A LC-MS/MS Method for the Quantitation of Ibuprofen, Phenylephrine, Chlorpheniramine, and Phenylephrine-Succinate in Rat Plasma (NaF/Na_2 EDTA)

Ginny James, Alan Dzerk, Curtis Sheldon
Celerion, Lincoln, NE

INTRODUCTION:

- Phenylephrine-succinate is a degradation product formed from the reaction between the maleate of chlorpheniramine and phenylephrine. It is found in over-the-counter products containing maleates and phenylephrine.
- The purpose of this project was to develop a single sample processing method that would allow for injection using multiple LC-MS/MS methods, if necessary, for the quantitation of ibuprofen (IBU), phenylephrine (PE), chlorpheniramine (CHL), and phenylephrine-succinate (PES) in rat plasma (NaF/EDTA).

CHALLENGES:

- Differences in the structures, active functional groups, and acid dissociation constants of the target compounds made it impossible to develop a single solid phase or liquid-liquid extraction method for all four compounds from rat plasma.
- Instead, a single protein precipitation method had to be developed to provide “clean” samples that would not cause high back-pressure or blown fittings when injected onto the LC-MS/MS.
- The protein precipitation method also required high recovery for the less sensitive compounds (PE and PES) and the addition of an appropriate dilution solvent for the more sensitive compounds (IBU and CHL).
- Differences in instrument sensitivity and required ion modes (positive vs. negative) made it necessary to develop three LC-MS/MS methods that would separate the compounds from matrix components inherently present in protein precipitated samples.

METHODS AND INSTRUMENTATION:

- Proteins in rat plasma samples (0.05 mL) were precipitated by the addition of dilute trichloroacetic acid.
- After centrifugation, portions of the supernatant were transferred to two separate 96 well plates using a Zymark/Caliper Sciclone automated sample handling system.
- One plate (for PE and PES) was diluted with an acidified aqueous:organic solution.
- The second plate (for IBU and CHL) was diluted with acetonitrile.
- PE and PES were resolved from each other and from other matrix components on a Waters Xterra MS C18 analytical column using trifluoroacetic acid in an aqueous rich mobile phase.
- CHL and IBU were separated from matrix components on ZORBAX 300-SCX and Luna C_8 analytical columns, respectively, using solvent rich mobile phases.
- An AB SCIEX 4000 using an ESI interface detected positive ions (for PE, PES, and CHL) and negative ions (for IBU) in the multiple reaction monitoring mode.
- The acquisition times for PE/PES, CHL, and IBU were 3.0, 1.5, and 1.5 minutes, respectively.

RESULTS:

- A weighted ($1/x^2$) linear regression over the ranges of 0.250-50.0 µg/mL (IBU), 5.00-1000 ng/mL (PE), 1.00-200 ng/mL (CHL), and 2.00-400 ng/mL (PES) was used.
- Inter-batch precision (% CV) and accuracy (% Bias) of IBU (Table 1), PE (Table 2), CHL (Table 3), and PES (Table 4) quality control samples met predefined validation acceptance criteria.
- Assay selectivity was demonstrated by quantitation of six separate rat plasma lots fortified near the LLOQ and at the high quality control concentrations for all compounds. No significant matrix effect was observed (Tables 5-6).
- A post-column infusion demonstrated that IBU, PE, CHL, PES and their respective internal standards did not co-elute with any areas of significant suppression or enhancement.
- Samples having a concentration above the upper limit of the calibration standard ranges were diluted with blank control matrix, and results show that samples with concentrations up to 100 µg/mL, 5000 ng/mL, 1000 ng/mL, and 2000 ng/mL for IBU, PE, CHL, PES, respectively, could be quantified after the application of an appropriate dilution factor.
- The average extraction recoveries of IBU, PE, CHL, and PES were 48%, 83%, 93%, and 106%, respectively. The average extraction recoveries of d_3-IBU, d_3-PE, and d_6-CHL were 43%, 83%, and 95%, respectively.
- Stability evaluations of IBU, PE, CHL, and PES (Table 7) quality control samples met predefined validation acceptance criteria.
- Method robustness was demonstrated with multiple column lots, mass spectrometers, and extraction scientists.
- The validated batch size was 96 samples.

Figure 1. IBU Structure

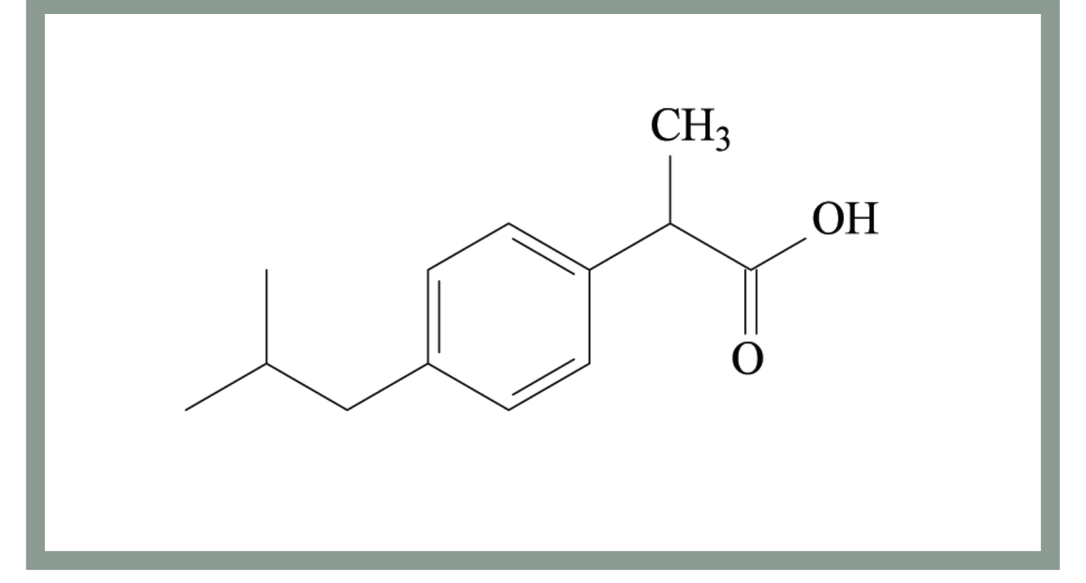
CH3
OH
O

Figure 2. PE Structure

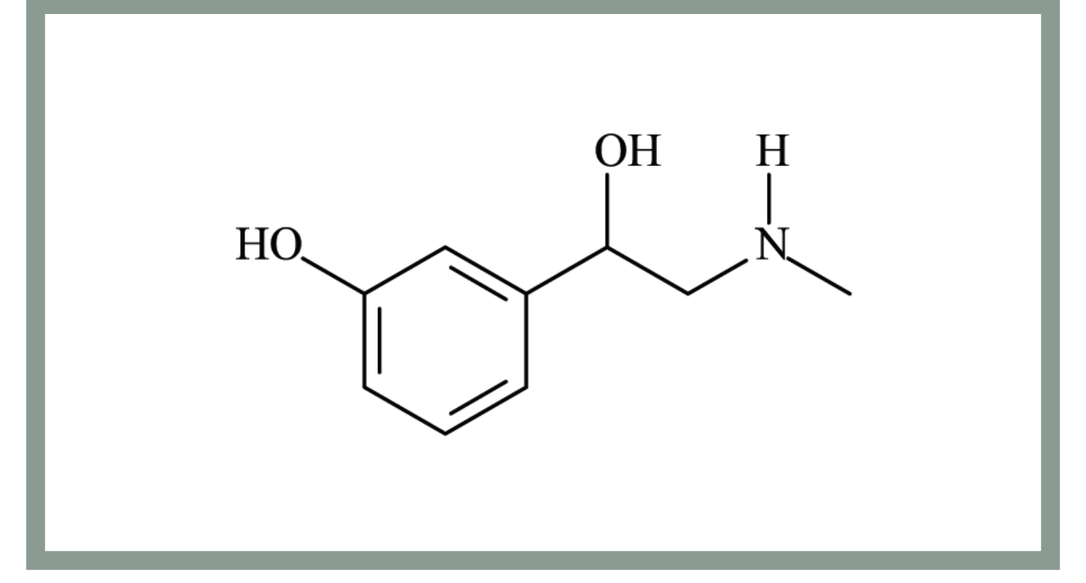
OH
H
HO
N

Figure 3. CHL Structure

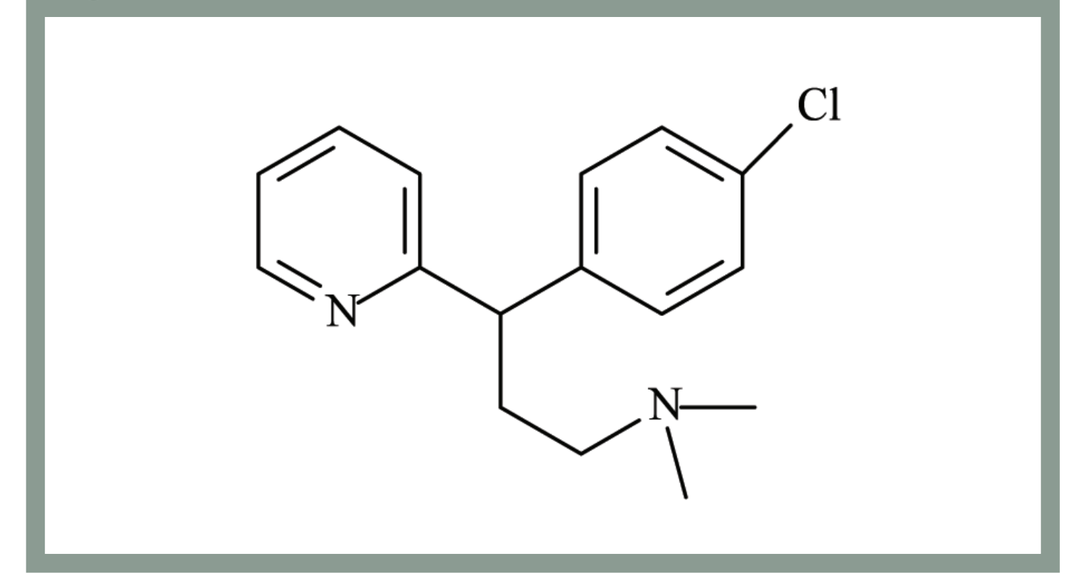
Cl
N
N

Figure 4. PES Structure

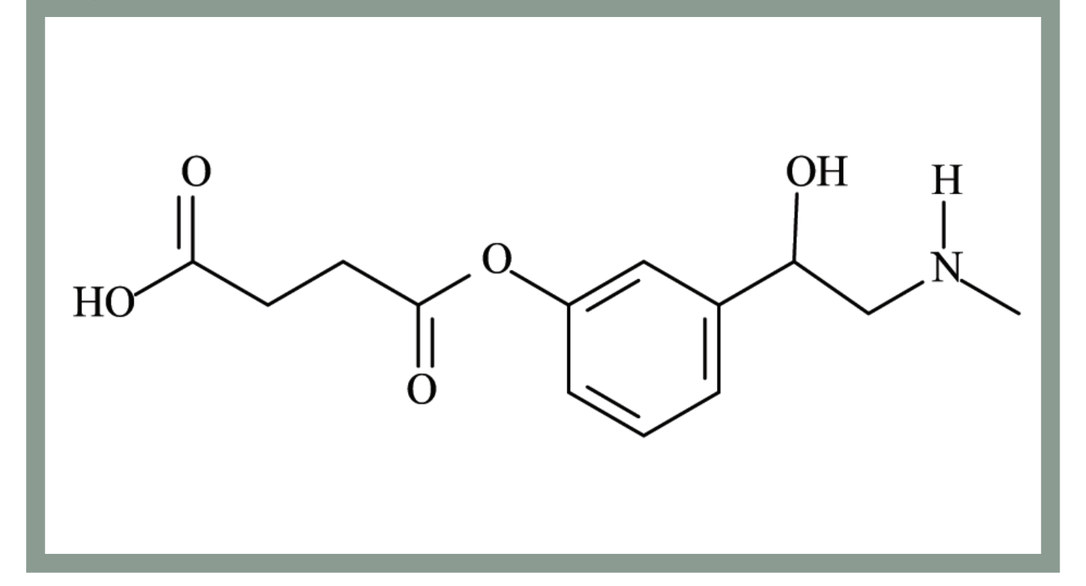
O
OH
H
HO
O
N
O

Table 1. IBU Inter-Batch Precision and Accuracy

	LLOQ QC 0.250 µg/mL	QC A 0.750 µg/mL	QC B 3.75 µg/mL	QC C 37.5 µg/mL
Inter-Batch Mean	0.258	0.780	3.95	35.5
Inter-Batch % CV	6.2	4.6	4.3	5.9
Inter-Batch % Bias	3.2	4.0	5.3	-5.3
n	18	18	18	18

Table 2. PE Inter-Batch Precision and Accuracy

	LLOQ QC 5.00 ng/mL	QC A 15.0 ng/mL	QC B 75.0 ng/mL	QC C 750 ng/mL
Inter-Batch Mean	5.47	15.2	74.6	704
Inter-Batch % CV	12.4	5.6	6.5	4.3
Inter-Batch % Bias	9.4	1.3	-0.5	-6.1
n	18	18	18	18

Table 3. CHL Inter-Batch Precision and Accuracy

	LLOQ QC 1.00 ng/mL	QC A 3.00 ng/mL	QC B 15.0 ng/mL	QC C 150 ng/mL
Inter-Batch Mean	0.927	2.88	15.1	147
Inter-Batch % CV	5.5	3.4	4.6	2.8
Inter-Batch % Bias	-7.3	-4.0	0.7	-2.0
n	18	18	18	18

Table 4. PES Inter-Batch Precision and Accuracy

	LLOQ QC 2.00 ng/mL	QC A 6.00 ng/mL	QC B 30.0 ng/mL	QC C 300 ng/mL
Inter-Batch Mean	2.03	6.02	31.2	308
Inter-Batch % CV	11.5	8.3	5.5	6.1
Inter-Batch % Bias	1.5	0.3	4.0	2.7
n	18	18	18	18

Table 5. IBU and CHL Matrix Effect Test

	IBU				CHL			
	LLOQ		High		LLOQ		High	
Lot#	0.250 µg/mL	% Dev.	37.5 µg/mL	% Dev.	1.00 ng/mL	% Dev.	150 ng/mL	% Dev.
1	0.242	-3.2	31.9	-14.9	1.00	+0.0	142	-5.3
2	0.262	+4.8	35.0	-6.7	0.988	-1.2	143	-4.7
3	0.244	-2.4	34.8	-7.2	1.02	+2.0	145	-3.3
4	0.240	-4.0	33.0	-12.0	1.11	+11.0	146	-2.7
5	0.249	-0.4	31.0	-17.3	0.983	-1.7	139	-7.3
6	0.227	-9.2	33.2	-11.5	1.05	+5.0	145	-3.3
Mean	0.244		33.2		1.03		143	
% CV	4.7		4.7		4.7		1.8	
% Theoretical	97.6		88.5		103.0		95.3	
n	6		6		6		6	

Table 6. PE and PES Matrix Effect Test

	PE				PES			
	LLOQ		High		LLOQ		High	
Lot#	5.00 ng/mL	% Dev.	750 ng/mL	% Dev.	2.00 ng/mL	% Dev.	300 ng/mL	% Dev.
1	6.69	+33.8	663	-11.6	2.06	+3.0	293	-2.3
2	5.25	+5.0	682	-9.1	1.94	-3.0	272	-9.3
3	5.46	+9.2	695	-7.3	2.28	+14.0	312	+4.0
4	5.53	+10.6	658	-12.3	2.02	+1.0	294	-2.0
5	5.48	+9.6	692	-7.7	2.22	+11.0	288	-4.0
6	5.25	+5.0	656	-12.5	2.24	+12.0	302	+0.7
Mean	5.61		674		2.13		294	
% CV	9.7		2.6		6.5		4.6	
% Theoretical	112.2		89.9		106.5		98.0	
n	6		6		6		6	

Table 7. Stability Evaluations

Information Requested	Data
Bench-Top Stability (Short-term Stability) at Ambient Temperature Under UV-Shielded Light	IBU, CHL, PES: 7 hours PE: 2 hours
Bench-Top Stability (Short-term Stability) in an Ice Water Bath Under UV-Shielded Light	IBU: 19 hours PE, CHL, PES: 5 hours
Processed Stability (Post-Preparative Stability) in a Polypropylene 96 Well Plate at 5°C	IBU: 106 hours PE: 105 hours CHL, PES: 126 hours
Processed Stability (Processed Sample Integrity) in a Polypropylene 96 Well Plate at 5°C	IBU: 101 hours PE, PES: 157 hours CHL: 107
Freeze-Thaw Stability in Polypropylene Tubes Under UV-Shielded Light	IBU, PE, CHL, PES: 4 freeze (-80°C) -thaw (ambient temperature) cycles
Long-Term Storage Stability in Polypropylene Tubes at -80°C	IBU, PE, CHL, PES: 110 days

Figure 5. IBU / d_3-IBU Extracted Blank Sample (Top) and Extracted LLOQ Sample (Bottom)

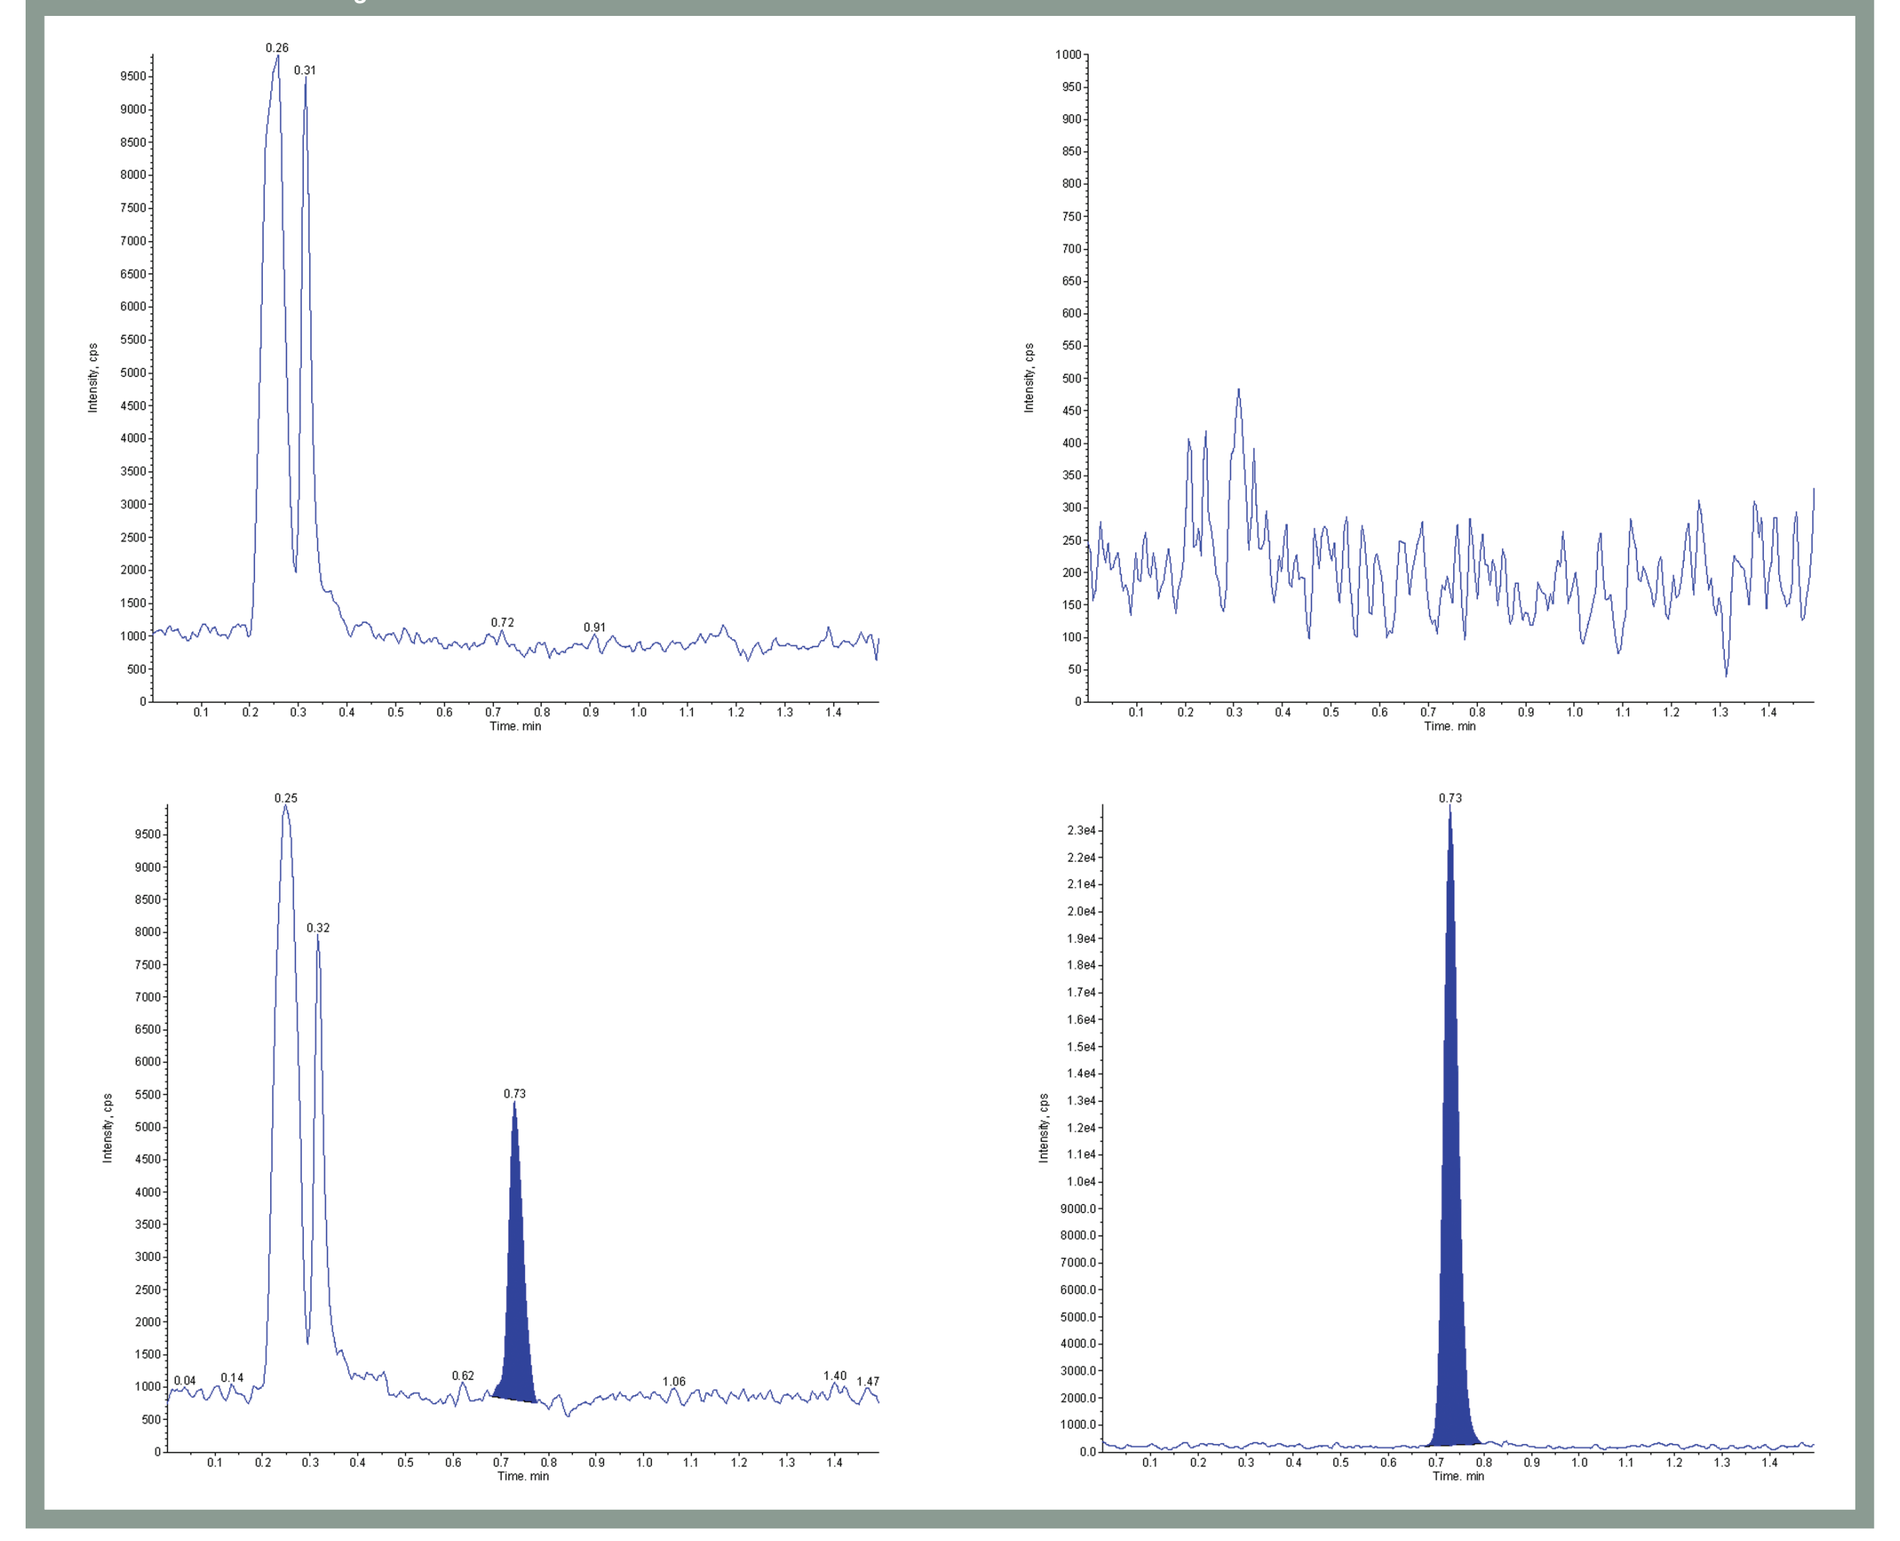

Figure 6. PE / d_3-PE Extracted Blank Sample (Top) and Extracted LLOQ Sample (Bottom)

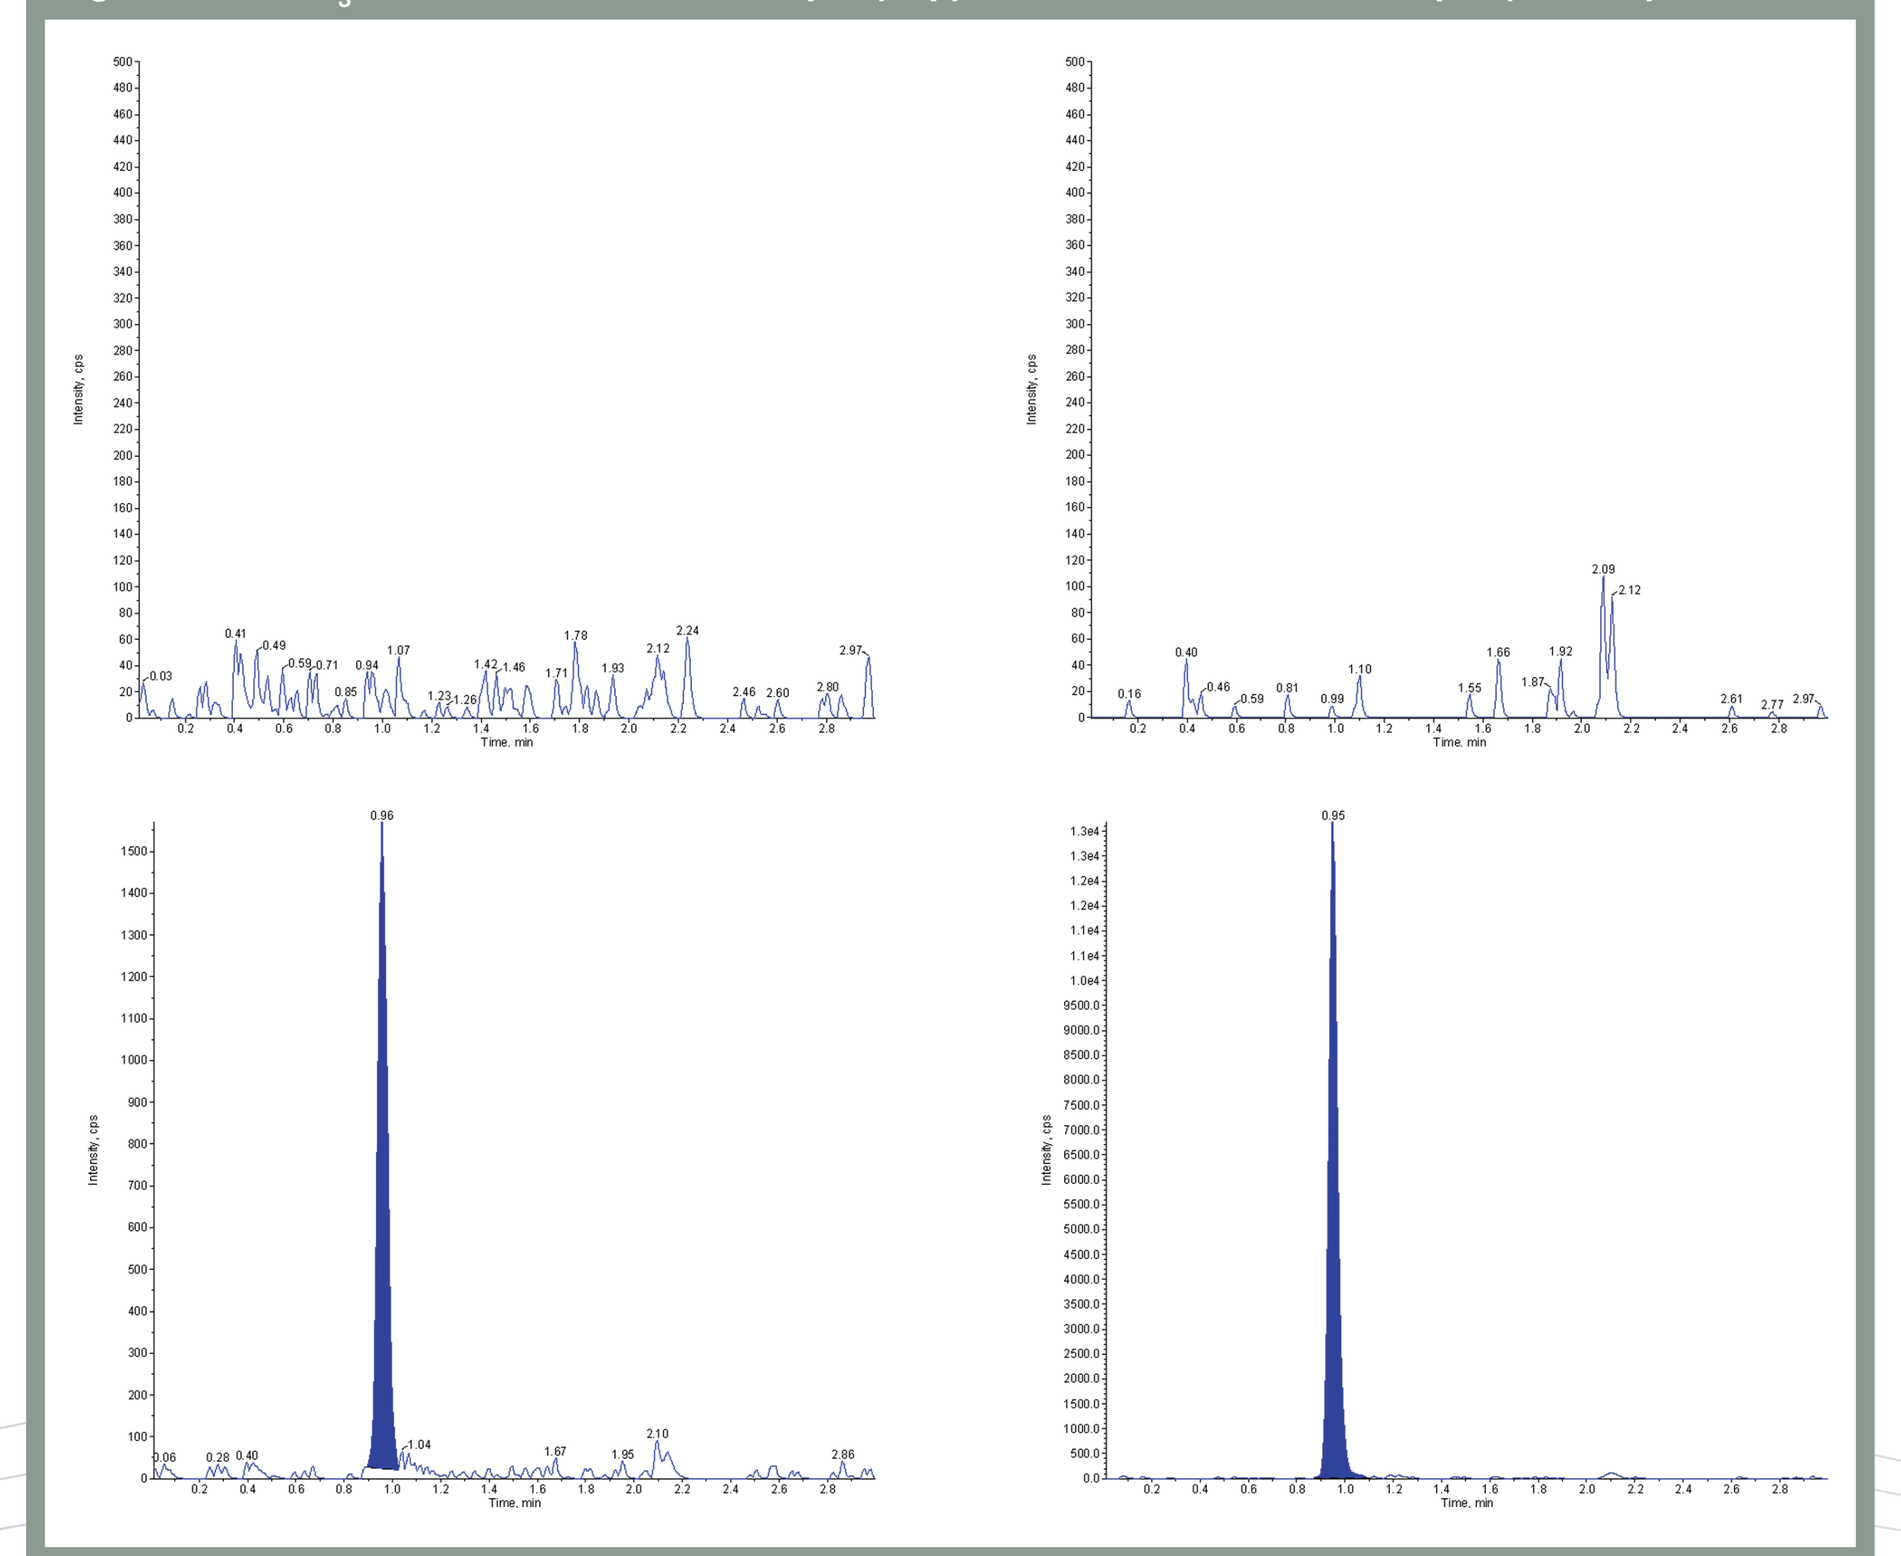

Figure 7. CHL / d_6-CHL Extracted Blank Sample (Top) and Extracted LLOQ Sample (Bottom)

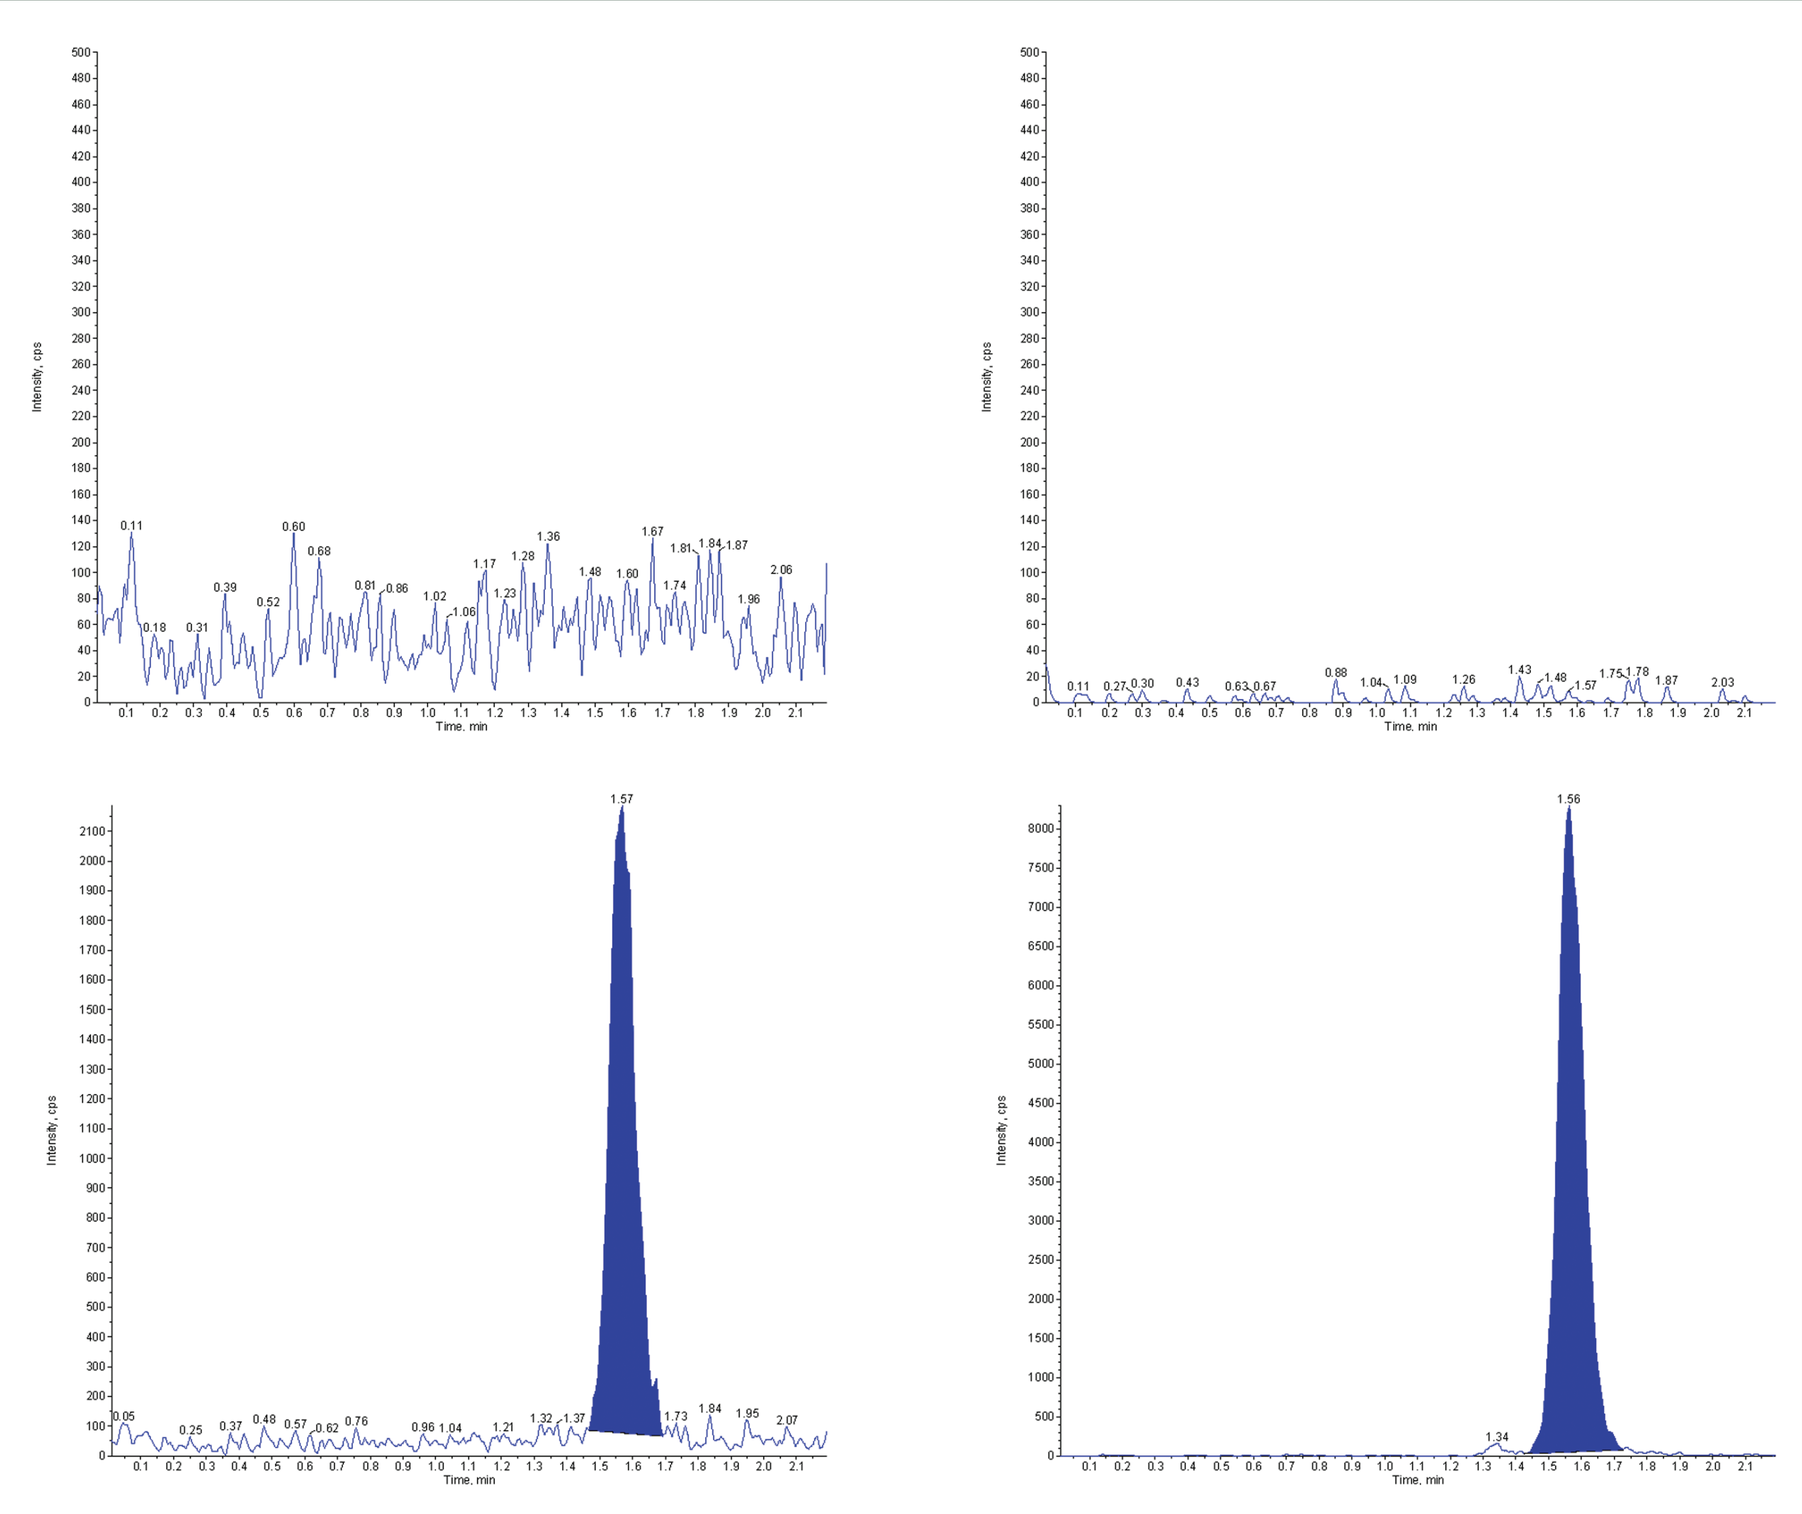

Figure 8. PES / d_3-PE Extracted Blank Sample (Top) and Extracted LLOQ Sample (Bottom)

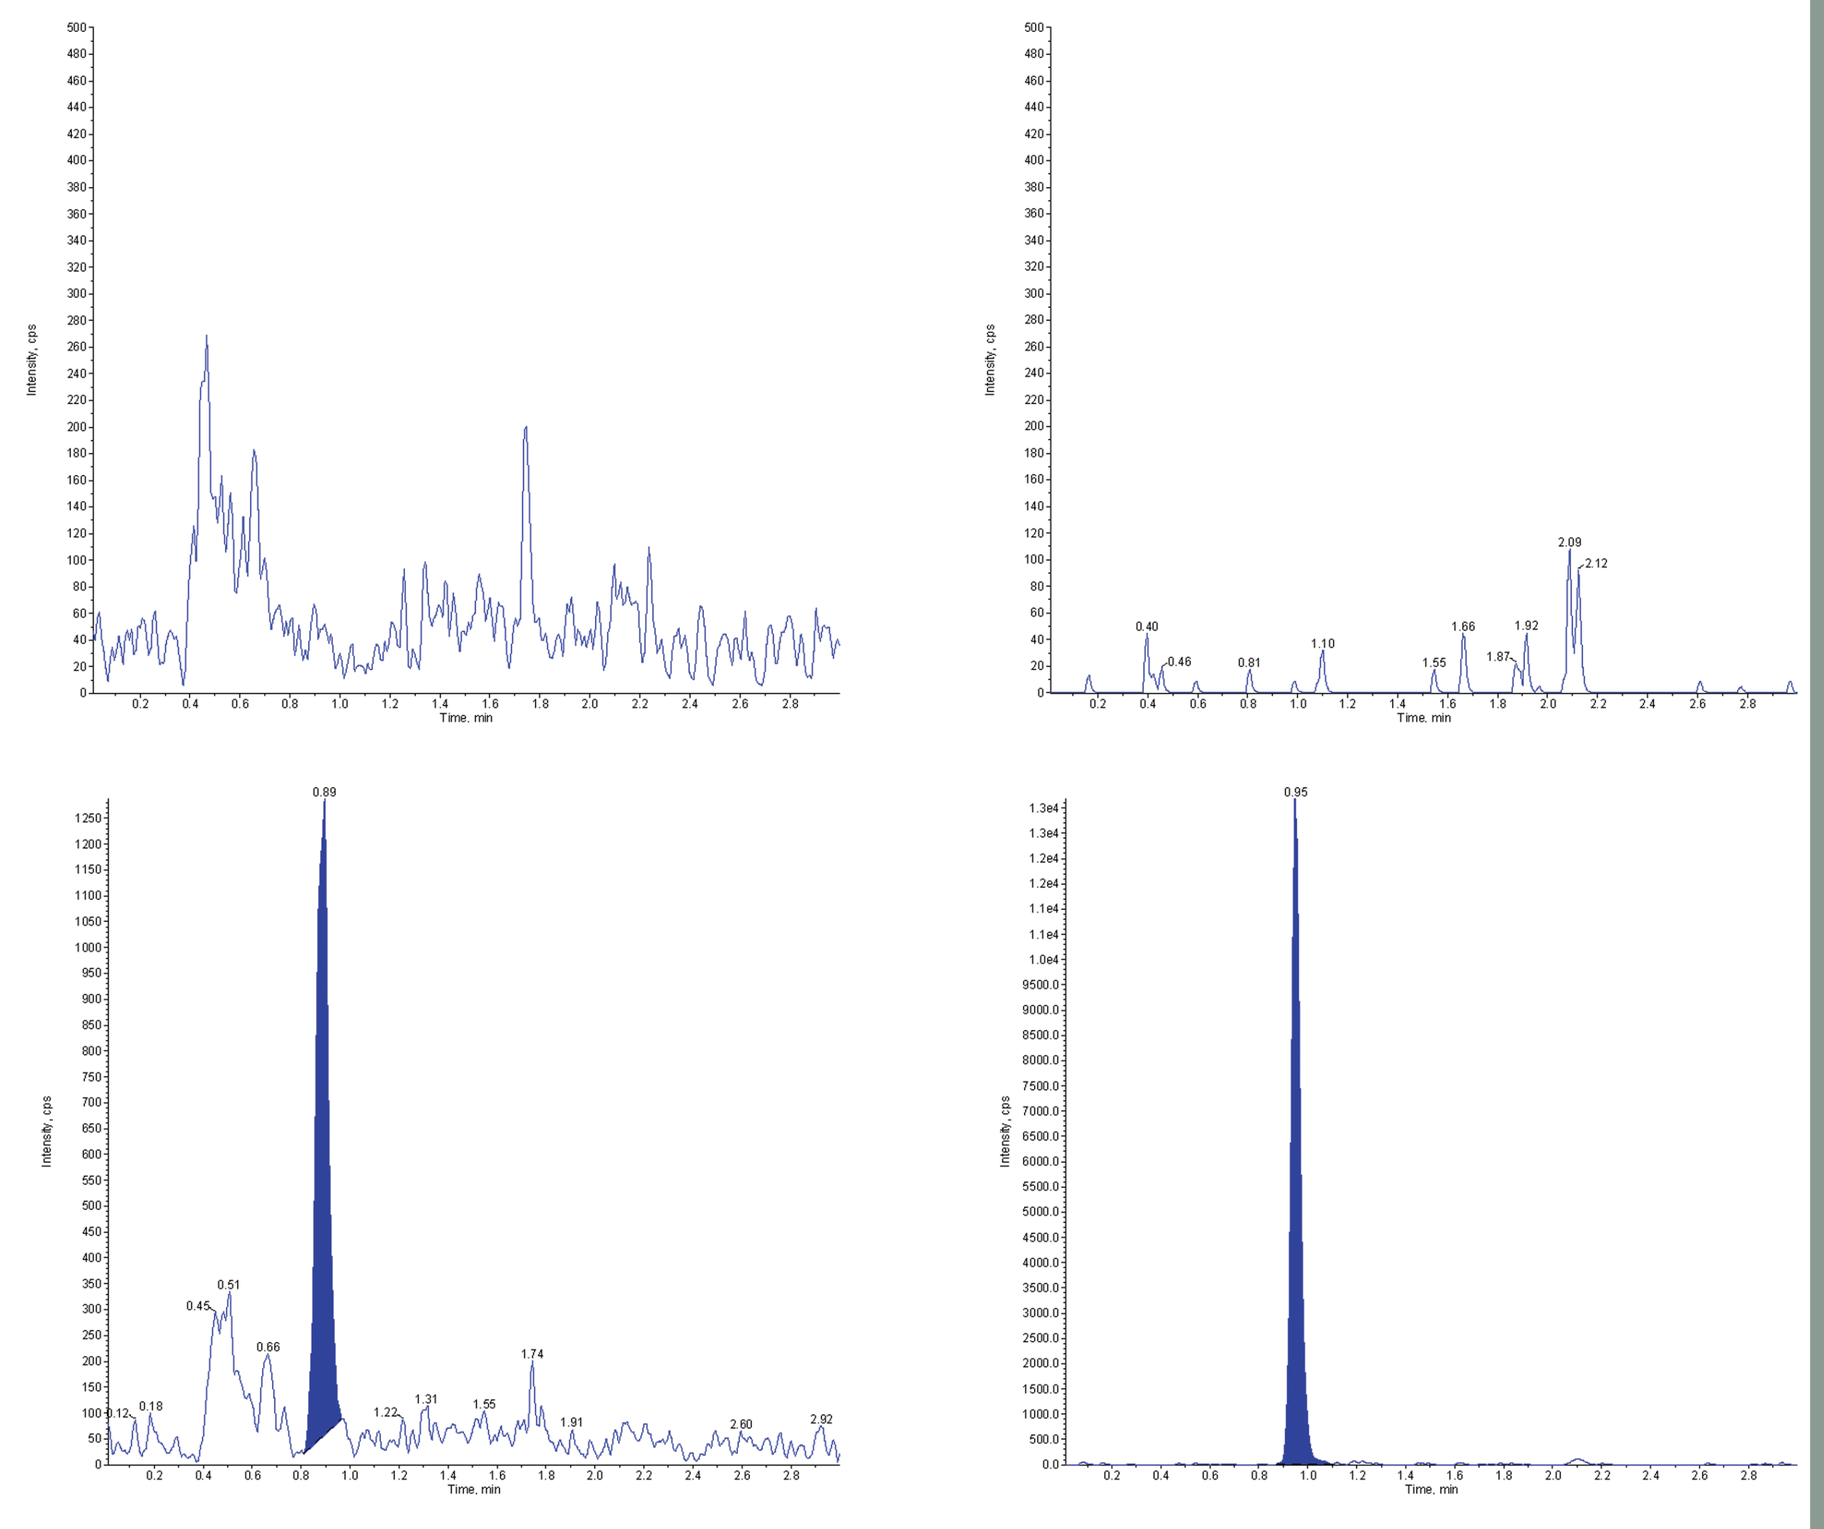

CONCLUSIONS:

- The validated method utilized a single sample processing method and three LC-MS/MS methods to appropriately quantitate all four compounds and eliminate significant matrix interference.
- The bioanalytical assay for the quantitation of ibuprofen, phenylephrine, chlorpheniramine, and phenylephrine-succinate in rat plasma (NaF/EDTA) met acceptance criteria for precision, accuracy, sensitivity, selectivity, and stability.
- The validation results demonstrate that a precise, accurate, sensitive, selective, rugged and reproducible assay was developed.

www.celerion.com