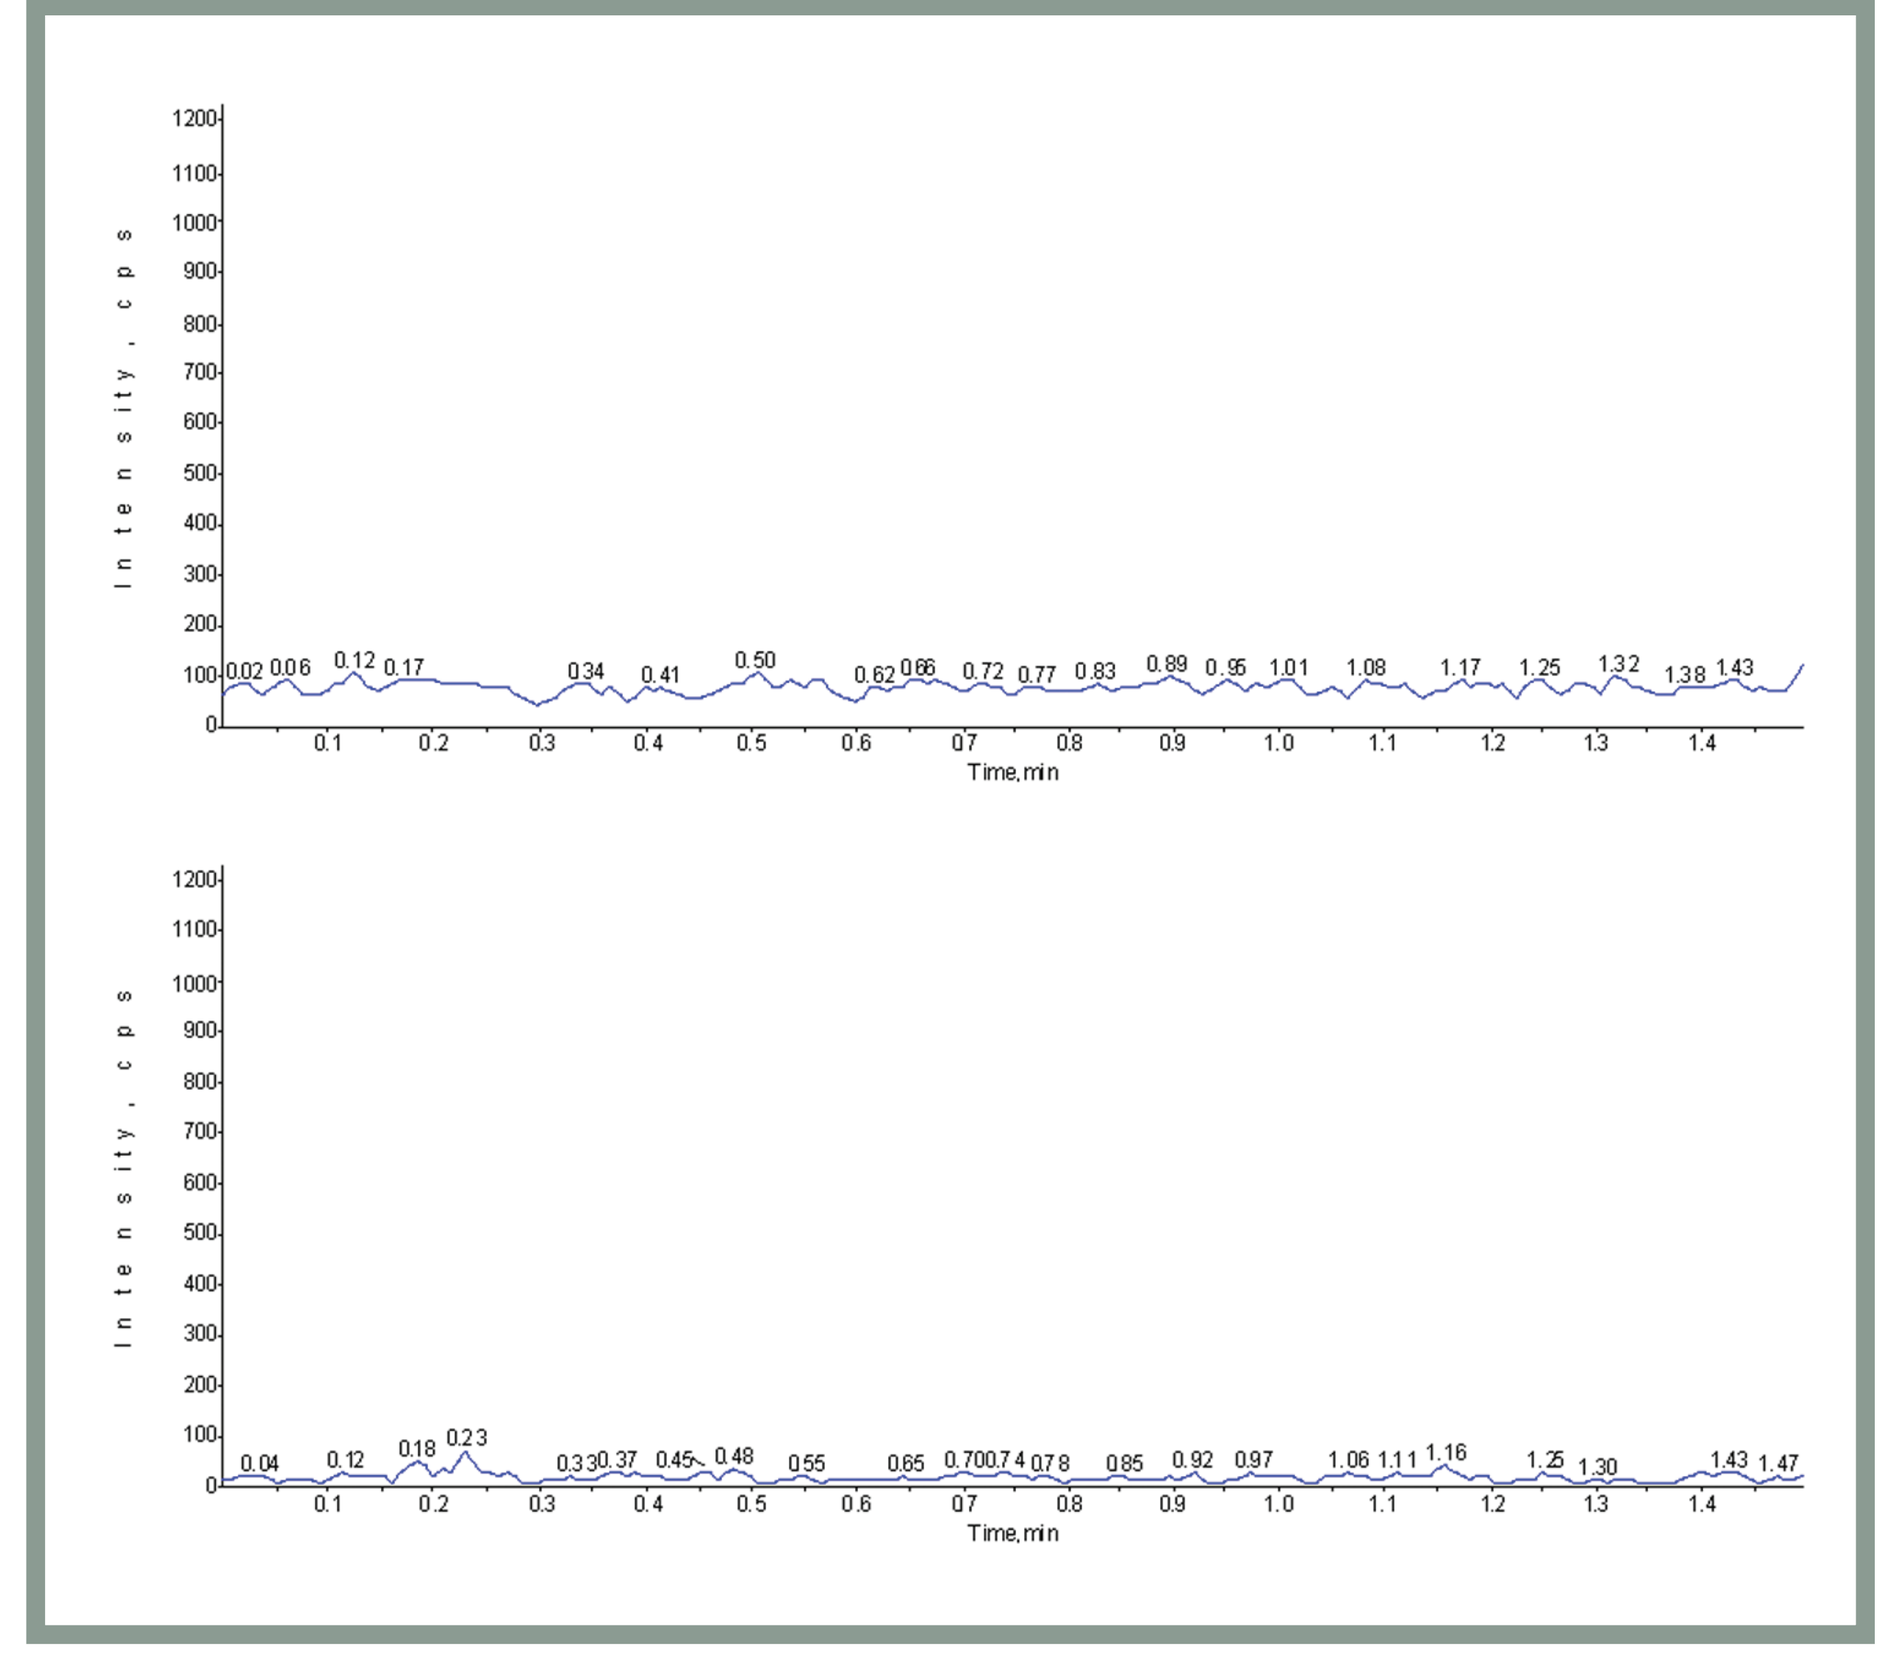
Intensity, cps
Time, min

Figure 4. Representative Chromatograms of Voriconazole and d_3-Voriconazole (IS) from an Extracted Human Plasma (EDTA) Sample Fortified with d_3-Voriconazole (IS) Only

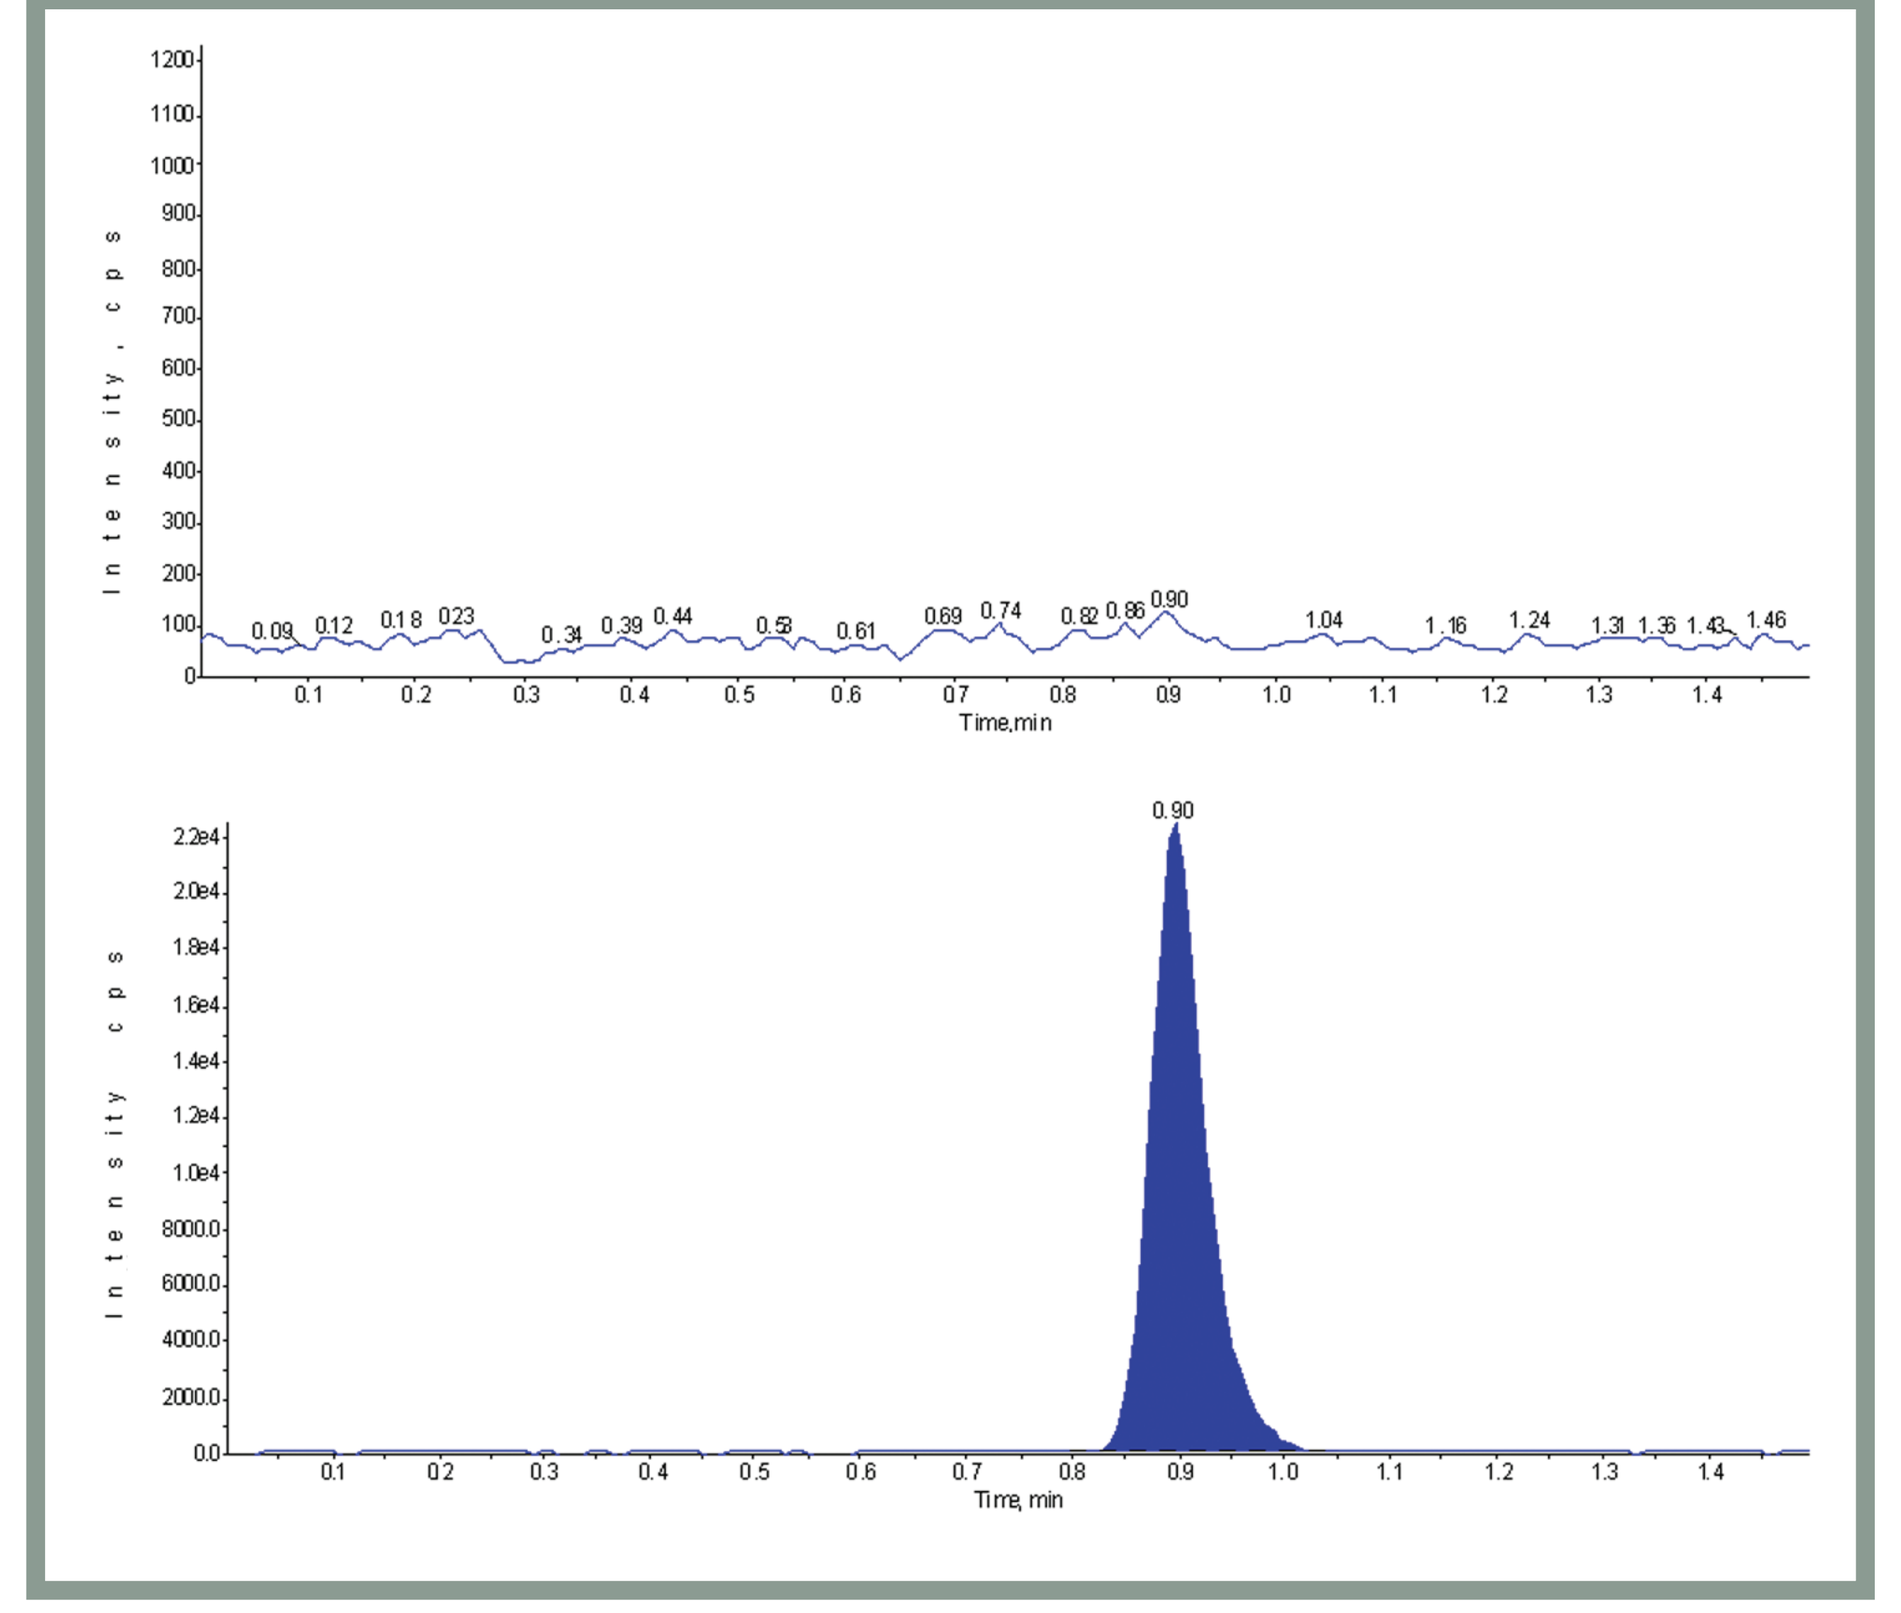
Intensity, cps
Time, min
0.90

Figure 5. Representative Chromatograms of Voriconazole and d_3-Voriconazole (IS) from an Extracted Human Plasma (EDTA) LLOQ Sample

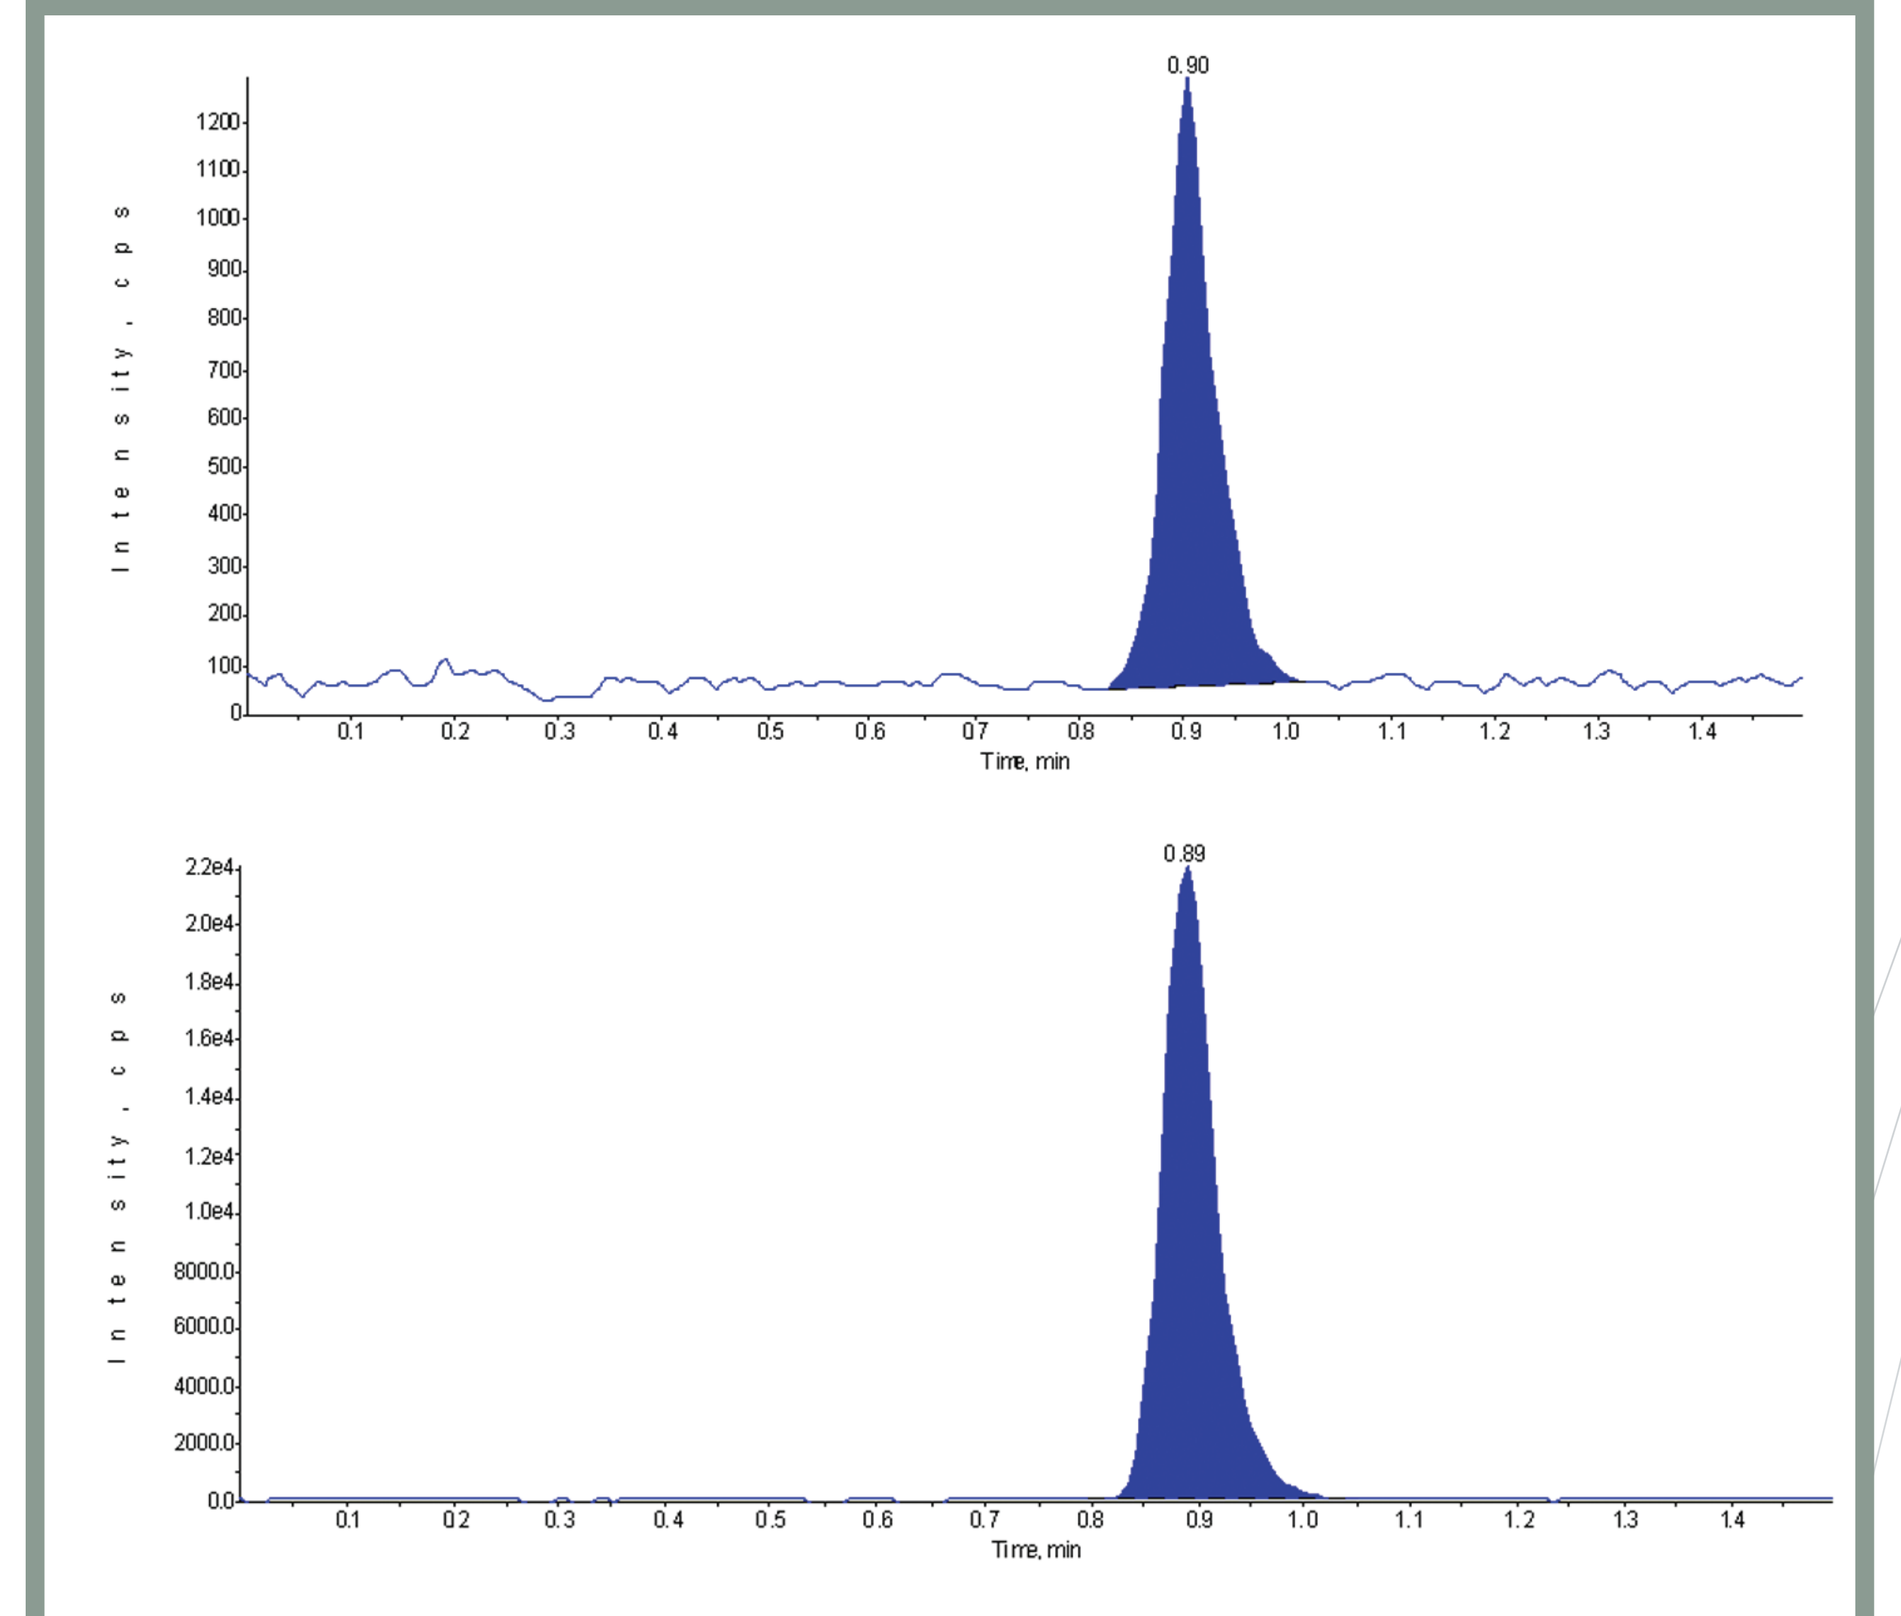
0.90
0.89
Intensity, cps
Time, min

Figure 6. Representative Chromatograms of Voriconazole and d3-Voriconazole (IS) from an Extracted Human Plasma (EDTA) ULOQ Sample

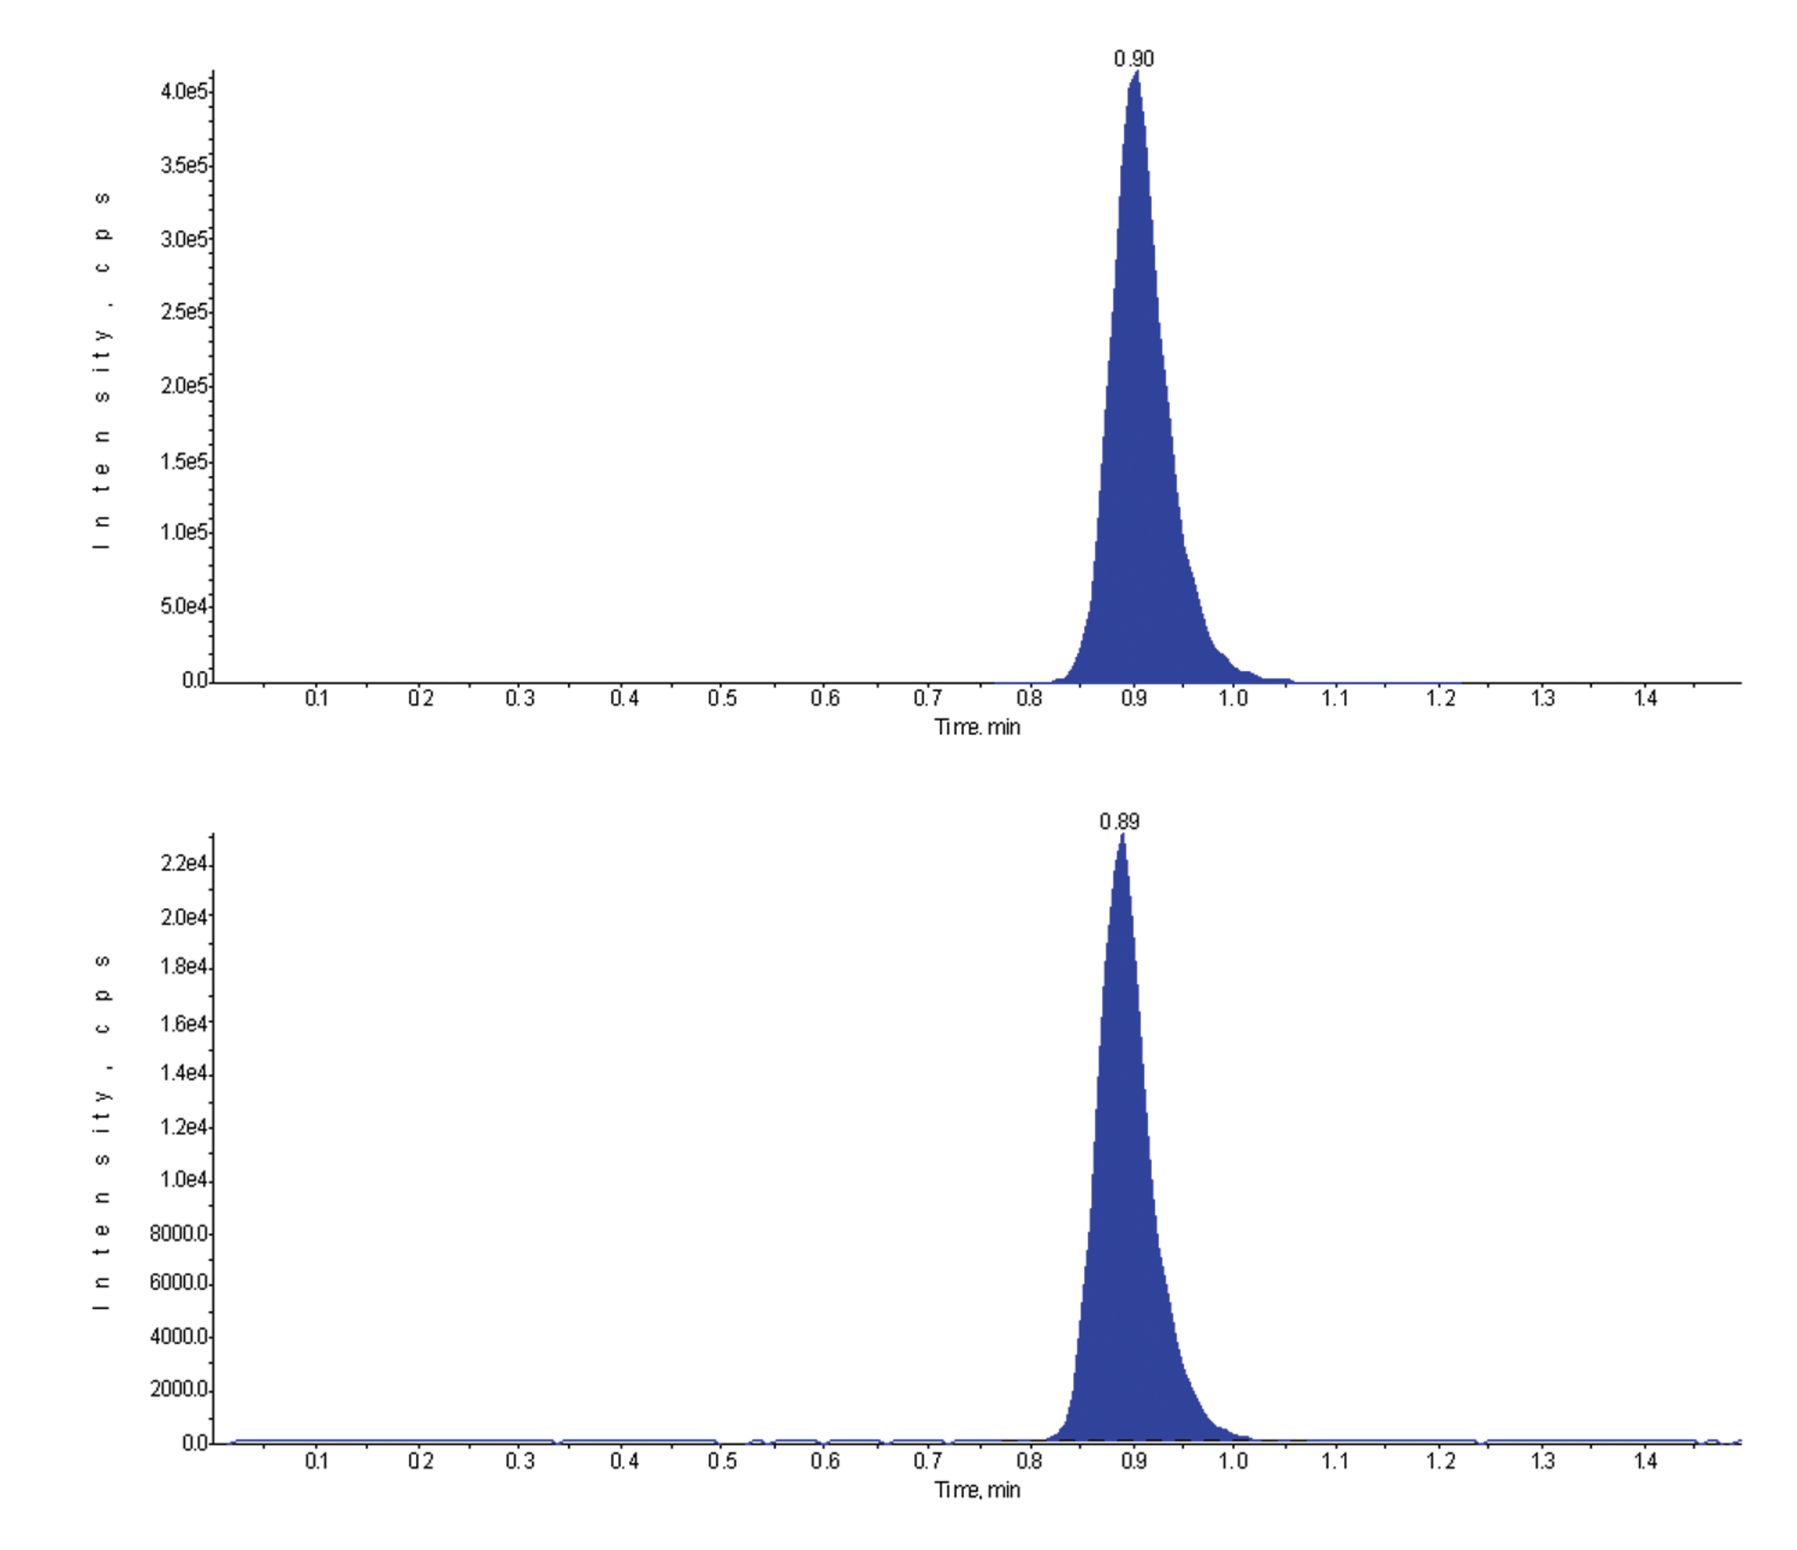
0.90
0.89
Intensity, cps
Time, min

www.celerion.com