A NOVEL APPROACH TO COST EFFECTIVE THOROUGH QT STUDIES UTILIZING A HIGHLY AUTOMATED HYBRID PHASE I/ECG CORE LABORATORY

Bill Wheeler, MD, FACC and Joy Olbertz, PharmD, PhD
Celerion

UNMET NEED:

ICH E14 mandates a dedicated study for essentially all small molecules to assess the risk of QT prolongation on the ECG that can lead to the potentially fatal cardiac arrhythmia, Torsade de Pointes. This highly specialized study, the Thorough QT/QTc (TQT) study, is expensive and a challenge for sponsors to perform. Traditional ECG core labs have resisted the implementation of cost effective approaches for ECG processing in TQT studies. We present a comprehensive plan to simplify planning, bidding, contracting, and executing of TQT studies while markedly decreasing overhead and ECG processing costs by about 50%. By using optimal automation of ECG processing and measurement, the cardiologist review costs and time are reduced by approximately 90%. Celerion performed a randomized, double-blind placebo and active control trial to test the system and presented that data to the FDA.

PROPOSED SOLUTION:

Based upon experience working with all the major ECG core labs in 36 TQT studies and over 140 Phase I studies with intensive ECG monitoring, Celerion has identified three area where costs efficiences could be realized while maintaining or increasing data quality. These areas are hybrid Phase I/ECG Core lab formation, equipment selection and ECG processing.

Traditional core labs were designed to service large, global late stage trials that require much greater resources than do TQT studies. One traditional ECG core lab recently revealed that 92% of their cost for an ECG was not related to ECG processing, but to support of that overhead[1]. By combining the functionality of both the Phase I unit and the ECG core lab into a single Hybrid Phase I/ECG core lab serving only Celerion Phase I units we are able to eliminate most of the overhead of traditional ECG core labs and duplication of services already operating in Celerion clinics. Combining the two functions into one entity also provides for smoother execution and coordination of functions.

Another opportunity for cost savings was identified in equipment selection. Celerion selected the Global Instrumentation M12R Holter monitor as our 12 lead ECG data collection device. The M12R has been tailored specifically for Phase I use to increase ease and efficiency of data collection. Capabilities such as a computer generated date/time stamps and preconfigured demographics uploaded by bluetooth from Celerion central computer systems help to minimize errors. Bluetooth functioinality also improves conduct efficiency by allowing the capture of safety ECGs while the Holter is acquiring without the concurrent use of a stand alone ECG as is required with most Holters. A relatively small number of Holters can service all four of our Celerion Phase I sites which is much more cost effective than four fixed telemetry systems. Holter monitors also have the advantage that the ECG data is stored on a flash card, rather than transmitted to a central station minimizing potential for lost data due to signal disruption. The M12R Holter monitor stores that data on an SD card, which has a lower failure rate than Compact Flash cards used in many Holter systems.

Finally, implementation of optimal automation in the processing of ECGs was another opportunity for significant cost savings. Celerion helped develop and implement a suite of software with AMPS, LLC a world leader in ECG measurement software. First, AMPS' Antares software is used to automatically extracts optimal 10 second ECG recordings from the Holter recording based upon levels of artifact and stable preceding heart rate (Fig. 1). This has been shown to decrease variability over manual methods[2]. Next, to decrease measurement variability further by smoothing beat to beat heart rate variability and minimizing artifact, representative beats for each of the 12 leads are generated and used for measurement, rather than three "raw" beats as is done in some ECG core labs. FAT QT and CalECG software then perform the appropriate measurements and classifies the recordings based upon predetermined characteristics that impair automated measurement accuracy. Those recordings with insufficient quality or other characteristics that prevent accurate automated measurements are routed to a cardiologist for review while the automated measurement for the acceptable recordings are transferred directly into the database (Fig.2). This decreases the cardiologist review by about 90%.

To identify parameters that determine whether an ECG is reviewed by a cardiologist, Celerion evaluated several previously analyzed QT studies with this software, but felt that a true test required a clinical trial testing the organization, equipment and software. Therefore, a 36 subject randomized, double-blind, placebo and moxifloxacin controlled, two way crossover study was performed to evaluate the entire system.

This study revealed a typical time course and magnitude of moxifloxacin QT prolongation (Fig.3). Holter monitors came off on a Friday and the preliminary data was presented at a symposium on the following Tuesday morning. The study required approximately 8 hours of cardiologist review time. Approximately 11% of ECGs were reviewed by a cardiologist and 2% of ECG measurements were actually adjusted by the cardiologist. The intra-reader variability was 2.5 ms and the average within subject variability was 6.5 ms. Using these quality parameters, a typical single dose, crossover TQT study could be perfomed with approximately 20-25 subjects (Fig. 4).

Celerion presented this data to the Interdisciplinary Review Team (IRT) at the FDA who agreed that data was typical for a moxifloxacin effect. They also made the point that there was no concern using this approach on TQT trials assuming that moxifloxacin response is identified appropriately, an an expectation of any TQT regardless of how the data is reviewed.

SUMMARY:

Celerion has developed a Hybrid Phase I/ ECG core lab that minimizes overhead and utilizes bluetooth Holter monitors that eleminate the need for stand alone ECG machines and decreases data entry errors. The highly automated approach to ECG analyses not only decreases costs significantly, but also decreases variability resulting in the potential for smaller TQT sample sizes and further savings to sponsors. The FDA expressed no concern using this system in a TQT study provided an appropriate moxifloxacin effect was seen, a requirement for any TQT study regardless of the measurement method used.

Potential Commercial Markets:	All nontoxic, small molecule compounds in development targeting FDA approval.
IP Status:	IP for software property of AMPS, LLC and Global Instrumentation
Potential Collaborators:	AMPS, LLC and Global Instrumentation
Next Steps:	Marketing Roll out

***Figure 1:** Antares® Optimal ECG Extraction. The time window selected around nominal time point is at the top. On the left side the preceding HR and artifact level are acceptable. On the right side at the nominal extraction time there is significant preceding HR instability and artifact. Decreasing HR instability and artifact decreases variability.*

In the HR scale, the blue line is the beat to beat variability and the fuchsia is smooth HR

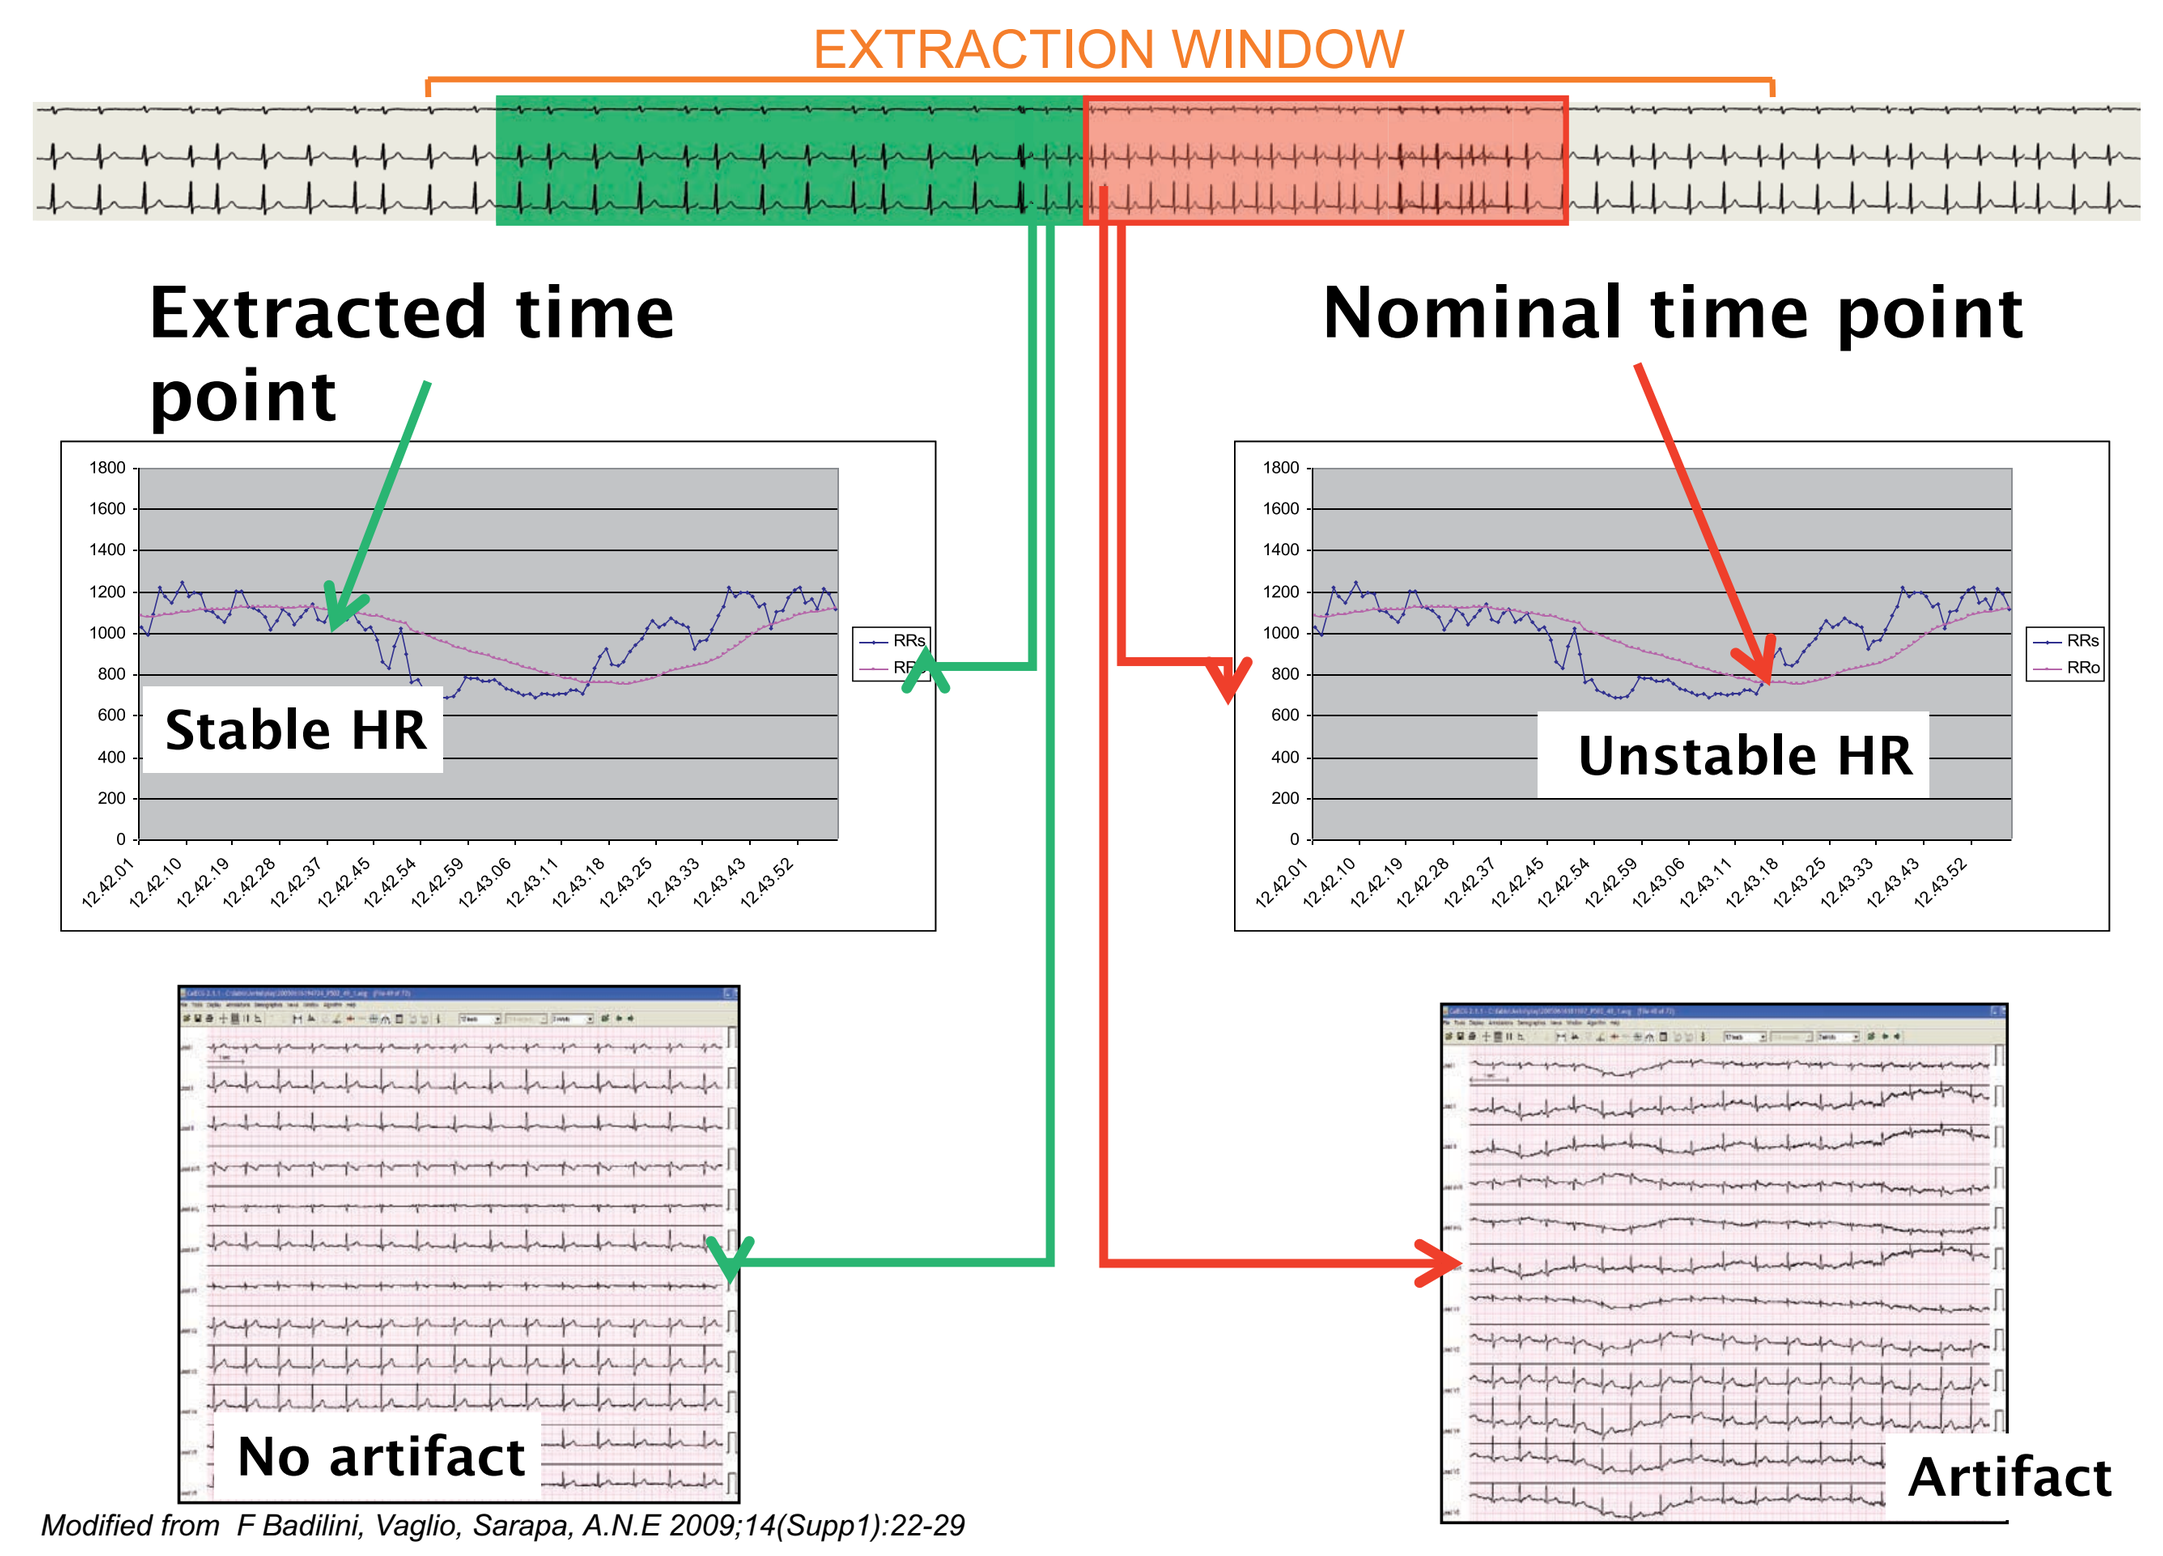
EXTRACTION WINDOW
Extracted time point
Nominal time point
Stable HR
Unstable HR
No artifact
Artifact
Modified from F Badilini, Vaglio, Sarapa, A.N.E 2009;14(Supp1):22-29

***Figure 2:** This graph shows the correlation of automated ECG measurements with manually adjudicated measurements. The closer to the zero line the better the correlation. Although statistically the correlation is good, there are outliers that increase variability. FATQT is able to identify outliers based upon specific criteria in the recording and assign them to a cardiologist for review , minimizing variability. The tracing below shows superimposed representative complex method used for ECG reading at Celerion. The gray area outlines the automated measurements, while the red arrow demonstrates the true end of the T wave identified by the cardiologist . This ECG demonstrates an abnormal T wave morphology which impairs automated measurement accuracy.*

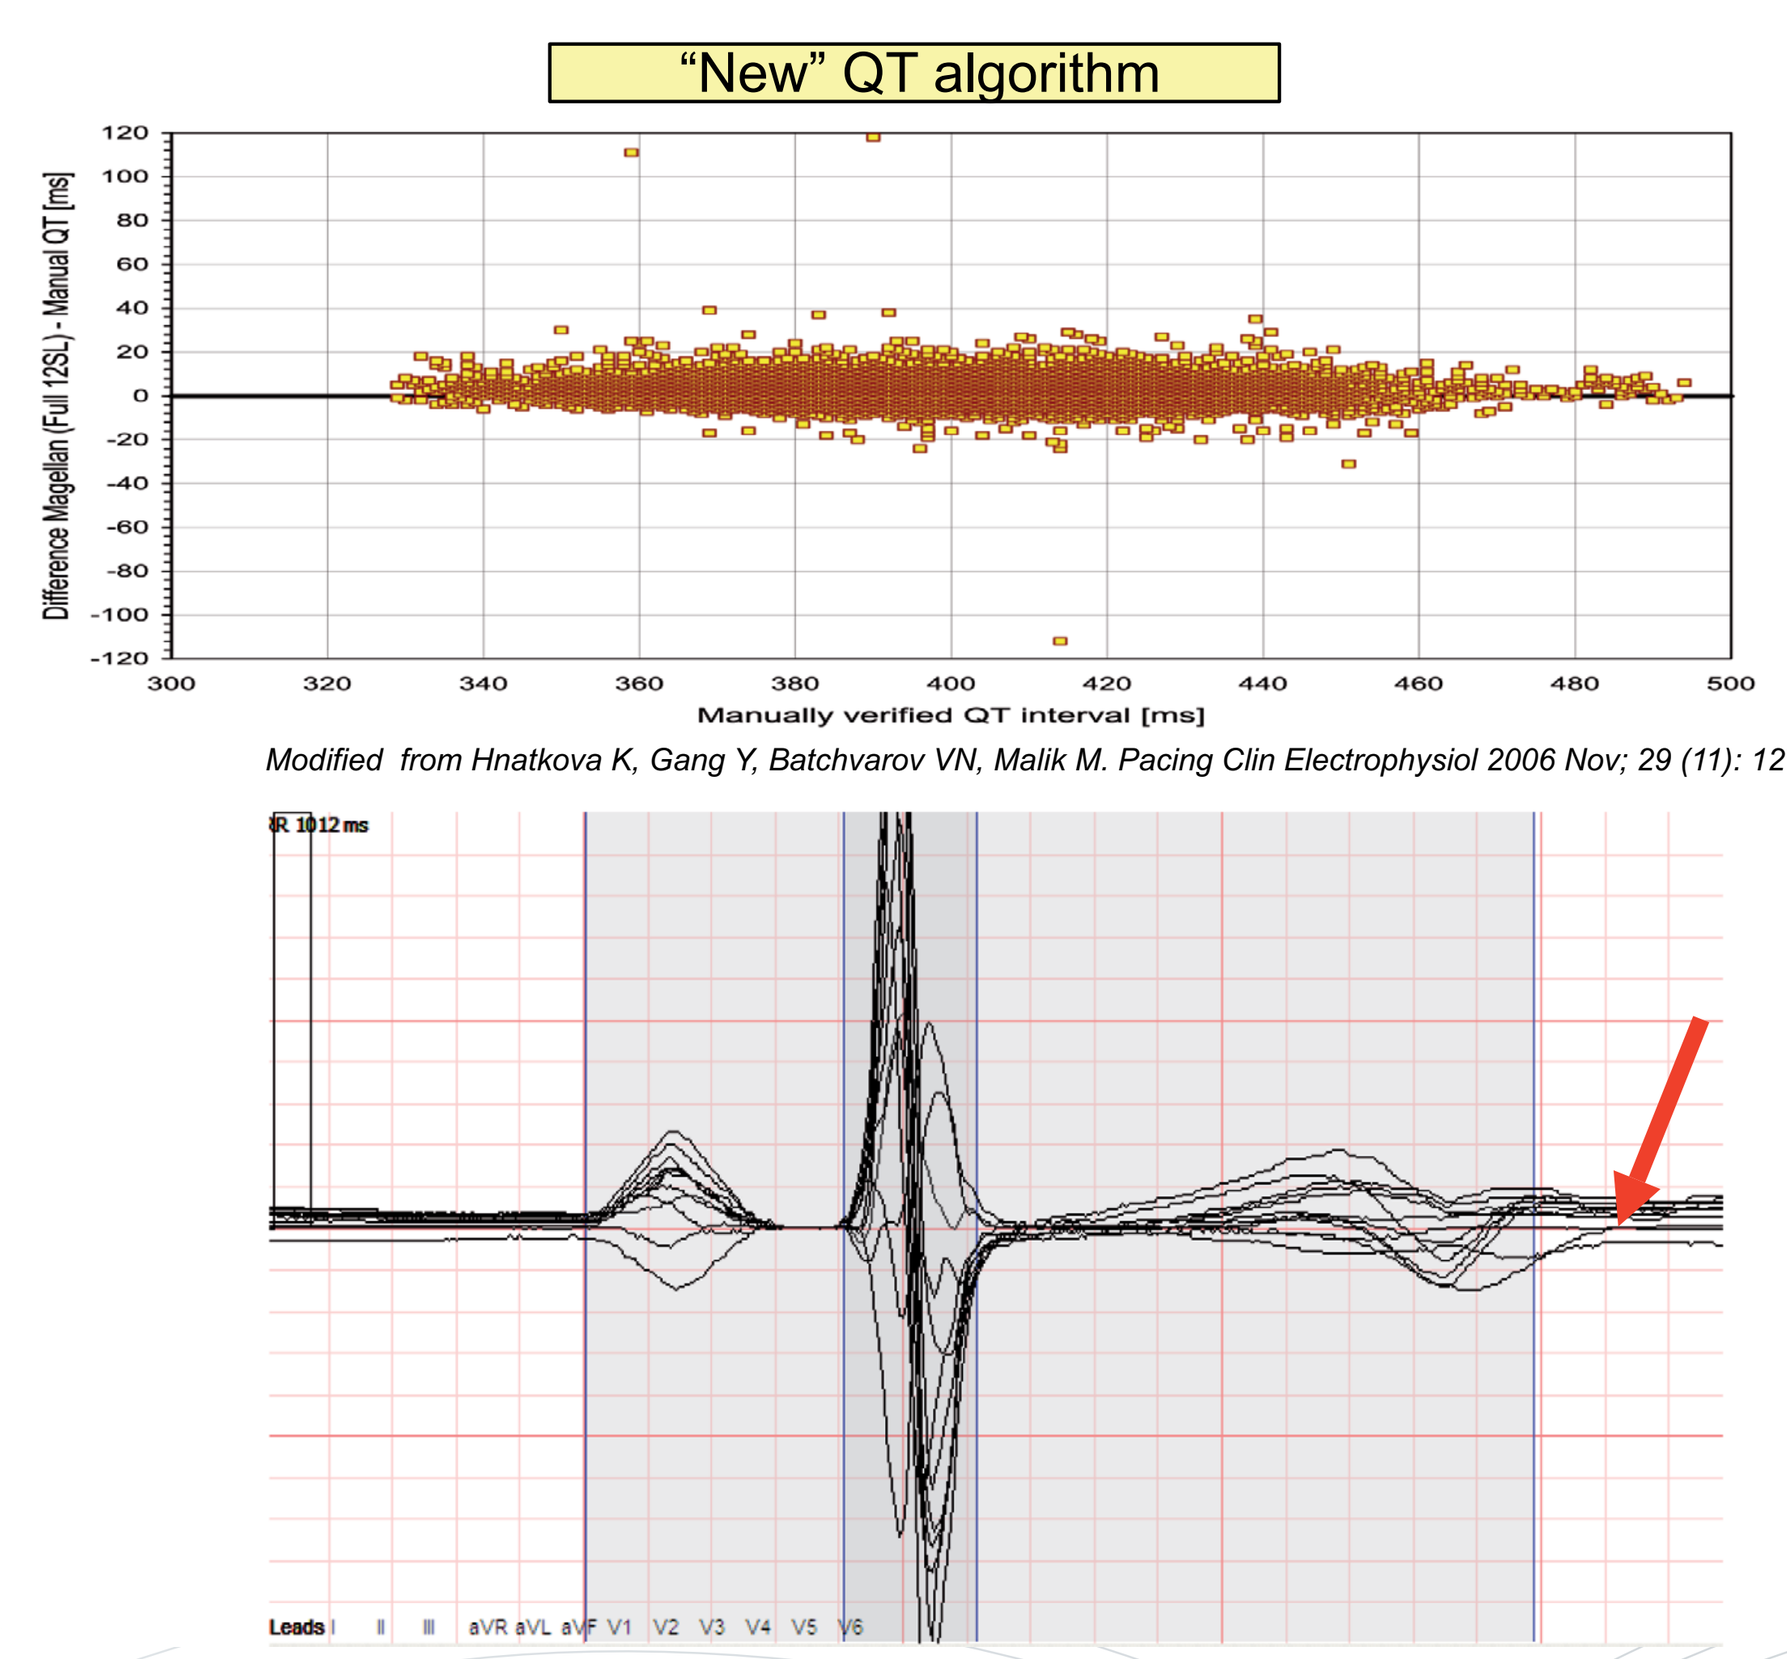
"New" QT algorithm
Difference Magellan (Full 12SL) - Manual QT [ms]
Manually verified QT interval [ms]
Modified from Hnatkova K, Gang Y, Batchvarov VN, Malik M. Pacing Clin Electrophysiol 2006 Nov; 29 (11): 1277-84.
RR 1012 ms
Leads I II III aVR aVL aVF V1 V2 V3 V4 V5 V6

***Figure 3:** Change in baseline adjusted, placebo extracted (so-called double delta) moxifloxacin effect on QTcF interval in Celerion trial. Note typical magnitude and time course of the drug effect. For a TQT study to be judged adequate by the FDA, the moxifloxacin effect must have a lower 95% confidence intervals (CI) greater than 5 ms (red dashed line) around maximal effect. At 24 hours moxifloxacin effect has decreased to 5 ms. At the 5 ms effect level this study would be able to exclude a upper 95% CI of 10 ms (green dashed line), the criteria for a positive study.*

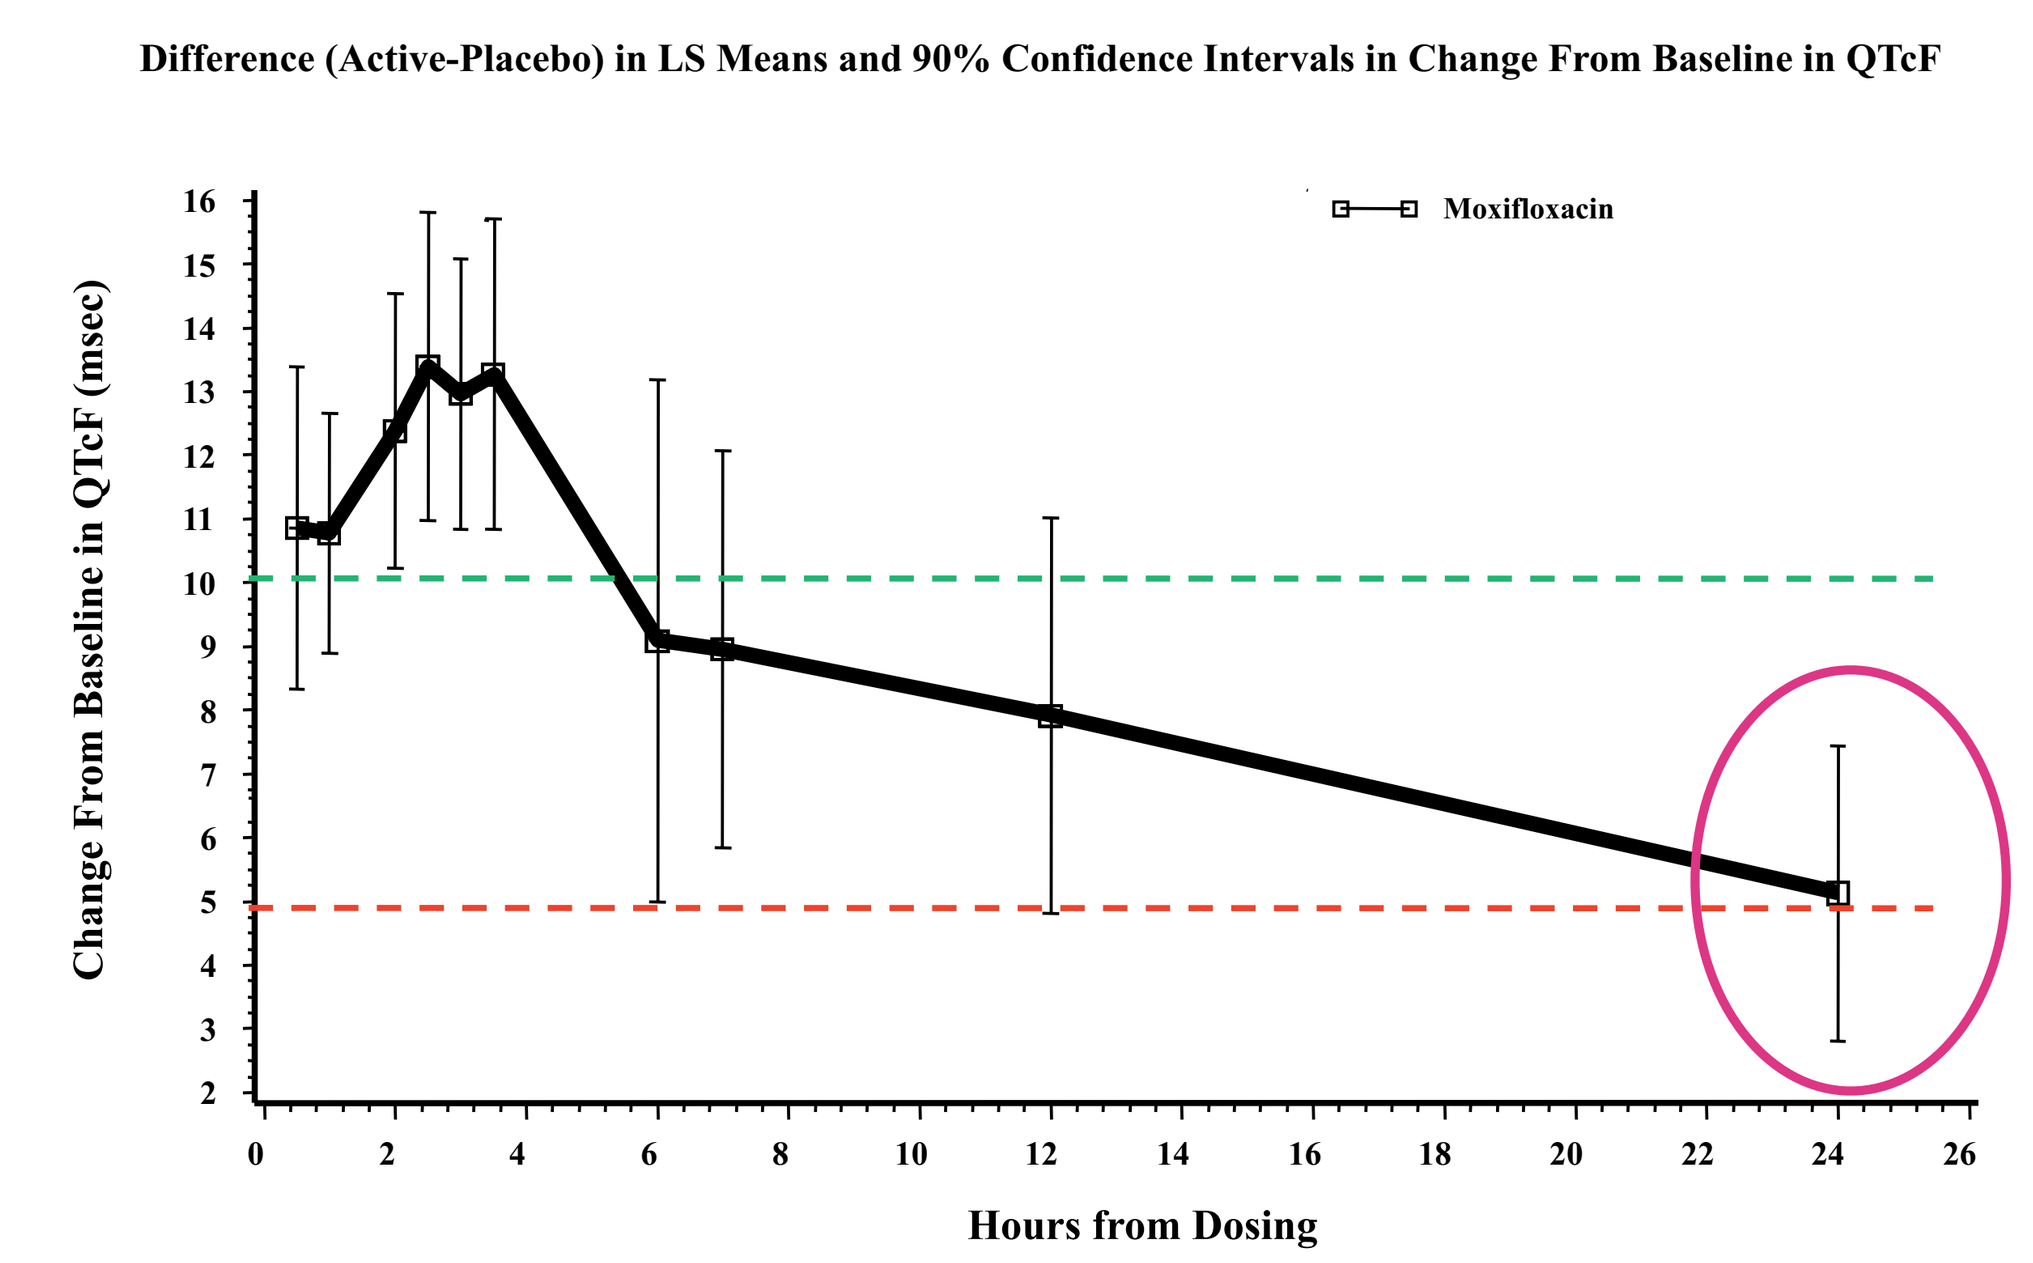
Difference (Active-Placebo) in LS Means and 90% Confidence Intervals in Change From Baseline in QTcF
Moxifloxacin
Change From Baseline in QTcF (msec)
Hours from Dosing

***Figure 4:** The sample size for a TQT study is related to the square of the variability (σ^2). Typically a within subject variability of approximately 10 ms is used for sample size calculations. With an assumed 3 ms drug effect that variability results in Sample size of 45. Using the variability obtained in our trial, average of 6.5 ms, that sample size would be about 20.*

$$N = \frac{2\,\sigma^2 (Z_{1-\alpha} + Z_{1-\beta})^2}{[10-(\mu_{drug}-\mu_{placebo})]^2}$$

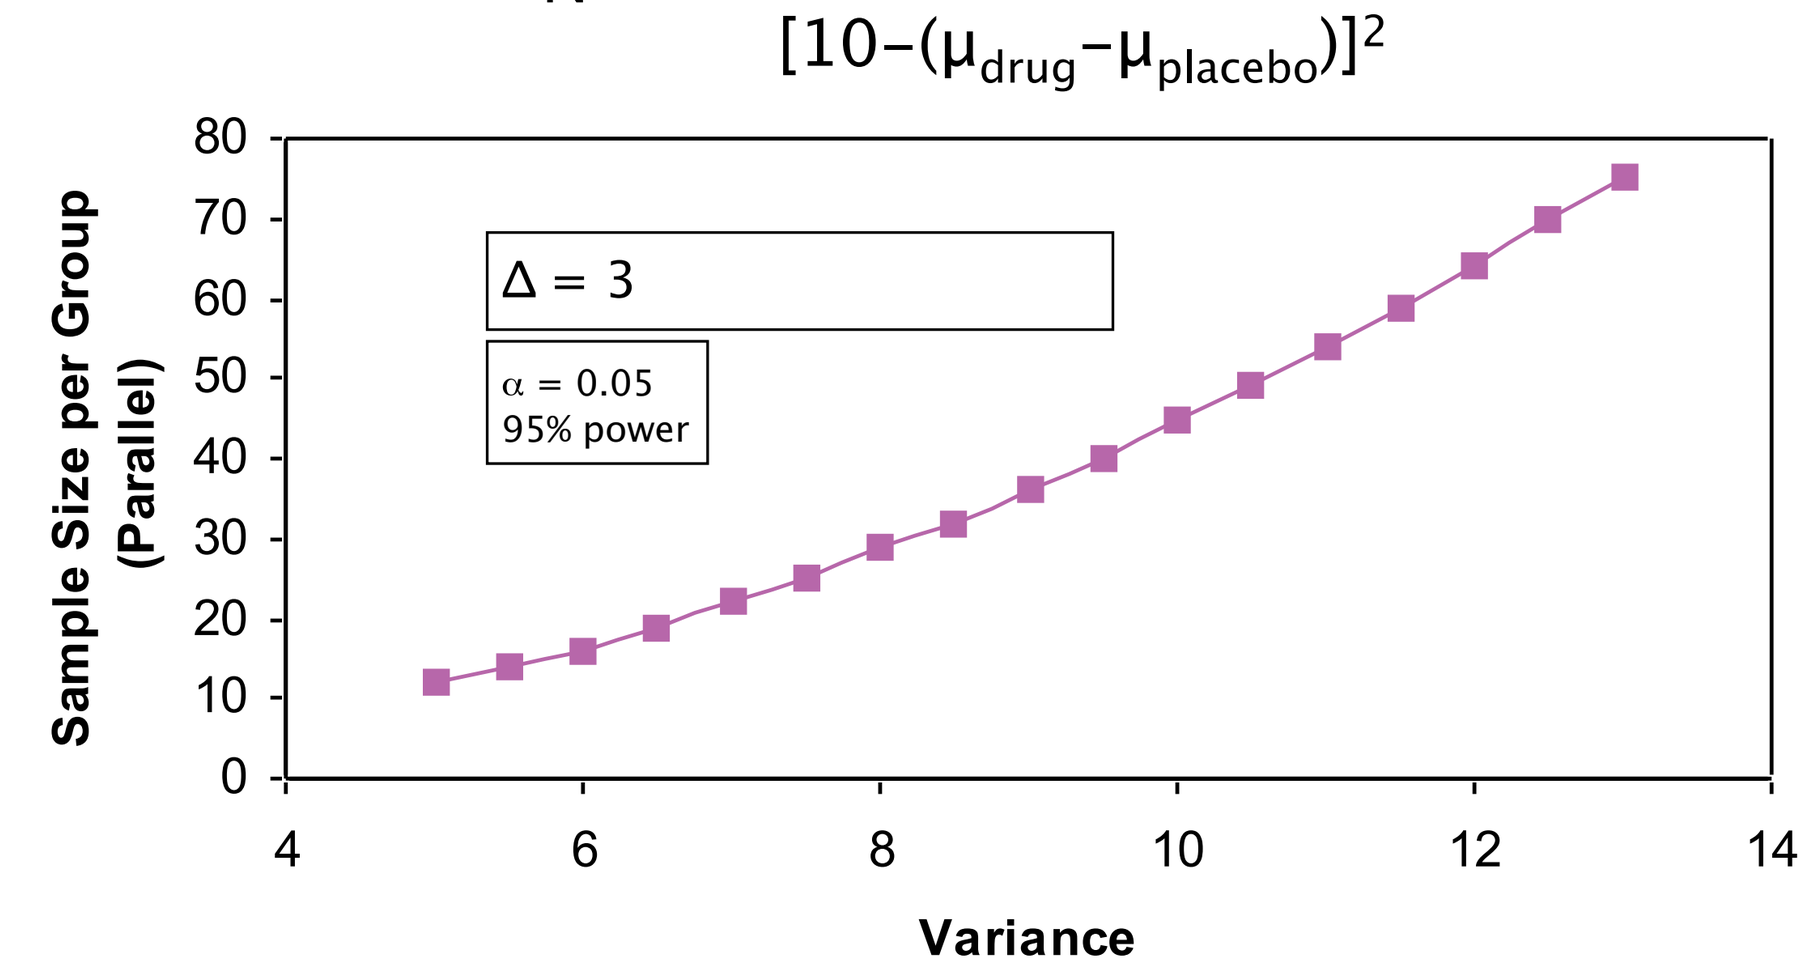
Δ = 3
α = 0.05
95% power
Sample Size per Group (Parallel)
Variance

σ^2	N
5	12
5.5	14
6	16
6.5	19
7	22
7.5	25
8	29
8.5	32
9	36
9.5	40
10	45
10.5	49
11	54
11.5	59
12	64
12.5	70
13	75

Modified from Zhang, L et al. 2008. Sample Size Calculations in Thorough QT Studies. J Biopharmaceutical Statistics 18:468.

REFERENCES:

1 Morganroth J. 'Auto Algorithm vs Manual Measurement of ECGs', presentation given at DIA Cardiovascular Safety in Drug Development: Arrhythmias, Thrombosis, Bleeding, held in Arlington, Virginia 12-13 April 2010.

2 Badilini F, Vaglio M, Sarapa N.'Automated extraction of ECG strips from continuous 12-lead Holter recordings for QT analyses at prescheduled versus optimized time points', *Ann Noninvsive Electrocardiol* 2009, 14(Suppl1):S22-29.

www.celerion.com