A highly selective and sensitive LC-MS/MS method for analysis of glucagon in Human Plasma

Veni N. Lapko, Paul Brown, Ridha Nachi, Chris Kafonek, Alan Dzerk, John Rollag and Corey Ohnmacht
Celerion, Lincoln, NE

celerion

INTRODUCTION:

- Glucagon is a polypeptide hormone that increases blood glucose concentration and has been used as an emergency treatment of hypoglycemia
- Historically, methods for glucagon analysis of bioequivalence studies, were based on immunochemical approaches intended for diagnostic purposes. The concentration ranges of these methods were inappropriate and insufficient even modification, and the methods often lack selectivity and typically display strong matrix effect
- An LC-MS/MS approach offers high selectivity of the analysis by the ability to discriminate most of modification/degradation products from the intact analyte
- An automated solid-phase procedure has been developed coupled with LC-MS/MS detection enabling sensitive glucagon analysis using only 0.250 mL of plasma sample. Due to insufficient isotopic purity of isotopically labeled glucagon, an analog with a deletion in the amino acid sequence of glucagon was used as an internal standard

SAMPLE PREPARATION

- Aliquot of human plasma (0.250 mL) was spiked with internal standard and diluted with 1 mM glycine buffer pH 8.2.
- Samples were loaded to a 96-well ion-exchange plate and washed with glycine buffer followed by organic solvents
- Elution was performed by ammonia hydroxide in organic solvent

INSTRUMENTATION

- An Agilent Technologies, ZORBAX Rapid Resolution 300SB-C_{18}, 50 x 2.1 mm, 3.5 µm analytical column was used for LC-MS/MS
- Mobile phase containing 30% ACN with formic acid was used for separation
- AB|MDS Sciex API 4000 tandem mass spectrometer was used to detect multiply charged positive ions in the multiple-reaction-monitoring mode
- The acquisition time was less than 4 minutes

RESULTS:

- The validated analytical range was from 100 to 10,000 pg/mL with dilution integrity demonstrated up to 25,000 ng/mL
- The signal-to noise at the lower limit of quantitation (LLOQ, 100 pg/mL) was typically ≥ 15 with only about 20% of the sample volume injected (Figure 4)
- The inter-batch precision (% C.V.) of quality control samples at 300, 1000 and 7500 pg/mL was 6.5, 2.2 and 3.3%, respectively. The inter-batch accuracy (% Bias) of the same quality control samples was 6.0, 6.0 and 5.2%, respectively (Table 1)
- Assay selectivity was demonstrated by the accurate quantitation of standard spikes into six separate lots of blank human plasma (EDTA). No significant matrix effect was observed in all lots spiked at the LLOQ and in 5 of 6 lots at high QC concentration. Precision (% C.V.) of the LLOQ and high QC concentration spikes quantitation in multiple lots were 8.4 and 2.4%, respectively (Table 2)
- The average extraction recovery of glucagon was about 50% (Table 3)
- Demonstrated lot-dependent glucagon degradation in human plasma and established composition of an inhibitory cocktail to improve substantially stability of clinical samples
- Short-term stability in human plasma (EDTA) was established for at least 14 hours on ice bath under white light
- Freeze and thaw stability in human plasma (heparin) was established for six freeze (-80°C) and thaw cycles on ice bath in polypropylene tubes
- Demonstrated post preparative stability in injection solvent (quantitation against freshly extracted standards) for 129 hours at 5°C and processed sample integrity in injection solvent (re-injection stability) was established for 128 hours at 5°C
- Demonstrated accurate and precise glucagon quantitation in turbid and hemolyzed samples (Tables 4 and 5)

Table 1. Inter-Batch and Intra-Batch Precision and Accuracy for Glucagon in Human Plasma

Batch	LLOQ QC 100 pg/mL	QC A 300 pg/mL	QC B 11000 pg/mL	QC C 7500 pg/mL
15	89.6	297	1020	7600
	107	320	1040	7520
	89.8	297	1040	7650
	88.2	302	1040	7740
	116	317	1030	8110
	99.9	296	1070	8030
Intra-Batch Mean	98.4	305	1040	7780
Intra-Batch SD	11.3	10.8	16.7	241
Intra-Batch % CV	11.5	3.5	1.6	3.1
Intra-Batch % Bias	-1.6	1.7	4.0	3.7
n	6	6	6	6
16	93.5	283	1050	7640
	103	295	1030	7980
	92.9	320	1070	8460
	104	321	1080	8120
	95.8	338	1060	7760
	95.1	338	1050	8240
Intra-Batch Mean	97.4	316	1060	8030
Intra-Batch SD	4.86	22.5	17.5	305
Intra-Batch % CV	5.0	7.1	1.7	3.8
Intra-Batch % Bias	-2.6	5.3	6.0	7.1
n	6	6	6	6
17	109	~~353	1070	7850
	119	317	1060	7620
	109	~~360	1080	8010
	~123	329	1080	7600
	105	324	1080	7920
	119	312	1110	8090
Intra-Batch Mean	114	333	1080	7850
Intra-Batch SD	7.24	19.6	16.7	202
Intra-Batch % CV	6.4	5.9	1.5	2.6
Intra-Batch % Bias	14.0	11.0	8.0	4.7
n	6	6	6	6

~ = Greater than 20% theoretical
~~ = Greater than 15% theoretical

Table 2. Matrix Effect for Glucagon in Human Plasma (EDTA)

		LLOQ		High	
Batch	Lot#	100 pg/mL	% Dev.	7500 pg/mL	% Dev.
16	1	101	+1.0	8100	+8.0
	2	109	+9.0	8480	+13.1
	3	95.8	-4.2	8370	+11.6
	4	118	+18.0	8330	+11.1
	5	111	+11.0	8680	+15.7
	6	96.7	-3.3	8240	+9.9
Mean		105		8370	
% CV		8.4		2.4	
% Theoretical		105.0		111.6	
n		6		6	

Table 3. Recovery Data of Glucagon from Human Plasma (EDTA)

Theoretical Concentration:	300 pg/mL Peak Area		1000 pg/mL Peak Area		7500 pg/mL Peak Area	
Batch	Extracted	Unextracted	Extracted	Unextracted	Extracted	Unextracted
17	3395	7231	9557	21829	81065	173225
	2855	6964	10452	24084	76302	160634
	3701	6736	10136	22627	86073	178188
	3048	6593	10705	23077	77406	155176
	3219	7327	9860	20508	88088	160092
	2735	6447	11168	22759	82284	155780
Mean	3159	6883	10313	22481	81870	163849
% CV	11.3	5.1	5.7	5.4	5.7	5.8
% Recovery	46		46		50	
n	6	6	6	6	6	6

Table 4. Hemolyzed Sample Integrity for Glucagon in Human Plasma (EDTA)

		Peak Area		Interference	LLOQ		High	
Batch	Lot#	Blank	LLOQ	(% of LLOQ)	100 pg/mL	% Dev.	7500 pg/mL	% Dev.
16	1	0	969	0.0	90.6	-9.4	7070	-5.7
	2	0	1120	0.0	107	+7.0	8190	+9.2
	3	0	1055	0.0	116	+16.0	7980	+6.4
Mean					105		7750	
% CV					12.3		7.7	
% Theoretical					105.0		103.3	
n					3		3	

Table 5. Turbid Sample Integrity for Glucagon in Human Plasma (EDTA)

		Peak Area		Interference	LLOQ		High	
Batch	Lot#	Blank	LLOQ	(% of LLOQ)	100 pg/mL	% Dev.	7500 pg/mL	% Dev.
17	1	0	753	0.0	91.6	-8.4	7350	-2.0
	2	0	790	0.0	95.3	-4.7	8020	+6.9
	3	0	924	0.0	109	+9.0	8180	+9.1
Mean					98.6		7850	
% CV					9.3		5.6	
% Theoretical					98.6		104.7	
n					3		3	

Table 6. Validation Summary

Information Requested	Data
Validation Summary	Celerion Validation Study ZZ17705-05
Analyte	Glucagon
Method Description	Solid phase extraction with analysis/detection by LC MS/MS
Limit of Quantitation (pg/mL)	100 pg/mL
Average Recovery of Drug (% Mean)	46% at 300 pg/mL 46% at 1000 pg/mL 50% at 7500 pg/mL
Average Recovery of IS (% Mean)	65% (at all analyte levels)
Standard Curve Concentrations (pg/mL)	100, 150, 250, 500, 1000, 2500, 5000, 8000, and 10,000 pg/mL
QC Concentrations (pg/mL)	LLOQ QC, 300, 1000, and 7500 pg/mL
QC Intra-Batch Precision Range (% CV)	1.5 to 11.5%
QC Intra-Batch Accuracy Range (% Bias)	-2.6 to 14.0%
QC Inter-Batch Precision Range (% CV)	2.2 to 10.7%
QC Inter-Batch Accuracy Range (% Bias)	3.0 to 6.0%
Bench-Top Stability (Hrs)	Short-Term Stability: 14 hours in polypropylene tubes in an ice water bath under white light
Stock Stability (Days)	Long-Term Stability for Stock Solutions (Stock): 29 days at approximately 400 µg/mL in 25:75:0.1 acetonitrile:water:formic acid in a BSA-treated polypropylene container at -80°C
Processed Stability (Hrs)	Post-Preparative Stability: 129 hours in a polypropylene 96 well plate at 5°C Processed Sample Integrity: 128 hours in a polypropylene 96 well plate at 5°C
Freeze-Thaw Stability (Cycles)	6 freeze (-80°C)-thaw (ice water bath) cycles in polypropylene tubes under white light
Long-Term Storage Stability (Days)	Long-Term Stability: 37 days in polypropylene tubes at 80°C
Dilution Integrity	up to 25,000 pg/mL, diluted 5-fold
Selectivity	No significant interference at the retention time and mass transition of glucagon/IS was observed from endogenous components in any of the 6 human plasma (EDTA) lots screened

Figure 1. Amino acid sequence of glucagon

Analyte: Glucagon Molecular Weight: 3482 Da

NH_2-His-Ser-Gln-Gly-Thr-Phe-Thr-Ser-Asp-Tyr-Ser-Lys-Tyr-Leu-Asp-Ser-Arg-Arg-Ala-Gln-Asp-Phe-Val-Gln-Trp-Leu- Met-Asn-Thr-COOH

Figure 2. Extracted blank plasma sample

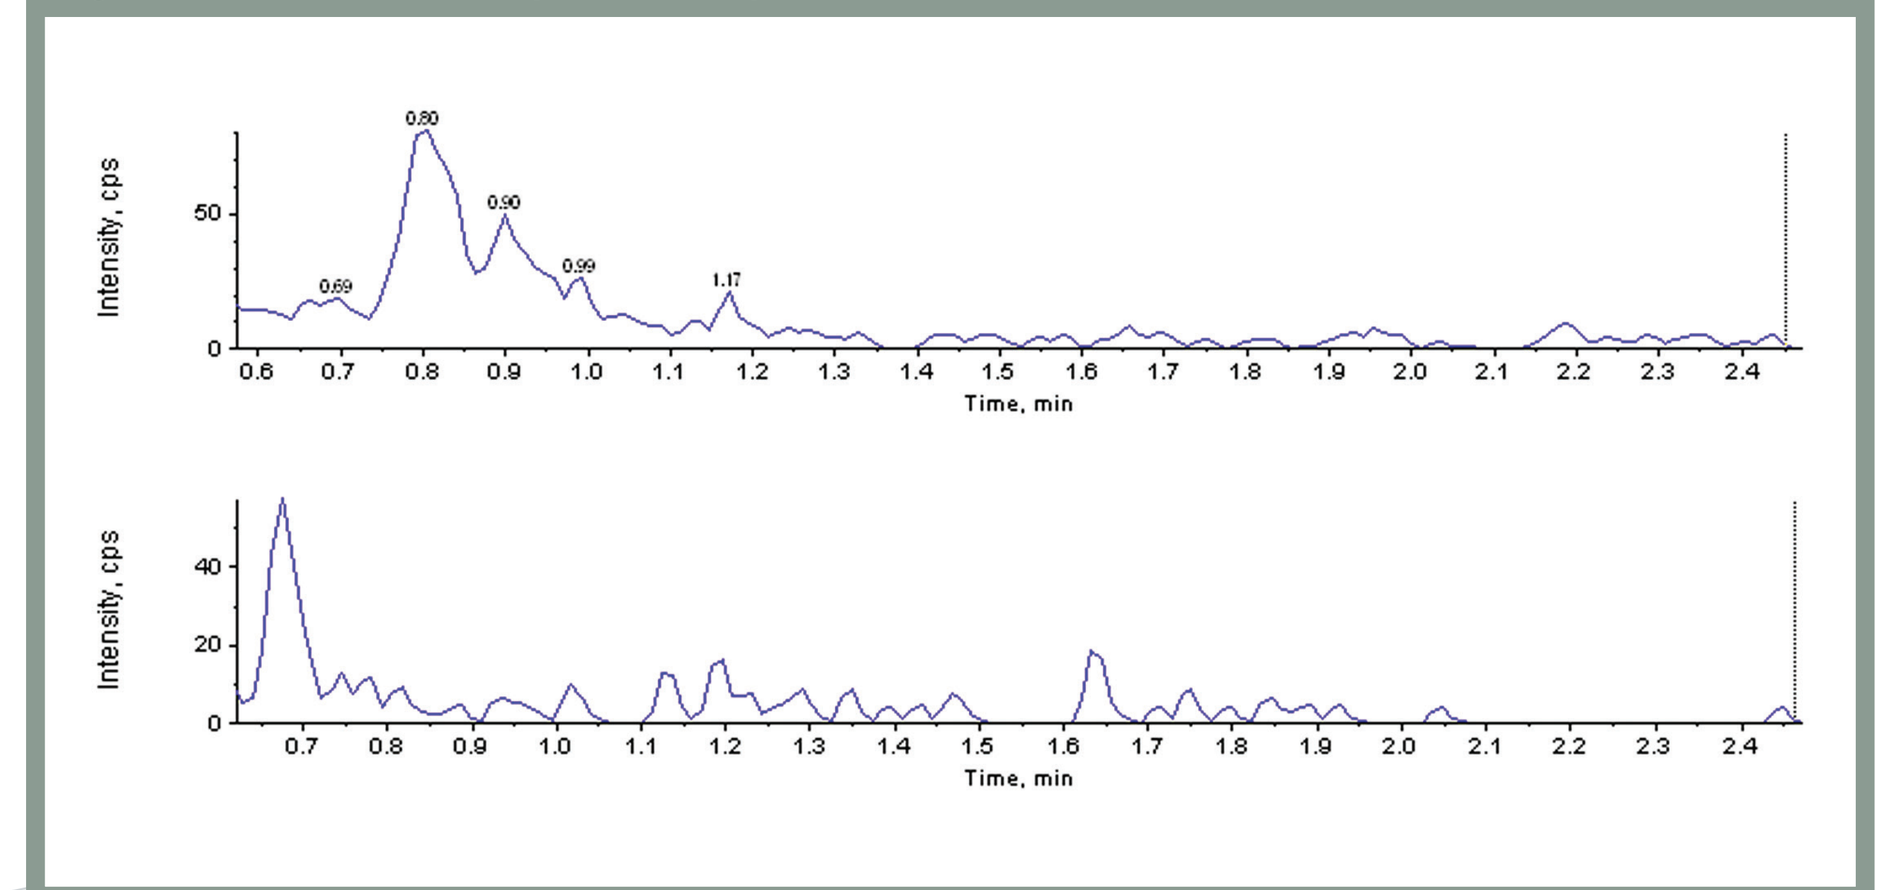
Intensity, cps
Time, min

Figure 3. Extracted plasma sample spiked with internal standard

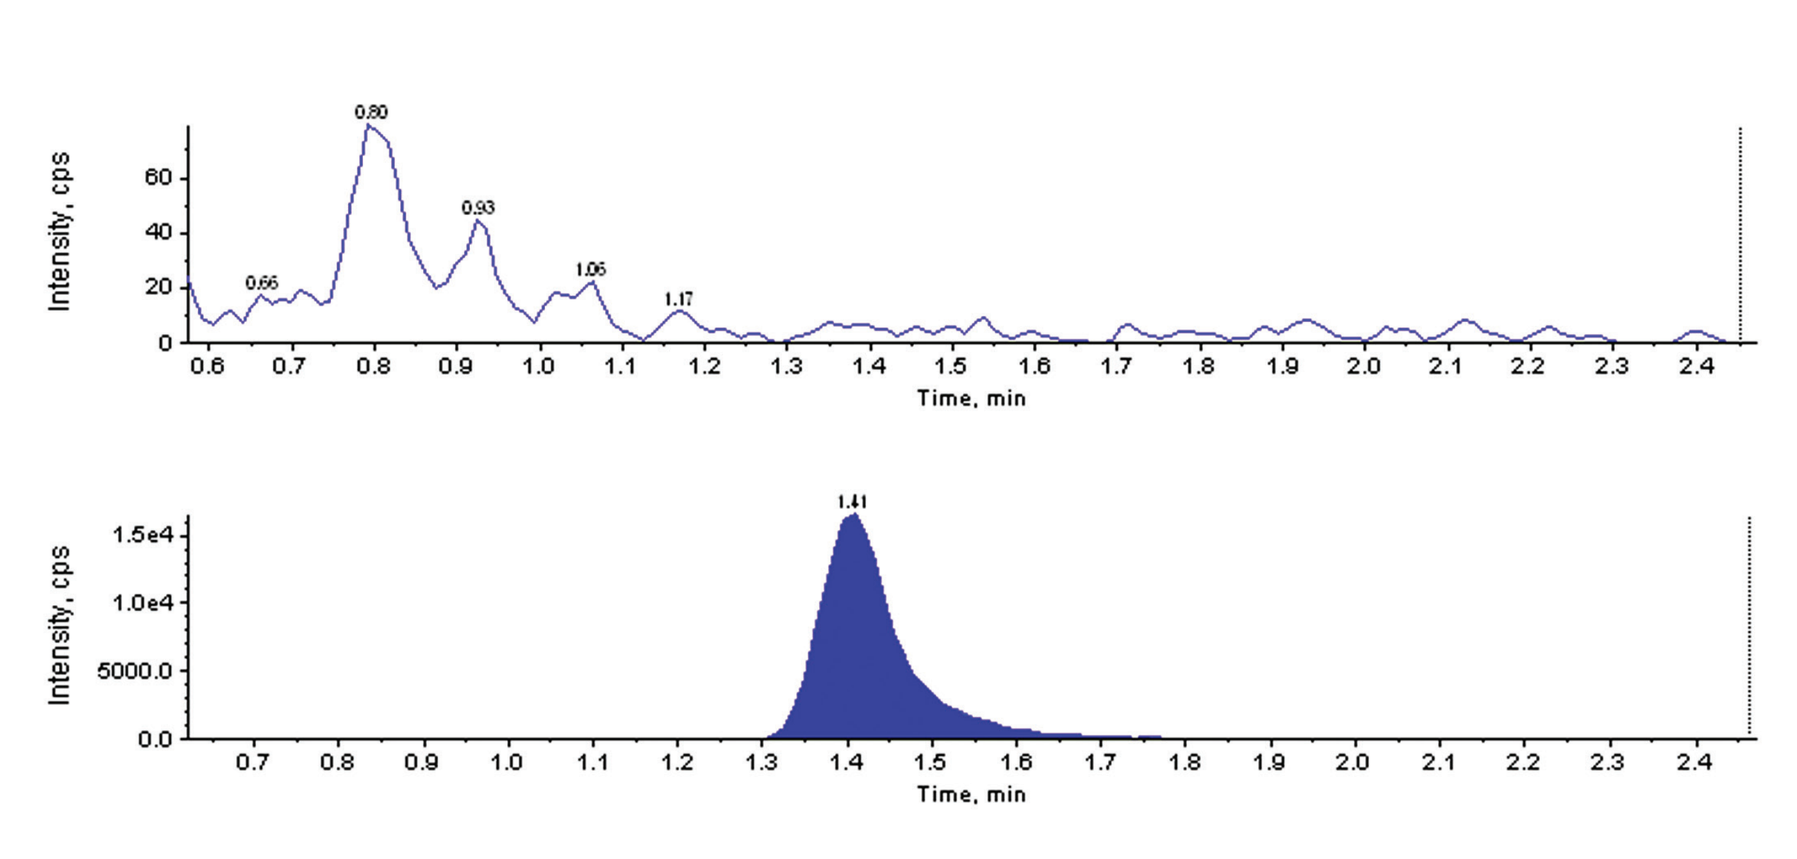
Intensity, cps
Time, min

Figure 4. Extracted LLOQ sample

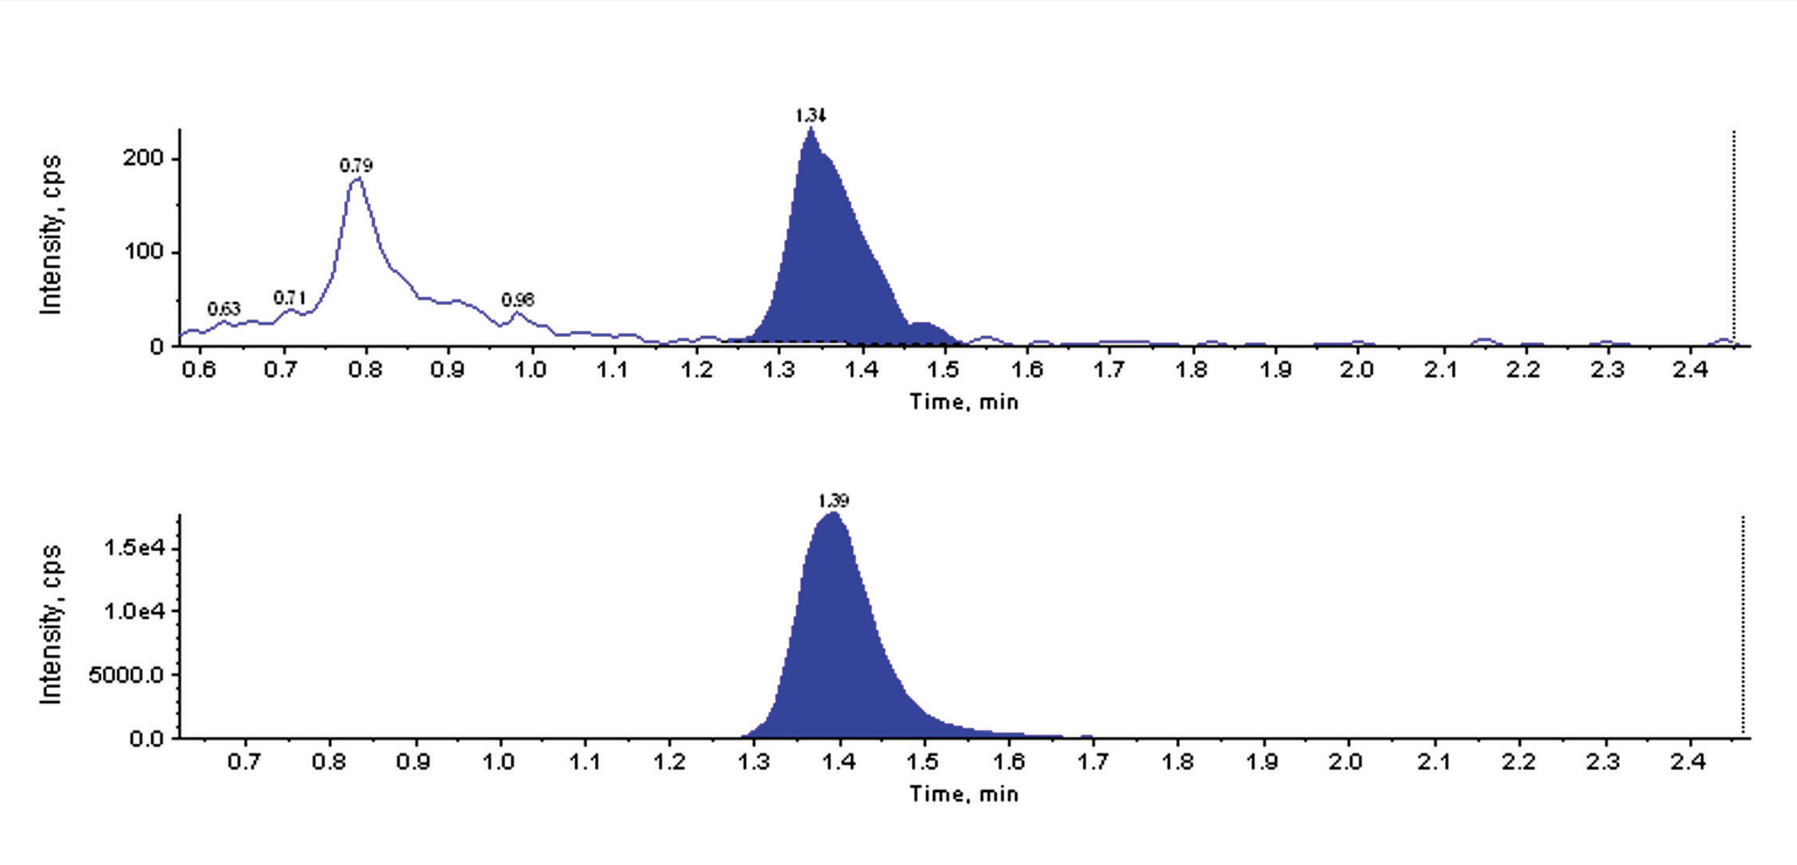
Intensity, cps
Time, min

Figure 5. Extracted ULOQ sample

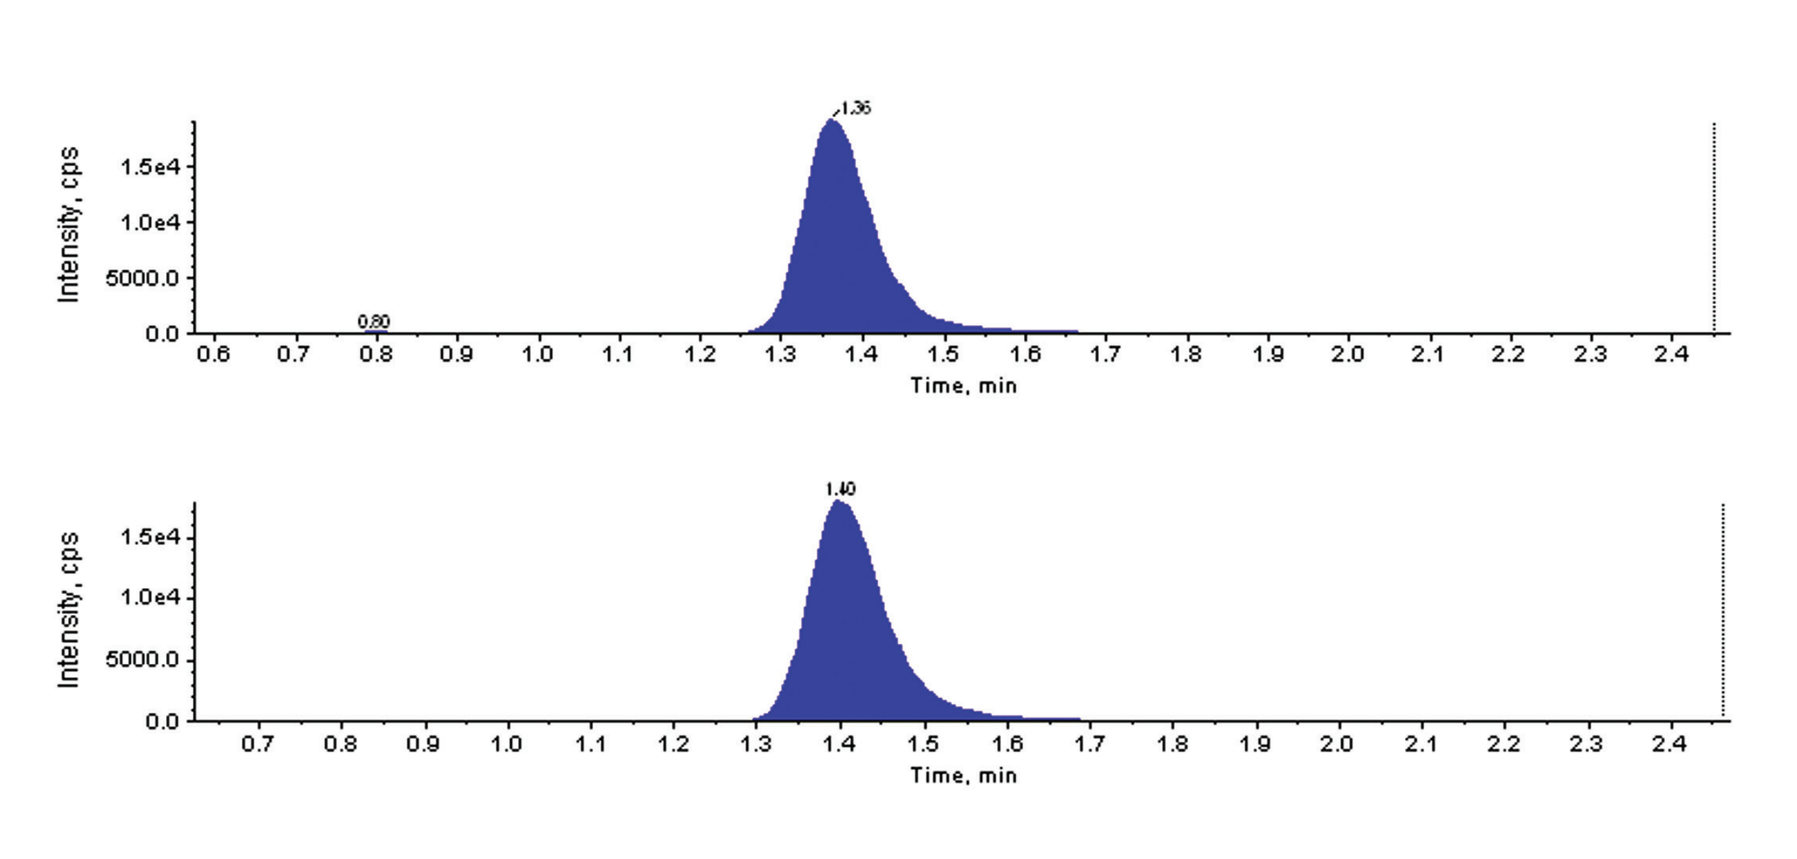
Intensity, cps
Time, min

CONCLUSIONS:

- A sensitive, accurate and reproducible method for glucagon was developed and validated with improved selectivity as compared to currently available immunochemical methods
- Developed an inhibitory cocktail to enhance significantly glucagon stability in plasma samples

www.celerion.com