T2222

Sterilization of Radiolabeled API in USP <797> Compliant Clinical Pharmacology Unit Clean Room: Application to ADME and Microtracer Studies

Jennifer L. Moorehead, Celerion, Lincoln, NE USA
Elliot Offman, Celerion, Montreal, QC Canada

celerion

BACKGROUND

Typically, parenterally administered products for clinical studies must be manufactured under GMP conditions. However, when reconstitution for injection can be performed under USP <797> Clean Room conditions by licensed pharmacists, sterility can be achieved at the clinical facility, potentially reducing the duration of finished product stability and sterility data required to enable dosing. USP <797> is a general chapter in the USP that describes requirements for the preparation of sterile drugs, including radiopharmaceuticals[1]. The rationale for USP <797> is to prevent harm and fatality to patients that could result from microbial contamination and excessive bacterial endotoxins. The regulations apply to healthcare institutions, pharmacies, physicians' offices, and other facilities where compounded sterile preparations are prepared.

PURPOSE

The purpose of this study was to determine whether sterile compounding of non-sterile radiolabeled API can result in a sterile finished product appropriate for parenteral administration in ADME and microtracer studies.

METHODS

The study included the preparation and testing of:

- A mock preparation using unlabeled API (Lot 1)
- A preparation using radiolabeled API (Lot 2)

Both preparations were prepared and handled in the same manner to minimize variability except for minor differences in the weight of the API.

The preparations consisted of weighing 12.8 mg of API into a 100 mL, depyrogenated sterile vial and reconstituting with 100 mL sterile water for injection followed by sonification.

Sterile filtration of the API solution was conducted in a Labconco Purifier® Logic® Class II Type A2 biological safety cabinet within a USP <797> Clean Room.

Both the biological safety cabinet and the USP <797> Clean Room are certified every six months to ensure that they meet the USP <797> guidelines with certification consisting of:

- Microbial surface testing
- Microbial air testing
- Air particle testing
- Magnahelic gauge pressure verification

Filtration was accomplished by filtering the contents of the 100 mL sterile vial through a Millex ®LG 0.20 micron hydrophilic filter.

The filtered solution was transferred into sterile vials containing a 1 mL aliquot of filtered solution and 14 mL of sterile 0.9% NaCl.

Testing of the final solution was performed at Microtest Laboratories (Agawam, Massachusetts) under cGMP, ISO 9001:2008 and ISO 17025:2011 and included:

- Bacteriostasis/fungistasis using the Steritest method[2] (to ensure our product did not interfere with sterility testing)
- Endotoxin using the Kinetic Turbidimetric method[3]
- Sterility

RESULTS OVERVIEW

Preparation with filtration resulted in a sterile, endotoxin free solution acceptable for administration to human participants via IV infusion. The product successfully qualified for USP <71>/ EP2.6.1/21CFR610.12[4,5]. The results of the mock preparation (Lot #1) are summarized in Tables 1, 2 and 4. The results were similar for Lot #2 and are summarized in Tables 3 and 5.

Table 1: Bacteriostasis and Fungistasis Steritest Qualification Testing
Product: Cold Mock Product, Lot #1
Test Date: 24 Feb 2012
Method: USP <71>/EP 2.6.1 Sterility Tests

Organism	Reference Plates CFU's (Plate1/Plate2)	Medium	Specification	Result	Pass/Fail
Bacillus Subtilis (B. atrophaeus)	(69/59)	TSB	Sample and fungal positive controls Growth observed < 5 days	Growth observed at day 3 for positive control and sample	Pass
Aspergillus brasiliensis (A. niger)	(36/43)	TSB		Growth observed at day 3 for positive control and sample	Pass
Candida albicans	(39/47)	TSB	Bacterial Positive controls Growth observed <3	Growth observed at day 3 for positive control and sample	Pass
Clostridium sporogenes	(47/72)	FTM		Growth observed at day 3 for positive control and sample	Pass

Table 2: Endotoxin Testing
Product: Cold Mock Product, Lot #1
Test Date: 24 Feb 2012
Method: EP 2.6.14/USP<85> Turbidimetric Method

Criteria	Result	Pass/Fail
Correlation Coefficient absolute value greater than or equal to 0.980	0.9981	Pass
Mean PPC must be within 50-200% of the known concentration of 0.5EU/mL	96% @ the neat concentration	Pass
Negative Control must be below the lowest point of the curve	LRW: <0.005 EU/mL	Pass
Test sample specification is "for information only"	<0.005EU/mL	Pass

Table 3: Endotoxin Testing
Product: Hot Product, Lot #2
Test Date: 21 Mar 2012
Method: EP 2.6.14/USP<85> Turbidimetric Method

Criteria	Result	Pass/Fail
Correlation Coefficient absolute value greater than or equal to 0.980	0.9994	Pass
Mean PPC must be within 50-200% of the known concentration of 0.5EU/mL	105% @ the neat concentration	Pass
Negative Control must be below the lowest point of the curve	LRW: <0.005 EU/mL	Pass
Test sample specification is "for information only"	<0.005EU/mL	Pass

Table 4: Sterility Testing
Product: Cold Mock Product, Lot #1
Date Started: 17 Feb 2012
Date Completed: 02 Mar 2012
Method: USP 34<71>/EP 2.6.1

Test/Procedure	Specification	Results
Sterility- Steritest Membrand Filtration	**USP 34 <71>/EP 2.6.1**	**No Growth**

Table 5: Sterility Testing
Product: Hot Product, Lot #2
Date Started: 19 Mar 2012
Date Completed: 02 Apr 2012
Method: USP 34<71>/EP 2.6.1

Test/Procedure	Specification	Results
Sterility- Steritest Membrane Filtration	**USP 34 <71>/EP 2.6.1**	**No Growth**

CONCLUSIONS

Sterilization of non-sterile radiolabeled API can be achieved on-site for parenteral administration to human participants by licensed pharmacists in a USP <797> compliant Clean Room. On-site sterile formulation is an innovative alternative for clinical studies which require an IV formulation for a specific study. This approach will save time and money in drug development by eliminating very lengthy and costly manufacturing steps in the formulation process.

REFERENCES

1. USP General Chapter <797> Pharmaceutical Compounding – Sterile Preparations. United States Pharmacopeia. 2010
2. http://www.millipore.com/downstream/cp1/steritest_sterility_testing_media
3. http://www.criver.com/SiteCollectionDocuments/qc_mm_r_kta2_package_insert.pdf
4. http://www.accessdata.fda.gov/scripts/cdrh/cfdocs/cfcfr/cfrsearch.cfm?fr=610.12
5. USP General Chapter <71> Sterility Testing. United States Pharmacopeia. 2010

www.celerion.com