COMPARATIVE BIOAVAILABILITY STUDY OF THREE DOSAGE FORMS OF LEVOTHYROXINE SODIUM TABLETS FOLLOWING SINGLE DOSES IN HEALTHY SUBJECTS

M. Obaidi, PhD[1], T. M. Grant, PhD[1], P. Chai, PhD[1], J. Tan-Fink, MA[1], L. C. Nix, PharmD[1], C. M. Brandquist, PharmD, MS[1], E. Offman, B.Sc. Pharm, MSc[2], and M. J. Lamson, PhD[3]

[1,2]Celerion, Lincoln, NE and Montreal, Canada, respectively
[3]King Pharmaceutical Research and Development, Inc, Cary, NC

OBJECTIVE

In October 2007, the U.S. Food and Drug Administration (FDA) mandated that all manufacturers of levothyroxine (T_4) sodium drugs used for thyroid replacement therapy tighten their potency specifications to ensure that the drug retains its potency over the shelf-life of the product.[1] To meet this challenge, King Pharmaceuticals, Inc. has developed a high-stability levothyroxine tablet dosage form, which has been shown *in vitro* to meet the proposed tighter potency requirements. The objective of this study was to determine the dosage form proportionality of this newly reformulated higher-stability tablet using a study design adapted from the Levothyroxine Guidance.[2]

METHODOLOGY

- The design was a randomized, open-label, single-dose, three-way crossover study to determine the dosage form proportionality of T_4 sodium tablets in healthy volunteers.
- Subjects were randomly assigned to receive the following treatments in the 3 dosing periods:
 - Levothyroxine sodium tablets 600 µg (12 x 50 µg tablets) with 240 mL of ambient-temperature water (Treatment A)
 - Levothyroxine sodium tablets 600 µg (6 x 100 µg tablets) with 240 mL of ambient-temperature water (Treatment B)
 - Levothyroxine sodium tablets 600 µg (3 x 200 µg tablets) with 240 mL of ambient-temperature water (Treatment C)
- There was a 35-day washout period between dosing periods.
- Peak (C_{max}), total exposure (AUC_{0-24} and AUC_{0-48}), and T_{max} were calculated for uncorrected and baseline-corrected serum concentrations of T_4 and uncorrected serum concentrations of T_3 (triiodothyronine) using WinNonlin® Version.5.0.1 (Pharsight, Mountain View, CA).
- Statistical comparisons of In-transformed PK parameters (C_{max}, AUC_{024h}, and AUC_{048h}) between Treatment A versus Treatment B, Treatment A versus Treatment C, and Treatment B versus Treatment C were performed with PROC MIXED of SAS® Version 9.1.3 (SAS Institute Inc., Cary, NC) using an analysis of variance (ANOVA) model with sequence, treatment, and period as fixed effects, and subject within sequence as a random effect.
- The least-squares (LS) means, the geometric mean ratios (GMR: ratio of exponentiated LS means), and the 90% confidence intervals (CIs) for each PK parameter were calculated to evaluate the dosage form proportionality for each of the treatment comparisons.
- Dosage form proportionality of levothyroxine was not rejected if the 90% CIs for the GRMs fell within the 80% to 125% CIs bioequivalence criteria for the uncorrected serum T_4 PK parameters, for each of the 3 comparisons. The 90% CIs for baseline-corrected serum T_4 and uncorrected serum T_3 were presented as supportive information.

RESULTS

- Thirty-six (36) subjects (16 female and 20 male) were enrolled and 34 subjects completed all 3 study periods. The demographic information of the subjects who were enrolled in the study is presented in Table 1.
- The concentration-time profiles of uncorrected and baseline-corrected serum T_4 and uncorrected serum T_3 following the administration of Treatments A, B, and C are presented in Figures 1, 2, and 3, respectively.
- The GMRs, LS means, and the 90% CIs derived from the analysis of the Intransformed C_{max}, AUC_{0-24h}, and AUC_{0-48h} for uncorrected and baseline-corrected serum T_4 and uncorrected serum T_3 are presented in Table 2.
- The GMRs of uncorrected serum T_4 C_{max}, AUC_{024h}, and AUC_{048h} for the comparisons of Treatments A, B, and C were approximately 100% (± 1 to 2%) indicating that the 3 tablet strengths had similar uncorrected serum T_4 exposure. The 90% CIs of the mean ratios for the PK parameters were all within the 80% to 125% range, indicating that the 3 tablet strengths were proportional with respect to uncorrected serum T_4 exposure.
- The GMRs of baseline-corrected serum T_4 C_{max}, AUC_{0-24h}, and AUC_{0-48h} for the comparisons of Treatments A, B, and C were within 100% (± 8%) indicating that the 3 tablet strengths had similar baseline-corrected serum T_4 exposure. The 90% CIs of the mean ratios for the PK parameters were all within the 80% to 125% range, indicating that the 3 tablet strengths were proportional with respect to baseline-corrected serum T_4 exposure.
- The GMRs of uncorrected serum T_3 C_{max}, AUC_{0-24h}, and AUC_{0-48h} for the comparisons of Treatments A, B, and C were approximately 100% (± 1%) indicating that the 3 tablet strengths had similar uncorrected serum T_3 exposure. The 90% CIs of the mean ratios for the PK parameters were all within the 80 to 125% range, indicating that 3 tablet strengths were proportional with respect to uncorrected serum T_3 exposure.

Table 1: Demographic Information of Subjects Enrolled in the Study

Variable	Female (N = 16)	Male (N = 20)	Total (N = 36)
Race, N (%)			
White	16 (100%)	20 (100%)	36 (100%)
Ethnicity, N (%)			
Hispanic or Latino	16 (100%)	18 (90%)	34 (94%)
Not Hispanic or Latino	0	2 (10%)	2 (6%)
Age (years)			
Mean ± SD	31.6 ± 9.47	35.1 ± 8.82	33.5 ± 9.15
Weight (kg)			
Mean ± SD	66.4 ± 11.82	76.9 ± 11.22	72.3 ± 12.50
Height (cm)			
Mean ± SD	156.4 ± 8.14	168.8 ± 7.42	163.3 ± 9.84
Body Mass Index (kg/m^2)			
Mean ± SD	27.0 ± 3.26	26.9 ± 2.70	26.9 ± 2.92

Table 2: Geometric Least Squares Mean Ratio and 90% Confidence Interval for Uncorrected and Baseline-corrected Serum T_4 and Uncorrected Serum T_3 Pharmacokinetic Parameters (PK Population)

Analyte	Pharmacokinetic Parameters	Treatment A Versus Treatment B	Treatment A Versus Treatment C	Treatment B Versus Treatment C
Uncorrected Serum T_4	C_{max}	99.27 (96.43, 102.19)	101.52 (98.58, 104.54)	102.26 (99.30, 105.31)
	AUC_{0-24h}	99.74 (97.73, 101.80)	101.56 (99.49, 103.67)	101.82 (99.74, 103.94)
	AUC_{0-48h}	99.38 (97.48, 101.31)	101.23 (99.27, 103.22)	101.86 (99.89, 103.87)
Baseline-corrected Serum T_4	C_{max}	95.34 (89.98, 101.02)	100.65 (94.93, 106.71)	105.57 (99.57, 111.93)
	AUC_{0-24h}	93.86 (88.21, 99.88)	100.77 (94.63, 107.30)	107.35 (100.81, 114.32)
	AUC_{0-48h}	91.64 (85.52, 98.19)	99.29 (92.59, 106.46)	108.35 (101.04, 116.19)
Uncorrected Serum T_3	C_{max}	99.76 (96.48, 103.15)	100.96 (97.61, 104.43)	101.20 (97.84, 104.68)
	AUC_{0-24h}	100.78 (98.29, 103.33)	100.56 (98.05, 103.13)	99.78 (97.29, 102.33)
	AUC_{0-48h}	100.50 (98.17, 102.88)	100.56 (98.21, 102.98)	100.07 (97.72, 102.47)

Treatment A = Levothyroxine sodium 600 µg (12 x 50 µg tablets), N = 36
Treatment B = Levothyroxine sodium 600 µg (6 x 100 µg tablets), N = 35
Treatment C = Levothyroxine sodium 600 µg (3 x 200 µg tablets), N = 34

CONCLUSIONS

- The statistical analyses, and the nearly superimposable mean serum concentration–time profiles of the 3 treatments, indicate that the exposures to uncorrected and baseline-corrected serum T_4 and uncorrected serum T_3 following the oral administration of the 3 tablet strengths studied were similar.
- The 90% CIs of the GMRs for the PK parameters of uncorrected and baseline-corrected serum T_4 and uncorrected serum T_3 fell within the 80% to 125% range, indicating that the administration of the 3 tablet strengths studied resulted in proportional exposure to serum T_4 and T_3.

REFERENCES

1 FDA NEWS. FDA Acts to Ensure Thyroid Drugs Don't Lose Potency Before Expiration Date. U.S. Department of Health and Human Services, Food and Drug Administration, Center for Drug Evaluation and Research (CDER). October 3, 2007.

2 Guidance for Industry: Levothyroxine Sodium Tablets – *In Vivo* Pharmacokinetic and Bioavailability Studies and *In Vitro* Dissolution Testing. U.S. Department of Health and Human Services, Food and Drug Administration, Center for Drug Evaluation and Research (CDER). December 2000.

Figure 1: Mean Uncorrected Serum T_4 Concentration-Time Profile Following Levothyroxine Treatments

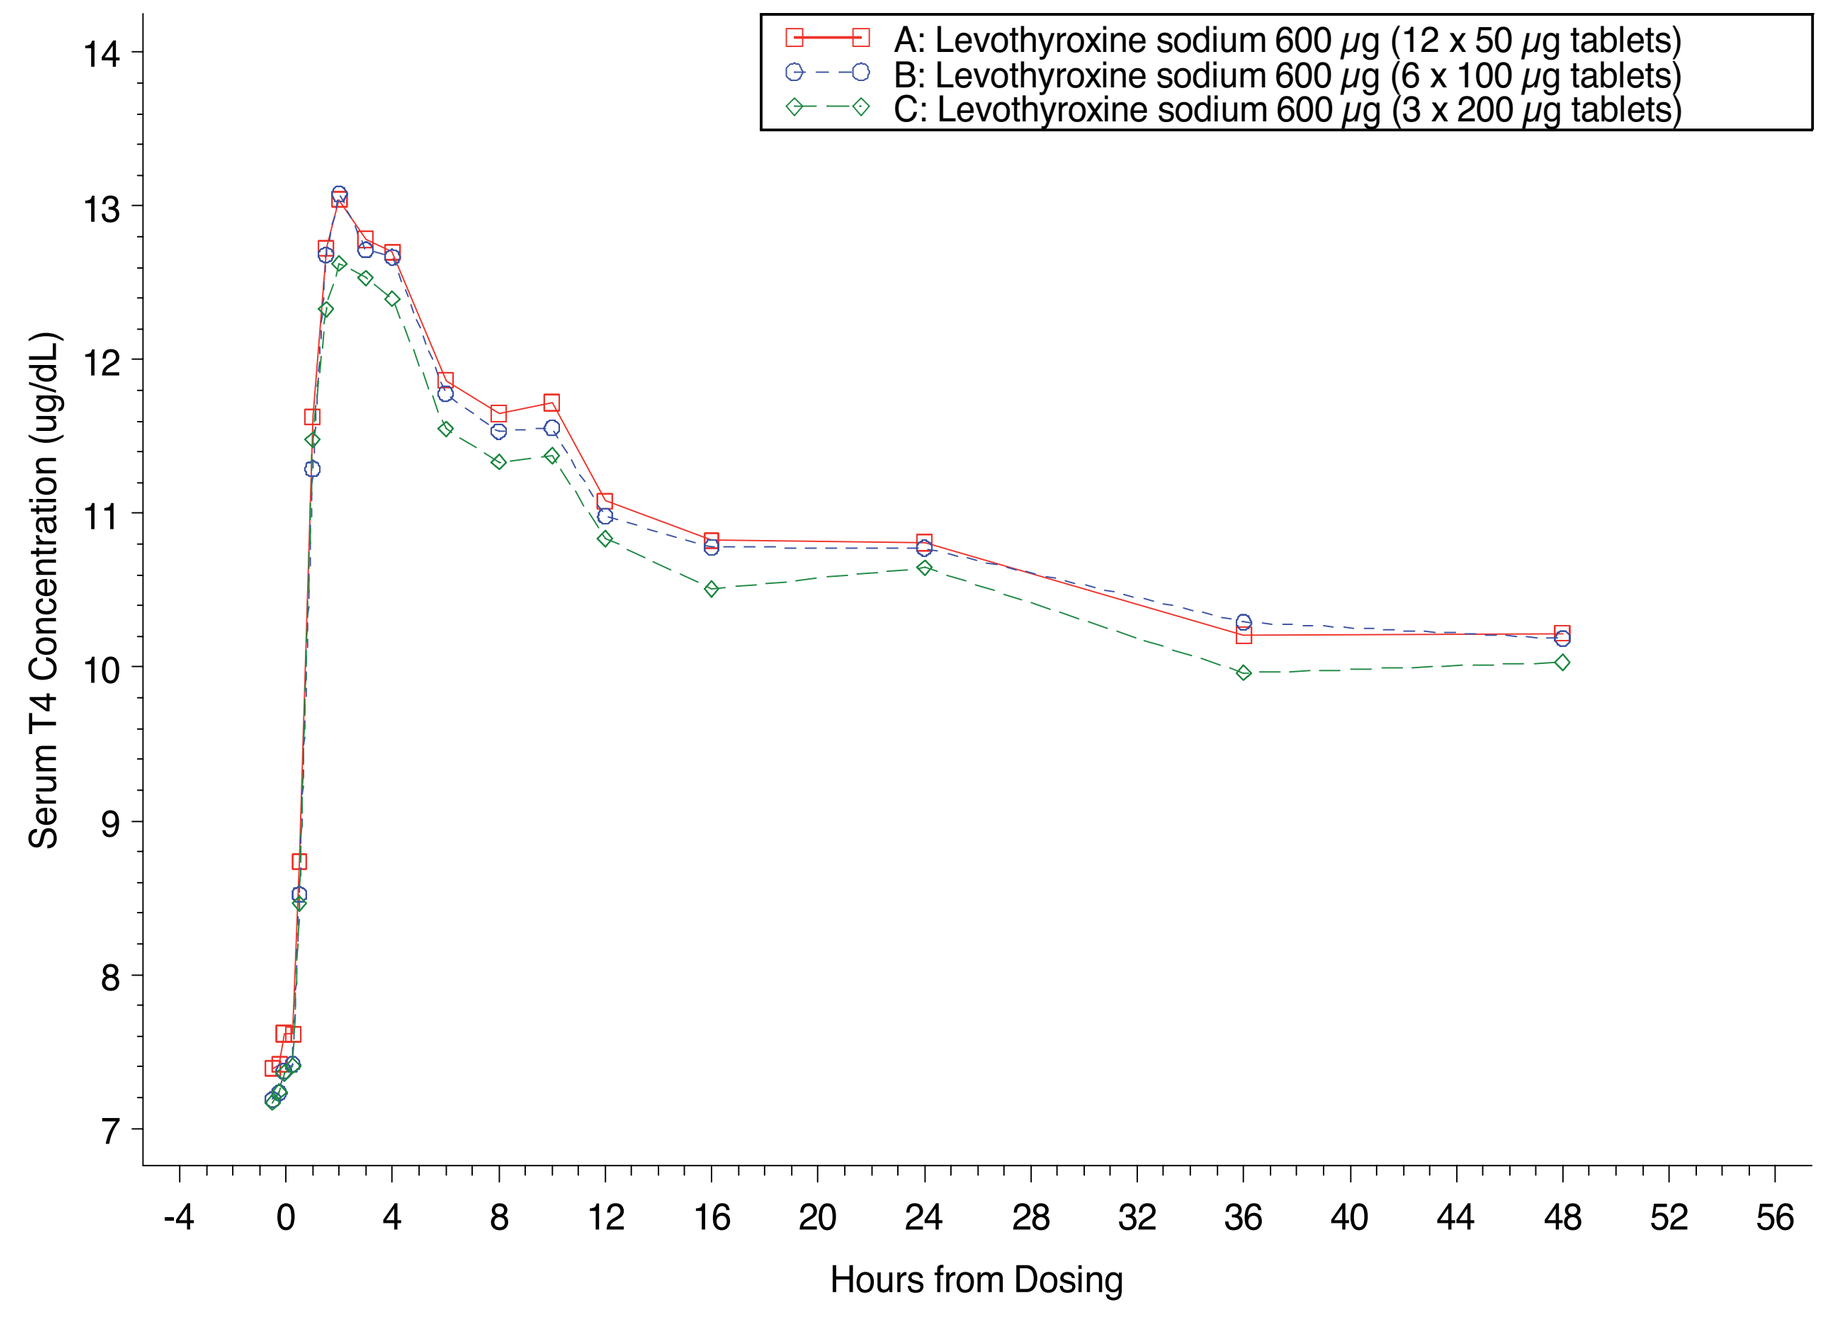
A: Levothyroxine sodium 600 µg (12 x 50 µg tablets)
B: Levothyroxine sodium 600 µg (6 x 100 µg tablets)
C: Levothyroxine sodium 600 µg (3 x 200 µg tablets)
Serum T4 Concentration (ug/dL)
Hours from Dosing

Figure 2: Mean Baseline-Corrected Serum T_4 Concentration-Time Profile Following Levothyroxine Treatments

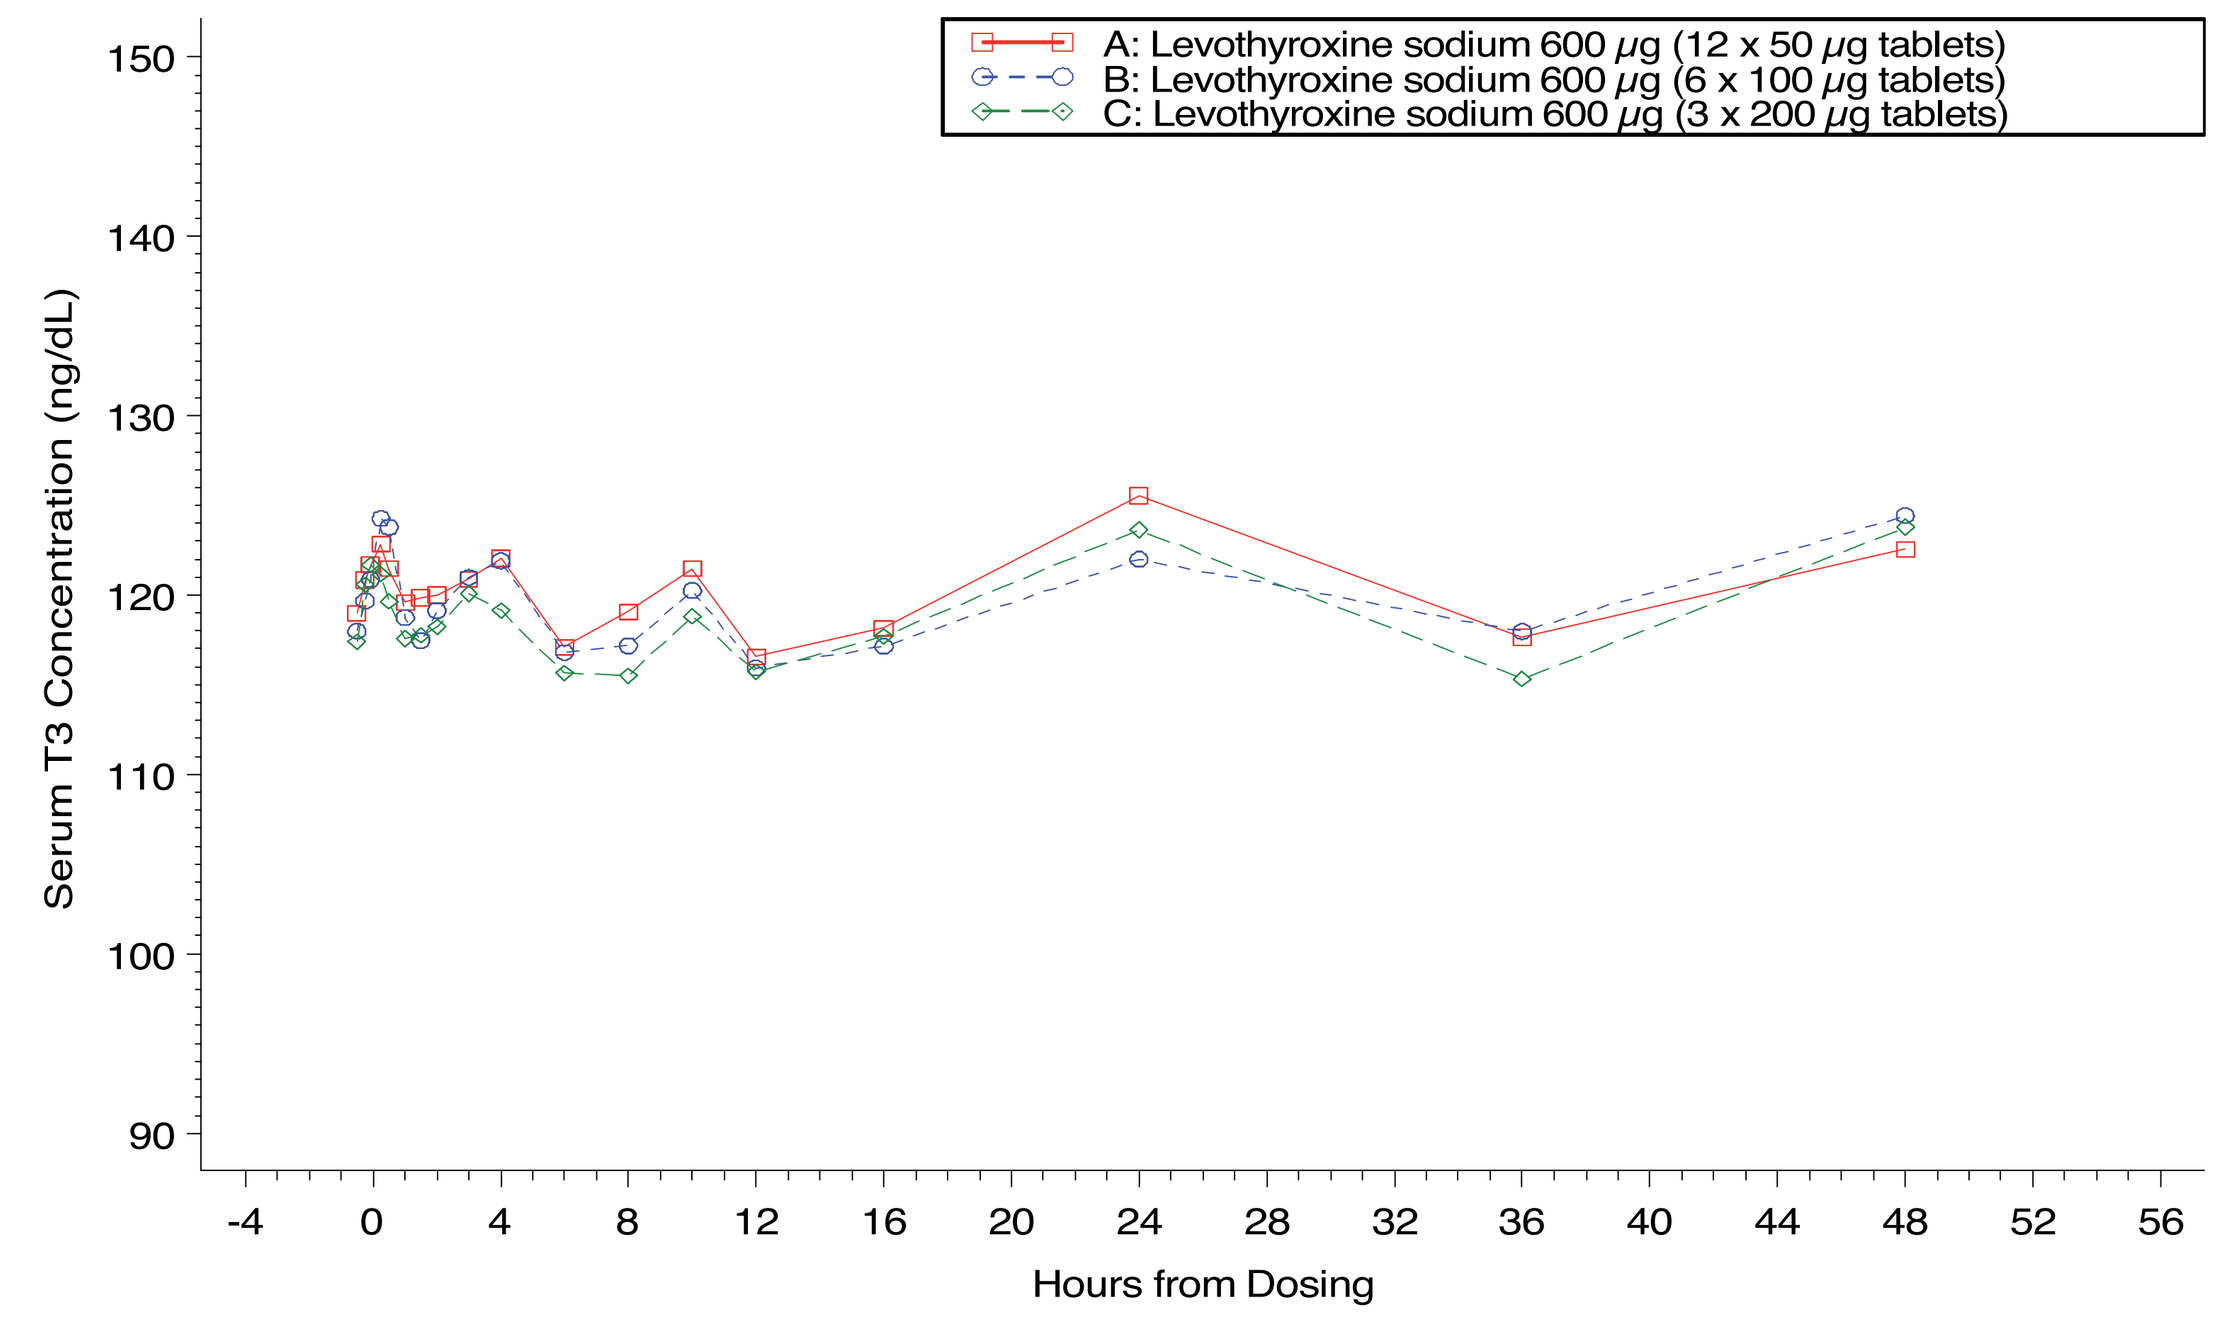
A: Levothyroxine sodium 600 µg (12 x 50 µg tablets)
B: Levothyroxine sodium 600 µg (6 x 100 µg tablets)
C: Levothyroxine sodium 600 µg (3 x 200 µg tablets)
Serum T3 Concentration (ng/dL)
Hours from Dosing

Figure 3: Mean Uncorrected Serum T_3 Concentration-Time Profile Following Levothyroxine Treatments

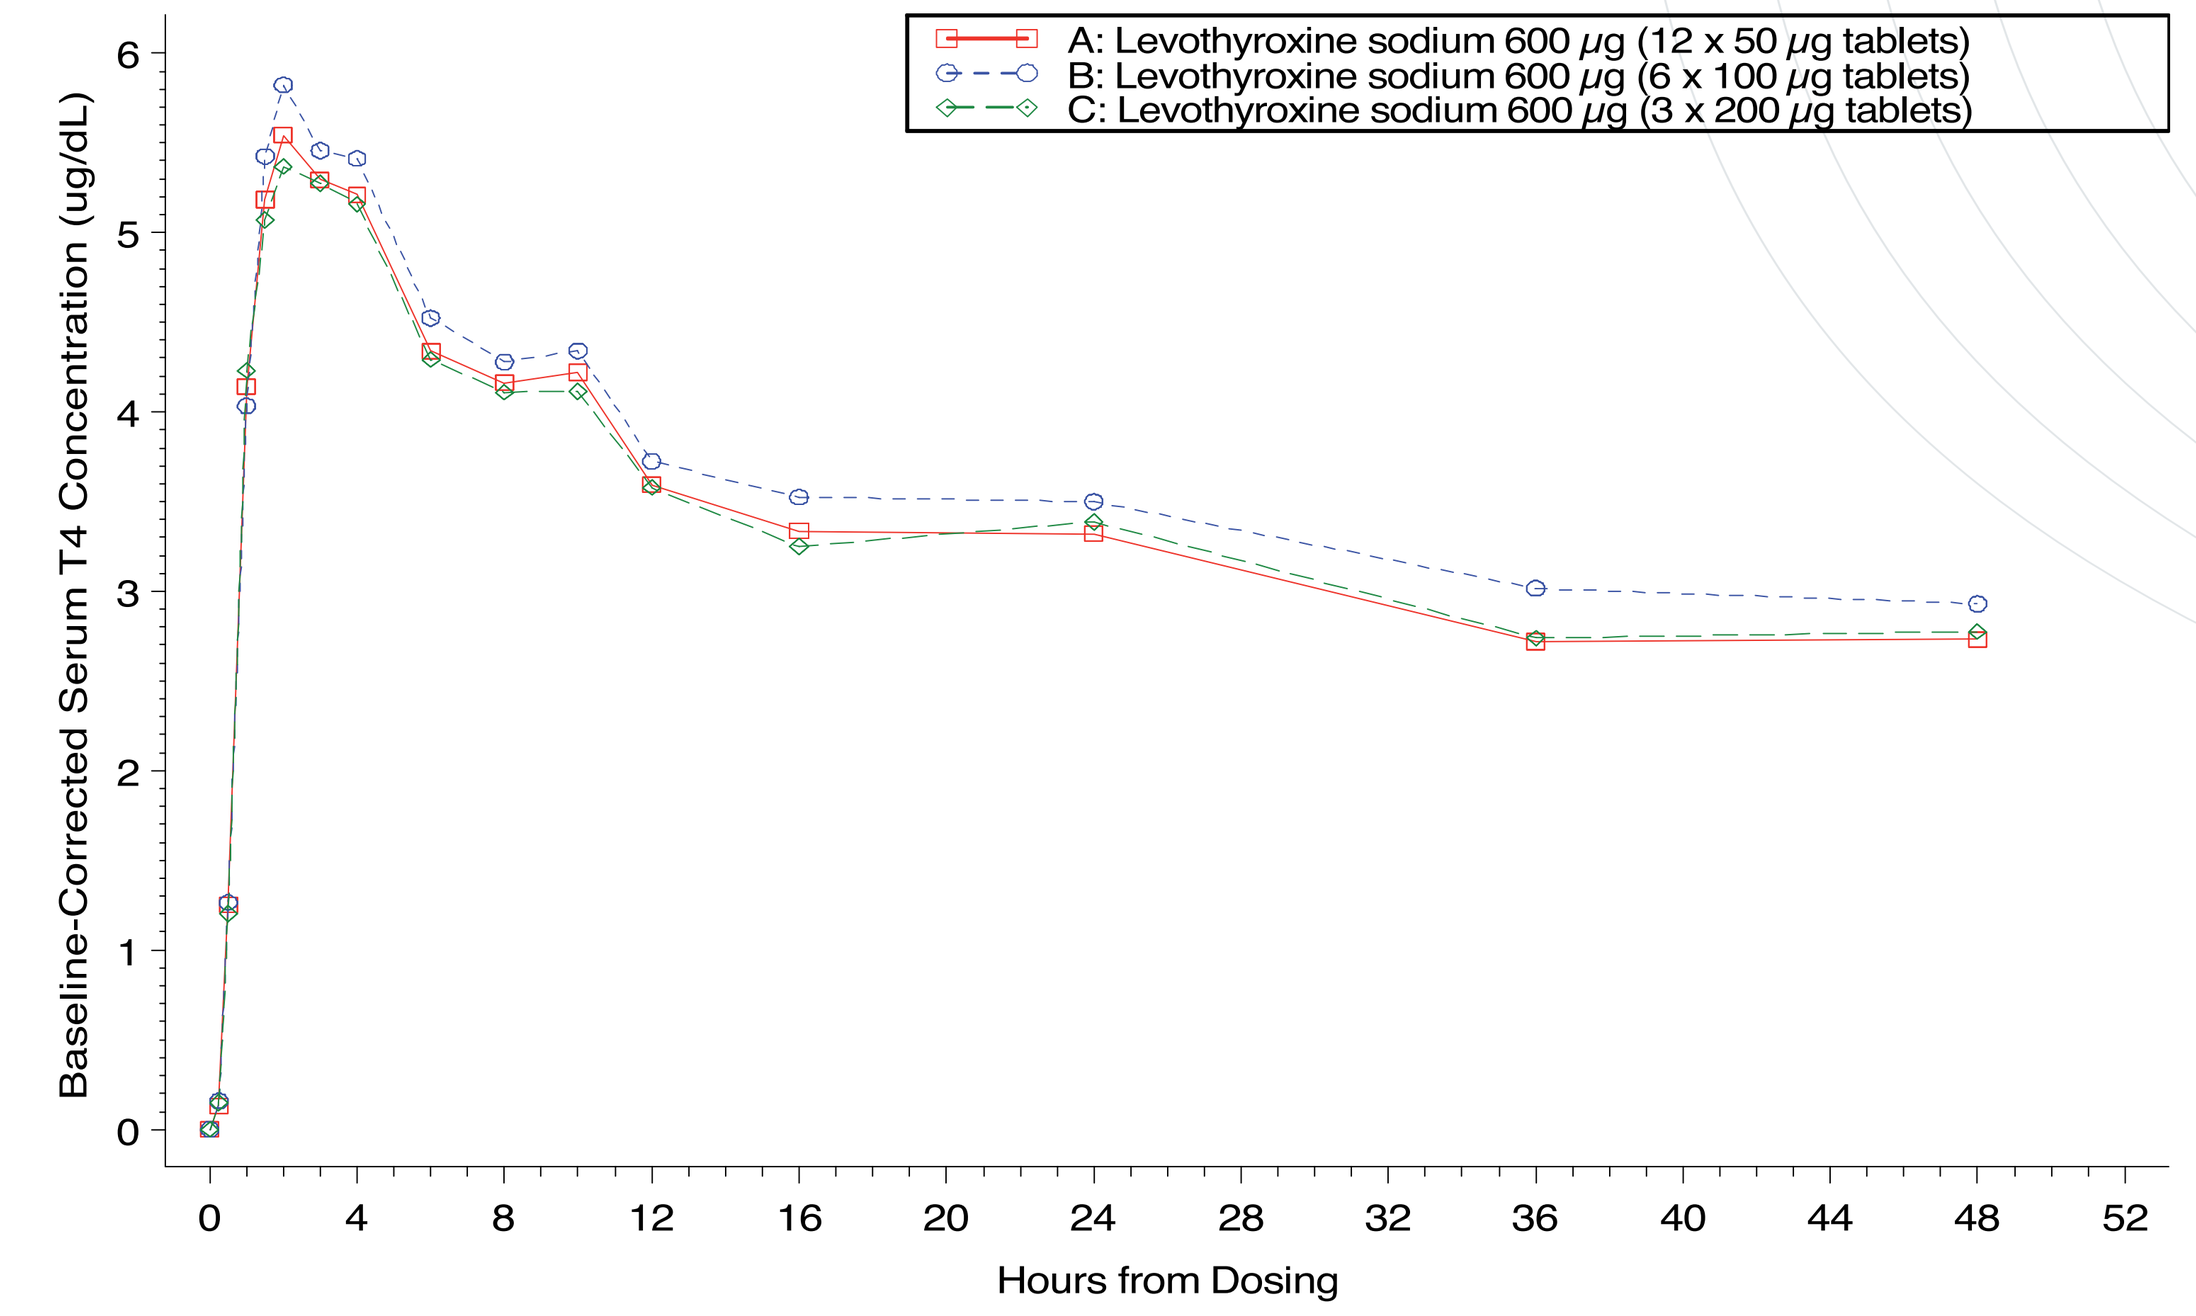
A: Levothyroxine sodium 600 µg (12 x 50 µg tablets)
B: Levothyroxine sodium 600 µg (6 x 100 µg tablets)
C: Levothyroxine sodium 600 µg (3 x 200 µg tablets)
Baseline-Corrected Serum T4 Concentration (ug/dL)
Hours from Dosing

www.celerion.com