Nº T2366

A RANDOMIZED, OPEN-LABEL, SINGLE-DOSE, CROSSOVER STUDY TO ASSESS THE EFFECTS OF ATORVASTATIN OR VALSARTAN CO-ADMINISTRATION ON THE PHARMACOKINETICS OF NALTREXONE HCL AND BUPROPION HCL SUSTAINED-RELEASE TABLETS IN HEALTHY ADULT SUBJECTS

Shawn Flanagan, PhD[1], Rahma Demnati, MD, PhD[2], Yuli Xie, MS[3], Georgia Tsaroucha MSPH[1], Erika Eichmann, PhD[4], Renae Chavira, BS, MA[3], Susan E. Hazel, BA[1], Sandra M. Connolly, MD[3], Eduardo Dunayevich, MD[1]

[1]Orexigen, La Jolla, CA, [2]Celerion, Montreal, QC, [3]Celerion, Lincoln, NE, [4] Tandem Labs, Salt Lake City, UT

OBJECTIVES

- To assess the effects of a single oral dose of 80 mg atorvastatin or 320 mg valsartan on the pharmacokinetics (PK) of naltrexone (N) and bupropion (B), given as a extended-release (SR) combination of NB (two 8/90 mg tablets) in healthy subjects.
- To compare the single dose PK of atorvastatin and valsartan (given in combination with NB) to previously published or reported results of these individual agents when given as monotherapy.
- To assess the safety and tolerability of the 3 evaluated treatments.

METHODS

Trial

- This was a single-center, randomized, open label, cross-over study.

Subject Population

- 20 healthy obese and non-obese subjects (16 males and 4 females), between 20-56 years of age with a body mass index (BMI) of 22 to 39 kg/m^2 (inclusive) participated in the study. All 20 subjects were included in the safety assessments and 19 subjects were included in the PK analysis (one subject was excluded for noncompliance).

Study design

- Subjects received a single oral dose of each treatment on Day 1 of each treatment period according to the randomization scheme, with a minimum 14-day washout period between dosing days.
 - Treatment A: Two naltrexone SR 8 mg/bupropion SR 90 mg tablets (NB; Reference).
 - Treatment B: Two NB tablets co-administered with 1 atorvastatin 80 mg tablet (Test 1).
 - Treatment C: Two NB tablets co-administered with 1 valsartan 320 mg tablet (Test 2)
- PK samples were collected over 120-hour postdose interval. Plasma samples were assayed using validated liquid chromatography tandem mass spectrometry methods. PK parameters were calculated from the individual plasma concentrations using noncompartmental methods.
- Safety monitoring included adverse events (AEs), vital signs, physical examination (including neurological assessment), clinical laboratory tests and ECG.

Analytical Assessments

Table 1: Validated Tandem Mass Spectrometry (LC-MS/MS) with Electrospray Ionization and Selected Reaction Monitoring Methods Used for Quantitation of Drugs in K_2-EDTA Human Plasma Specimens from Orexigen Study NB-232

Assay	Naltrexone	Bupropion	Valsartan	Atorvastatin
Analytes	naltrexone (N) 6ß-naltrexol (6ß)	bupropion hydroxybupropion threohydrobupropion erythrohydrobupropion	valsartan	atorvastatin p-hydroxyatorvastatin o-hydroxyatorvastatin
Ion Polarity	positive	positive	negative	positive
Extraction	liquid-liquid	solid phase	protein precipitation	supported liquid
Range (ng/mL)	0.0200-2.00 (N) 0.500-500 (6ß)	1.00-1000	50.0-10,000	0.300-50.0
Precision and Accuracy (only parent is shown)				
Intra-Day				
Precision	1.7 to 13.8%	1.8 to 6.7%	2.0 to 6.4%	1.0 to 7.3%
Accuracy	-5.6 to 10.5%	-14.8 to 1.0%	-6.5 to 2.0%	-8.3 to 4.0%
Inter Day				
Precision	0.5 to 5.0%	2.9 to 8.4%	0.0 to 2.8%	0.0 to 3.7%
Accuracy	-2.0 to 6.5%	-7.3 to -4.3%	-3.6 to -0.2%	-4.8 to -0.8%
Interference	**No interference from:** bupropion hydroxybupropion threohydrobupropion erythrohydrobupropion atorvastatin p-hydroxyatorvastatin o-hydroxyatorvastatin valsartan	**No interference from:** naltrexone 6ß-naltrexol atorvastatin p-hydroxyatorvastatin o-hydroxyatorvastatin valsartan	**No interference from:** bupropion hydroxybupropion threohydrobupropion erythrohydrobupropion naltrexone 6ß-naltrexol	**No interference from:** bupropion hydroxybupropion threohydrobupropion erythrohydrobupropion naltrexone 6ß-naltrexol
Stability	All samples were assayed within reported benchtop, freeze-thaw and long term storage stability			

RESULTS

Pharmacokinetic Results

Figure 1: Mean Plasma Naltrexone Concentration-Time Profiles (Linear Plot)

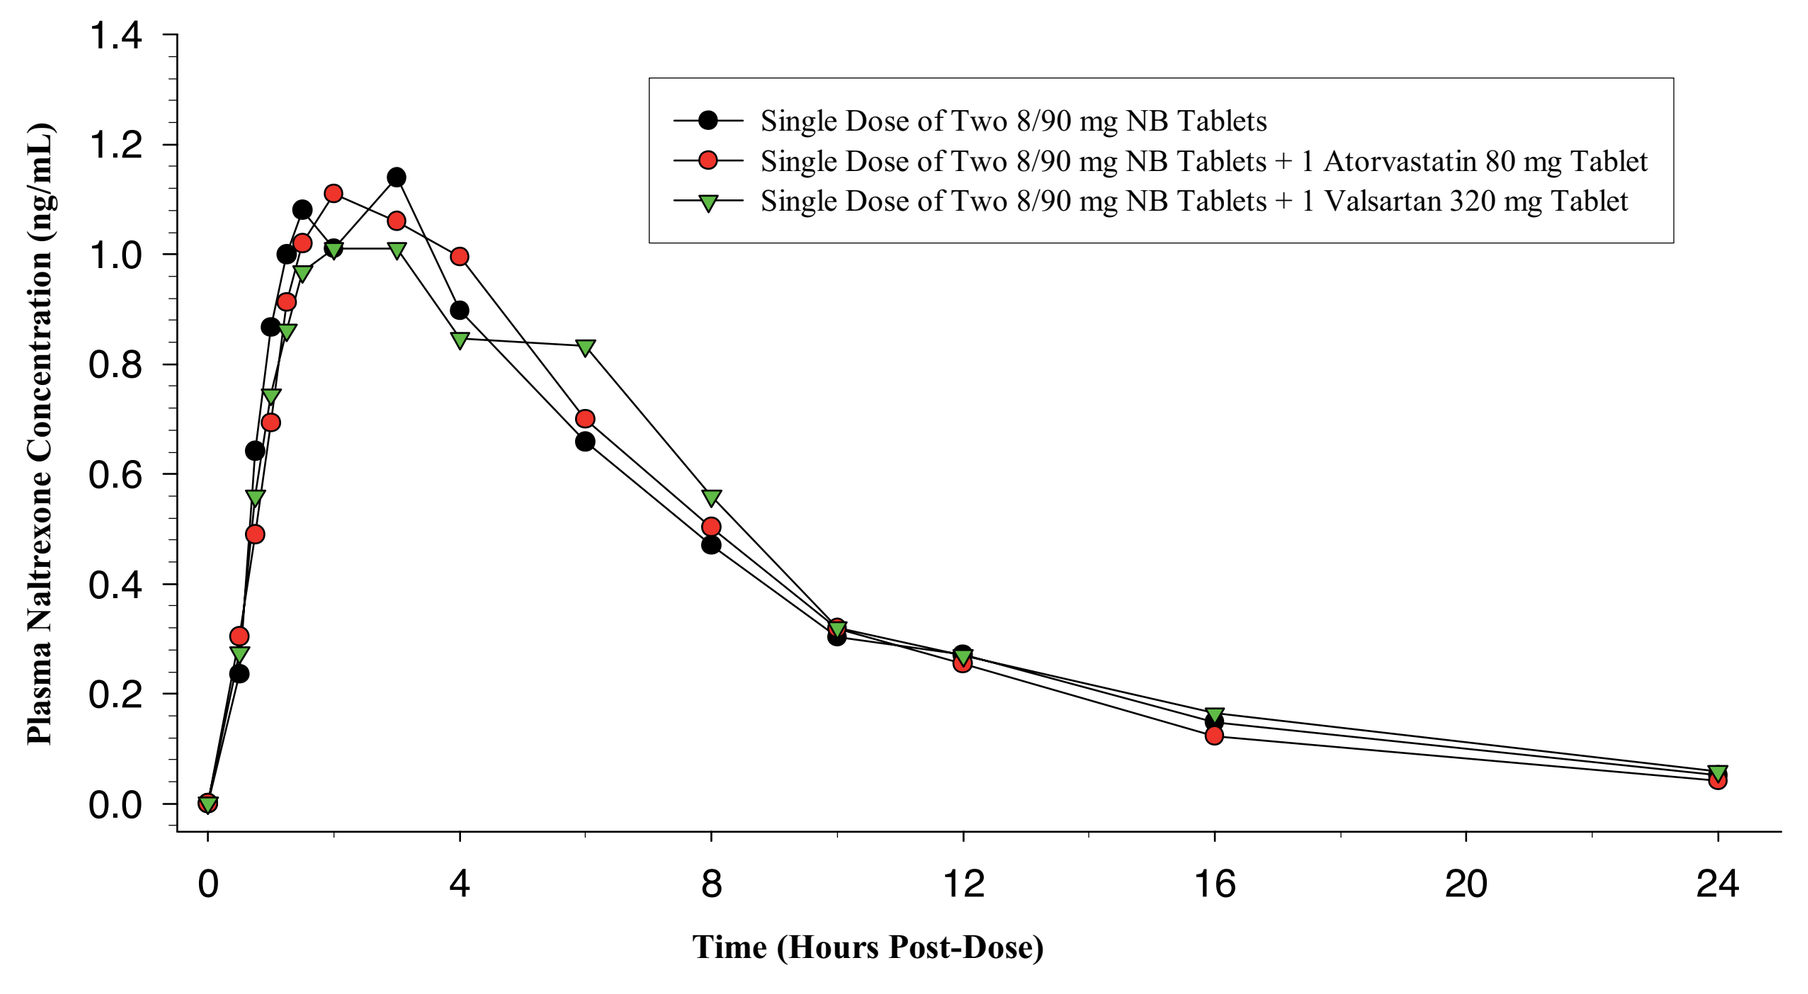
Plasma Naltrexone Concentration (ng/mL)
Time (Hours Post-Dose)
Single Dose of Two 8/90 mg NB Tablets
Single Dose of Two 8/90 mg NB Tablets + 1 Atorvastatin 80 mg Tablet
Single Dose of Two 8/90 mg NB Tablets + 1 Valsartan 320 mg Tablet

NB: naltrexone and bupropion combination

Table 2: Summary of PK Parameters and Statistical Analyses of Naltrexone and its Active Metabolite 6-Beta-Naltrexol for NB + Atorvastatin Versus NB Alone

Analyte	PK Parameter	NB Alone	NB + Atorvastatin	%GMR (90% CI)
Naltrexone	AUC_{0-t} (ng•h/mL)	9.62 ± 5.66	9.25 ± 5.13	98.22 (88.09 - 109.52)
	$AUC_{0-\infty}$ (ng•h/mL)	10.55 ± 5.76	9.46 ± 5.13	94.74 (85.32 - 105.19)
	C_{max} (ng/mL)	1.37 ± 0.943	1.34 ± 0.704	104.83 (90.85 - 120.97)
	T_{max} (h)	1.76 (0.75, 6.00)	2.00 (1.00, 5.99)	–
	$t_{1/2}$ (h)	4.94 ± 1.60	4.05 ± 1.01	–
6-Beta Naltrexol	AUC_{0-t} (ng•h/mL)	243.27 ± 55.81	259.11 ± 55.52	106.66 (102.38 - 111.12)
	$AUC_{0-\infty}$ (ng•h/mL)	257.85 ± 57.52	274.31 ± 55.31	106.68 (103.18 - 110.30)
	C_{max} (ng/mL)	15.8 ± 4.46	16.6 ± 4.06	105.64 (96.69 - 115.42)
	T_{max} (h)	2.50 (0.75, 6.00)	2.00 (1.00, 5.99)	–
	$t_{1/2}$ (h)	14.38 ± 2.94	15.36 ± 3.70	–

NB: Two naltrexone SR 8 mg/bupropion SR 90 mg tablets
GMR: Geometric mean ratio, CI: Confidence interval
AUCs, C_{max} and $t_{1/2}$ are presented as means ± SD; whereas T_{max} is presented as median (min , max)

Table 3: Summary of PK Parameters and Statistical Analyses of Naltrexone and its Active Metabolite 6-Beta-Naltrexol for NB + Valsartan Versus NB Alone

Analyte	PK Parameter	NB Alone	NB + Valsartan	%GMR (90% CI)
Naltrexone	AUC_{0-t} (ng•h/mL)	9.62 ± 5.66	9.89 ± 5.92	101.30 (90.63 - 113.22)
	$AUC_{0-\infty}$ (ng•h/mL)	10.55 ± 5.76	10.14 ± 5.91	98.58 (88.59 - 109.70)
	C_{max} (ng/mL)	1.37 ± 0.943	1.27 ± 0.743	93.39 (80.68 - 108.10)
	T_{max} (h)	1.76 (0.75, 6.00)	2.00 (0.50, 6.05)	–
	$t_{1/2}$ (h)	4.94 ± 1.60	4.75 ± 1.46	–
6-Beta Naltrexol	AUC_{0-t} (ng•h/mL)	243.27 ± 55.81	227.95 ± 47.30	92.64 (88.84 - 96.60)
	$AUC_{0-\infty}$ (ng•h/mL)	257.85 ± 57.52	245.67 ± 48.98	94.19 (91.02 - 97.47)
	C_{max} (ng/mL)	15.8 ± 4.46	15.4 ± 4.39	95.67 (87.39 - 104.73)
	T_{max} (h)	2.50 (0.75, 6.00)	3.00 (0.75, 6.05)	–
	$t_{1/2}$ (h)	14.38 ± 2.94	14.97 ± 3.55	–

NB: Two naltrexone SR 8 mg/bupropion SR 90 mg tablets
GMR: Geometric mean ratio, CI: Confidence interval
AUCs, C_{max} and $t_{1/2}$ are presented as means ± SD; whereas T_{max} is presented as median (min , max)

Figure 2: Mean Plasma Bupropion Concentration-Time Profiles (Linear plot)

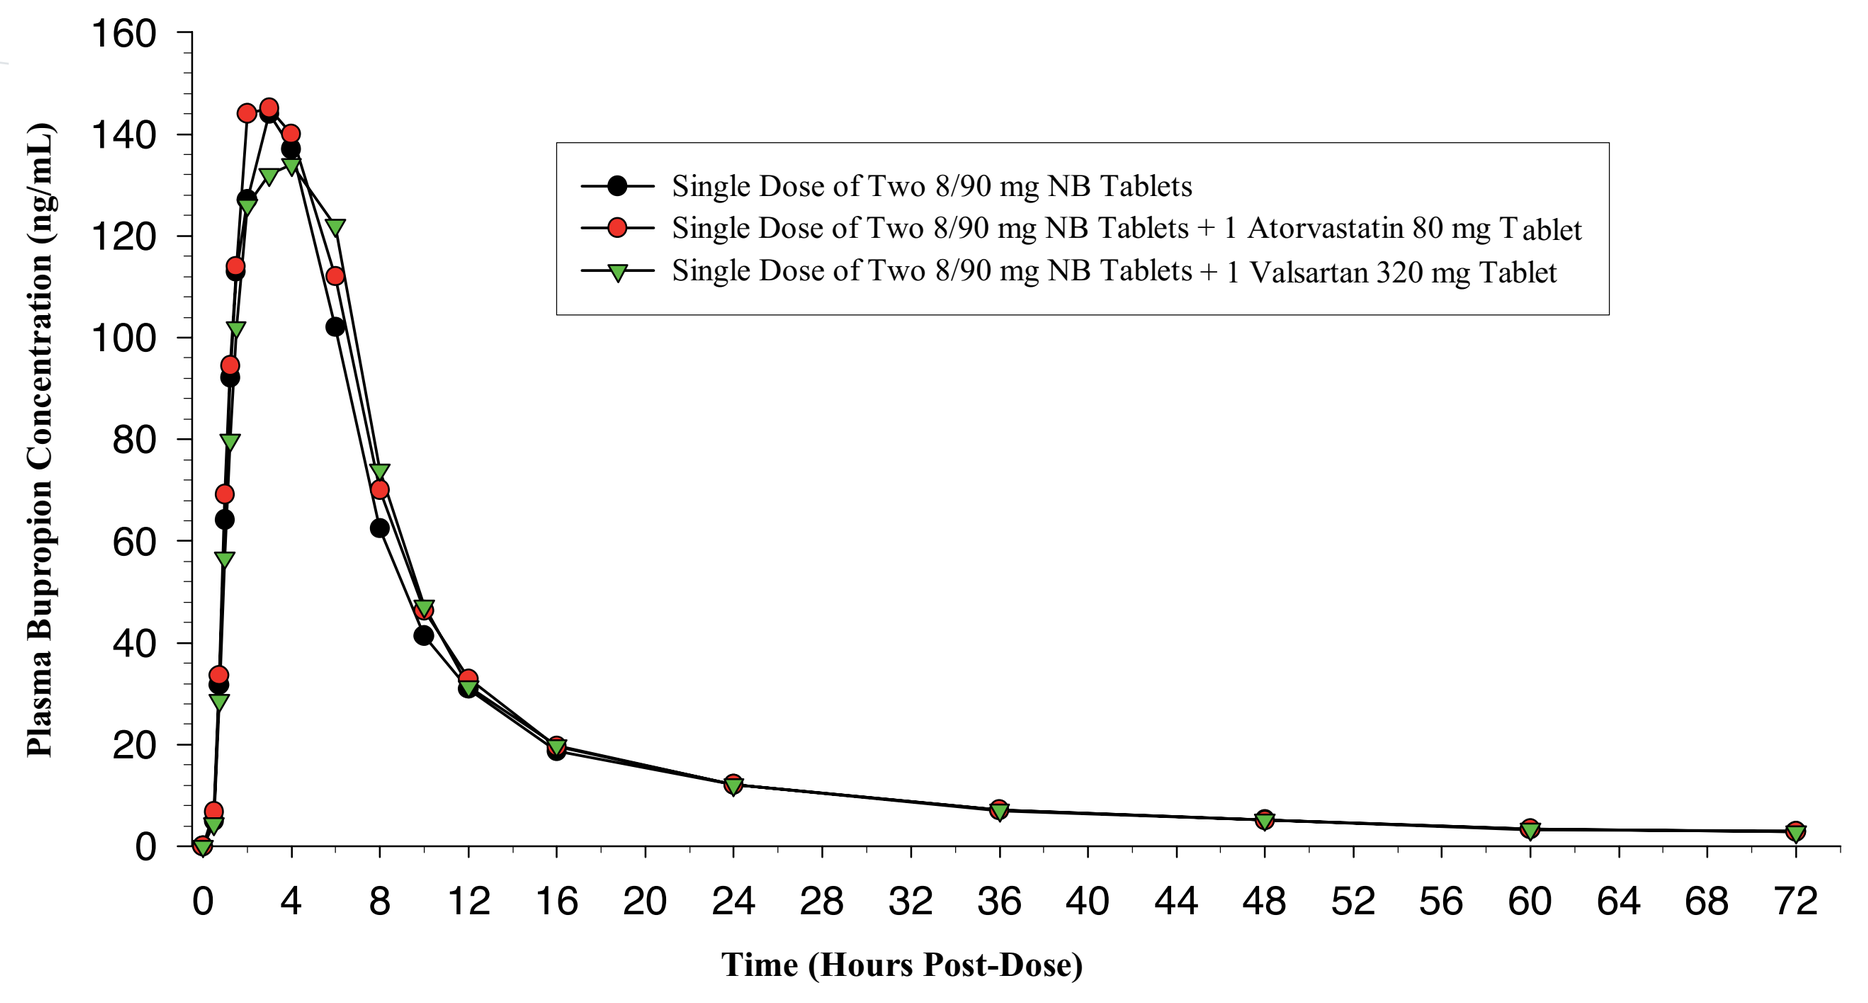
Plasma Bupropion Concentration (ng/mL)
Time (Hours Post-Dose)
Single Dose of Two 8/90 mg NB Tablets
Single Dose of Two 8/90 mg NB Tablets + 1 Atorvastatin 80 mg Tablet
Single Dose of Two 8/90 mg NB Tablets + 1 Valsartan 320 mg Tablet

NB: naltrexone and bupropion combination

Table 4: Summary of PK Parameters and Statistical Analyses of Bupropion and its Active Metabolites for NB+ Atorvastatin Versus NB Alone

Analyte	PK Parameter	NB Alone	NB + Atorvastatin	%GMR (90% CI)
Bupropion	AUC_{0-t} (ng•h/mL)	1523.25 ± 500.58	1603.55 ± 467.17	105.25 (99.67 - 111.15)
	$AUC_{0-\infty}$ (ng•h/mL)	1613.41 ± 549.55	1670.35 ± 484.67	103.88 (98.45 - 109.60)
	C_{max} (ng/mL)	157 ± 35.7	164 ± 29.1	104.85 (95.64 - 114.96)
	T_{max} (h)	3.00 (1.25, 4.00)	2.10 (1.50, 5.99)	–
	$t_{1/2}$ (h)	24.74 ± 6.74	25.33 ± 8.34	–
Hydroxybupropion	AUC_{0-t} (ng•h/mL)	17563.61 ± 8906.10	15353.69 ± 5921.43	89.96 (82.93 - 97.59)
	$AUC_{0-\infty}$ (ng•h/mL)	19087.41 ± 9761.76	15392.26 ± 6019.46	96.26 (88.61 -104.56)
	C_{max} (ng/mL)	365 ± 172	351 ± 123	96.77 (89.80 - 104.28)
	T_{max} (h)	6.01 (3.99, 72.00)	6.00 (4.00, 11.99)	–
	$t_{1/2}$ (h)	29.83 ± 5.97	28.38 ± 6.25	–
Threohydrobupropion	AUC_{0-t} (ng•h/mL)	6096.45 ± 4163.75	5833.65 ± 3828.94	96.93 (91.85 - 102.29)
	$AUC_{0-\infty}$ (ng•h/mL)	7715.50 ± 6582.76	7031.08 ± 4463.37	101.90 (94.63 - 109.72)
	C_{max} (ng/mL)	137 ± 57.6	136 ± 46.9	99.56 (93.76 - 105.72)
	T_{max} (h)	6.00 (3.99, 8.01)	6.00 (5.98, 8.00)	–
	$t_{1/2}$ (h)	45.06 ± 9.98	47.62 ± 7.66	–
Erythrohydrobupropion	AUC_{0-t} (ng•h/mL)	1154.17 ± 648.99	1155.09 ± 654.37	100.41 (94.67 - 106.49)
	$AUC_{0-\infty}$ (ng•h/mL)	1264.53 ± 702.59	1305.06 ± 792.81	98.64 (92.96 - 104.67)
	C_{max} (ng/mL)	23.5 ± 6.42	23.7 ± 6.23	99.73 (95.11 - 104.58)
	T_{max} (h)	6.01 (5.99, 12.00)	8.00 (5.98, 16.00)	–
	$t_{1/2}$ (h)	32.92 ± 8.93	31.69 ± 7.63	–

NB: Two naltrexone SR 8 mg/bupropion SR 90 mg tablets
GMR: Geometric mean ratio, CI: Confidence interval
AUCs, C_{max} and $t_{1/2}$ are presented as means ± SD; whereas T_{max} is presented as median (min, max)

Table 5: Summary of PK Parameters and Statistical Analyses of Bupropion and its Active Metabolites for NB + Valsartan Versus NB Alone

Analyte	PK Parameter	NB Alone	NB + Valsartan	%GMR (90% CI)
Bupropion	AUC_{0-t} (ng•h/mL)	1523.25 ± 500.58	1581.20 ± 494.70	101.12 (95.64 - 106.92)
	$AUC_{0-\infty}$ (ng•h/mL)	1613.41 ± 549.55	1649.44 ± 510.89	100.08 (94.74 - 105.72)
	C_{max} (ng/mL)	157 ± 35.7	165 ± 32.6	103.68 (94.38 - 113.89)
	T_{max} (h)	3.00 (1.25, 4.00)	3.00 (1.49, 6.00)	–
	$t_{1/2}$ (h)	24.74 ± 6.74	26.74 ± 8.27	–
Hydroxybupropion	AUC_{0-t} (ng•h/mL)	17563.61 ± 8906.10	14425.43 ± 5966.95	82.61 (76.02 - 89.78)
	$AUC_{0-\infty}$ (ng•h/mL)	19087.41 ± 9761.76	15366.31 ± 6817.73	85.60 (78.98 - 92.77)
	C_{max} (ng/mL)	365 ± 172	351 ± 125	96.09 (89.03 - 103.72)
	T_{max} (h)	6.01 (3.99, 72.00)	6.00 (4.00, 15.97)	–
	$t_{1/2}$ (h)	29.83 ± 5.97	28.23 ± 6.80	–
Threohydrobupropion	AUC_{0-t} (ng•h/mL)	6096.45 ± 4163.75	5468.16 ± 3695.85	89.52 (84.73 - 94.58)
	$AUC_{0-\infty}$ (ng•h/mL)	7715.50 ± 6582.76	5651.95 ± 2282.83	90.59 (83.69 - 98.06)
	C_{max} (ng/mL)	137 ± 57.6	140 ± 44.4	102.46 (96.36 - 108.94)
	T_{max} (h)	6.00 (3.99, 8.01)	6.00 (2.01, 8.02)	–
	$t_{1/2}$ (h)	45.06 ± 9.98	46.49 ± 9.48	–
Erythrohydrobupropion	AUC_{0-t} (ng•h/mL)	1154.17 ± 648.99	1011.37 ± 481.12	88.83 (83.65 - 94.33)
	$AUC_{0-\infty}$ (ng•h/mL)	1264.53 ± 702.59	1056.73 ± 395.38	87.98 (82.80 - 93.47)
	C_{max} (ng/mL)	23.5 ± 6.42	24.0 ± 5.78	101.21 (96.42 - 106.24)
	T_{max} (h)	6.01 (5.99, 12.00)	7.00 (2.01, 10.00)	–
	$t_{1/2}$ (h)	32.92 ± 8.93	32.21 ± 9.50	–

NB: Two naltrexone SR 8 mg/bupropion SR 90 mg tablets
GMR: Geometric mean ratio, CI: Confidence interval
AUCs, C_{max} and $t_{1/2}$ are presented as means ± SD; whereas T_{max} is presented as median (minimum, maximum)

Co-administration of either a single dose of atorvastatin or valsartan did not affect the PK of bupropion or naltrexone. The % GMR and the 90% CI of the bupropion PK parameters were within 80 - 125%.

Table 6: Pharmacokinetic Parameters of Atorvastatin, Ortho-Atorvastatin and Para-Atorvastatin PK Results - Comparison with Literature Data

Study	Analyte	$AUC_{0-\infty}$ (ng•h/mL)	C_{max} (ng/mL)	T_{max} (h)	$t_{1/2}$ (h)
NB-232, 80 mg					
	A	160.8 (43%) (78.4 - 336.2)	31.0 (43%) (11.3 - 66.0)	0.95 (97%) (0.43 - 3.99)	7.19 (58%) (3.51 - 18.76)
	o-A	168.8 (57%) (53.7 - 510.1)	18.8 (50%) (6.86 - 36.0)	2.17 (100%) (0.74 - 10.00)	9.29 (59%) (3.58 - 25.21)
	p-A	42.3 (112%) (13.7 - 167.2)	1.50 (76%) (0.62 - 5.33)	9.06 (36%) (0.74 - 12.0)	14.18 (54%) (6.16 - 26.78)
Mendoza L et al. (2006), 40 mg					
	A	57.7	14.5	0.69 (55%)	9.07 (30%)
	o-A	69.6	10.5	1.27 (78%)	9.11 (21%)
Data on file, 80 mg					
	A	148 (41%)	35 (55%)	1 (75%)	15 (45%)
	o-A	211 (43%)	32 (60%)	1.5 (70%)	16 (35%)
	p-A	29 (57%)	2 (100%)	6 (100%)	27 (50%)
Posvar EL et al. (1996), 80 mg					
	A	335 (50 - 850)	33.1 (10 - 100)	2.8 (0.5 - 6)	19.2 (14.7 - 57.6)
	o-A	Not Available	Not Available	Not Available	Not Available

PK data are expressed as mean with or without CV and/or range under parentheses if available.
NB: Two naltrexone SR 8 mg/bupropion SR 90 mg tablets
A: atorvastatin, o-A: o-hydroxy-atorvastatin, p-A: p-hydroxy-atorvastatin

Mean PK parameters of atorvastatin and hydroxy-atorvastatin were either comparable or within the range of values reported in the literature and the Celerion (formerly MDS Pharma Services) data on file.

Table 7: Mean (CV%) Valsartan PK Results - Comparison with Literature Data

Study	AUC_{0-t} (ng•h/mL)	$AUC_{0-\infty}$ (ng•h/mL)	C_{max} (ng/mL)	T_{max} (h)	$t_{1/2}$ (h)
NB-232 (320 mg)	41615 (45%)	43867 (45%)	4600 (39%)	4.00 (1.3 - 8)	11.9 (73%)
FDA[a] (320 mg)	41050 (33%)	42683 (31%)	6162 (34%)	2.75 (1 - 4.37)	14.7 (65%)
Séchaud R et al., 2002 (320 mg)	not applicable	42680	6162	2.75 (1 - 4.37)	not applicable
Schmidt EK et al., 1998 (160 mg)	not applicable	not applicable	2250 (48%)	2.0 (2.0 - 3.0)	5.73 (15%)

AUCs, C_{max} and $t_{1/2}$ are expressed as mean with or without CV whereas T_{max} is given as median (range).
[a]Data from FDA Clinical Pharmacology and Biopharmaceutics Review, Application Number 21-283 (Diovan tablets).
NB: Two naltrexone SR 8 mg/bupropion SR 90 mg tablets

Despite the small increase of 1.25 to 2.0 hours that was observed in median T_{max} when results from this study (NB-232) are compared to the literature data, the arithmetic mean plasma valsartan AUC_{0-t}, $AUC_{0-\infty}$, and t½ values were comparable to the values reported in the literature. In addition, mean C_{max} appeared to be 2-fold greater when compared to C_{max} values obtained at half the dose (160 mg), and with approximately 25% lower C_{max} values than previously reported data at the 320 mg dose.

Safety Results

There were no serious or severe adverse events and no treatment-related trends in clinical assessments including AEs, vital signs, and laboratory measures. There were no differences in adverse events or other safety measures when NB was administered alone relative to co-administration with atorvastatin or valsartan. The most common AEs in this study were nausea and dizziness. There were 4 events of mild nausea reported by 3 (15%) subjects (2 subjects following NB + valsartan, and 1 subject following NB + atorvastatin and NB alone) and 3 events of mild dizziness reported by 3 (15%) subjects (1 subject each following all treatments).

CONCLUSIONS

- Co-administration of either single dose of atorvastatin or valsartan with NB (two 8/90 mg tablets) did not affect the PK of naltrexone or bupropion. The % GMR and the 90%CI of the N and B PK parameters were within 80 - 125%.
- The comparison with literature data showed that co-administration of single dose of two 8/90 mg NB tablets in combination with one atorvastatin 80 mg tablet or with one valsartan 320 mg tablet had no effect on the PK exposure of atorvastatin or valsartan. A small increase in the valsartan median Tmax of 1.25 to 2.0 hours was observed when compared to the literature data.
- Single-dose administration of NB (two 8/90 mg tablets) alone or with either atorvastatin or valsartan appeared to be generally safe and well tolerated by the healthy male and female subjects.

REFERENCES

1. FDA CDER, Clinical Pharmacology and Biopharmaceutics Review for Application Number 21-283 (Diovan tablets). 2001.
2. Mendoza L, Hajdúch M, Plaušinaitis R, Platílová V, Emritte N, Svoboda M. Pharmacokinetic and bioequivalence testing of atorvastatin formulations in healthy male volunteers. Pharmazie 2006; 61: 805-806.
3. Posvar EL, Radulovic LL, Cilla DD, Jr., Whitfield LR, Sedman AJ. Tolerance and pharmacokinetics of single-dose atorvastatin, a potent inhibitor of HMG-CoA reductase, in healthy subjects. J Clin Pharmacol 1996; 36: 728-731.
4. Séchaud R, Graf P, Bigler H, Gruendl E, Letzkus M, Merz M. Bioequivalence study of a valsartan tablet and a capsule formulation after a single dosing in healthy volunteers using a replicated crossover design. Int J Clin Pharmacol Ther 2002; 40(1): 35-40.
5. Schmidt EK, Antonin K-H, Flesch G, Racine-Poon A. An interaction study with cimetidine and the new angiotensin II antagonist valsartan. Eur J Pharmacol 1998; 53: 451-458.

Funded by Orexigen Therapeutics, Inc.

For more information, contact medinfo@orexigen.com

celerion

www.celerion.com